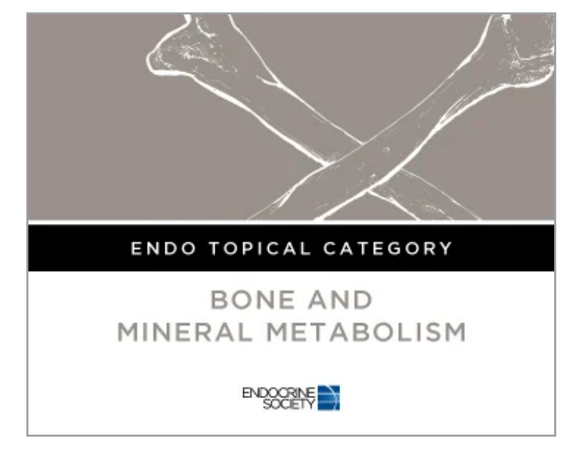

ENDO TOPICAL CATEGORY
BONE AND
MINERAL METABOLISM

Palopegteriparatide Enhances Work Functioning and Productivity in Employed Adults With Chronic Hypoparathyroidism: 156-Week Results From the PaTHway Trial

SUN-770

Wahidullah Noori,[1] Carol Zhao,[1] Alden R Smith,[1] Christopher T Sibley,[1] Aimee D Shu[1]

[1]Ascendis Pharma LLC, Palo Alto, CA, USA

Conclusions

Palopegteriparatide treatment was associated with early improvements in work functioning and productivity in adults with hypoparathyroidism.

- Positive changes were observed across all Work Limitations Questionnaire (WLQ)-25 domains and sustained over 3 years.
- The improvement in productivity scores may reflect increased functional capacity at work and may be economically relevant, indicating greater productivity while at work.
- These findings support the potential long-term functional and economic value of palopegteriparatide treatment for adults with hypoparathyroidism.

Background

- An intact parathyroid hormone (PTH) axis maintains normal serum and urine calcium and phosphate homeostasis.[1-3]
- PTH is the primary regulator of calcium/phosphate balance, acting directly on bone and kidney, and indirectly on the intestine.[4,5]
- Conventional therapy for hypoparathyroidism (active vitamin D [calcitriol] and oral calcium) aims to alleviate hypocalcemic symptoms but fails to restore normal PTH physiology.[6]
- PTH replacement therapy for hypoparathyroidism should provide PTH levels within the physiological range and restore downstream calcitriol, promoting independence from conventional therapy and normalizing:
 - Serum and urine calcium and phosphate
 - Skeletal health
 - Quality of life

Figure 1. **PTH Therapy for Hypoparathyroidism**[7]

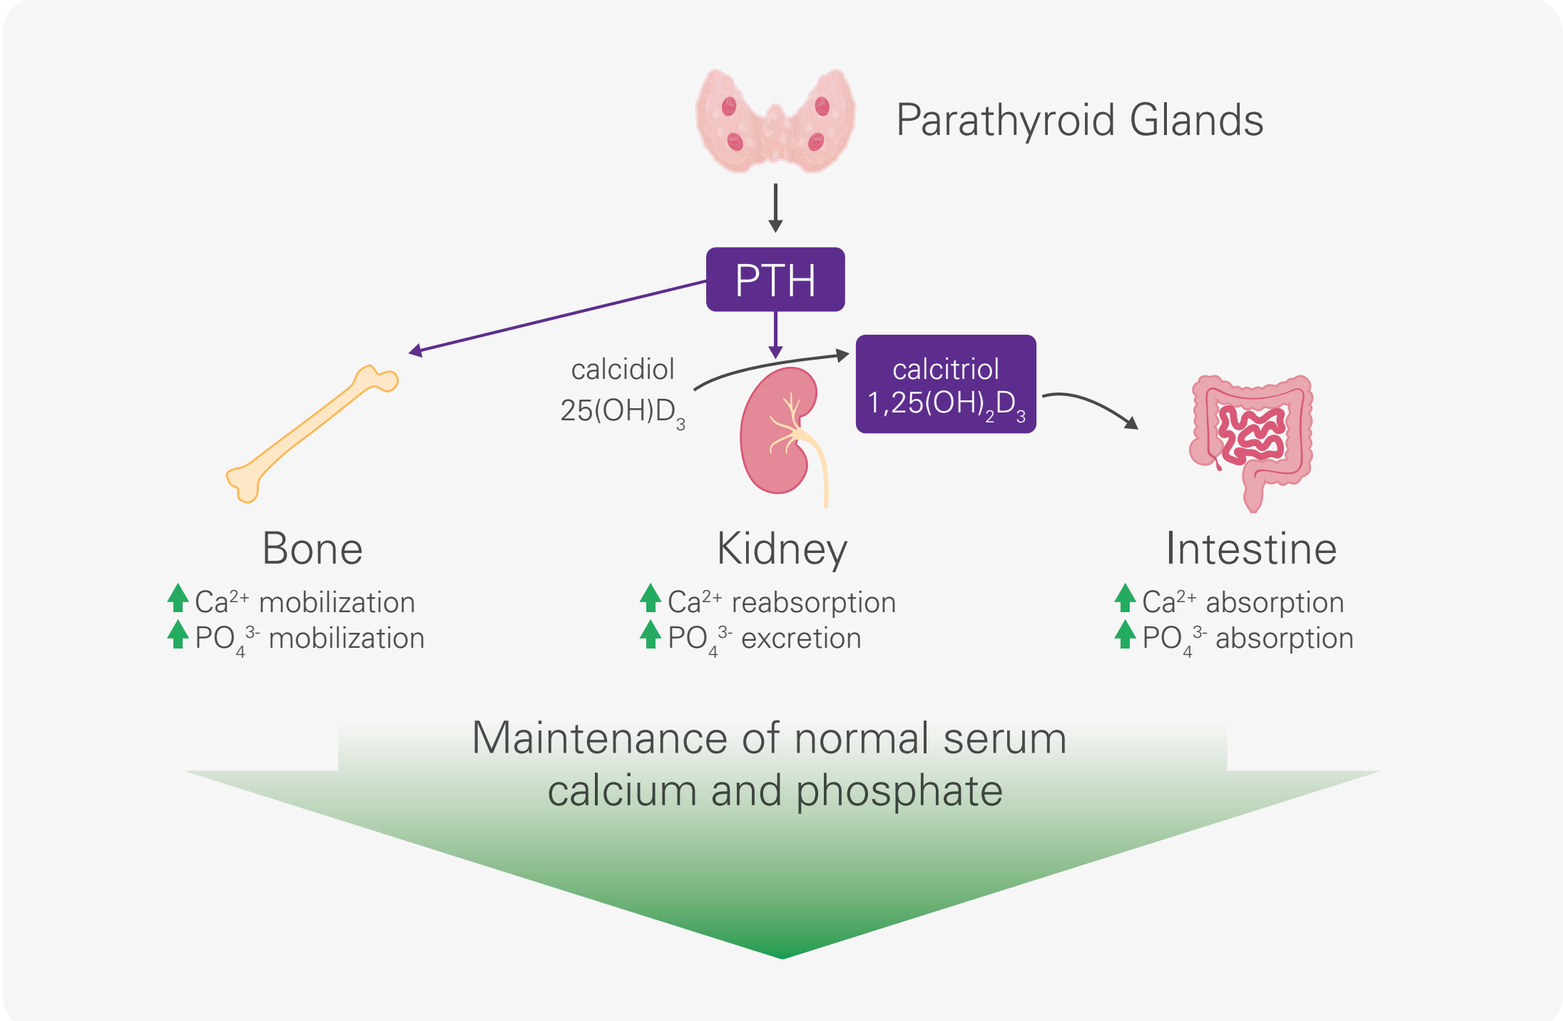

Parathyroid Glands
PTH
calcidiol 25(OH)D3
calcitriol 1,25(OH)2D3
Bone
Ca2+ mobilization
PO43- mobilization
Kidney
Ca2+ reabsorption
PO43- excretion
Intestine
Ca2+ absorption
PO43- absorption
Maintenance of normal serum calcium and phosphate

Figure 2. **Palopegteriparatide (TransCon® PTH) Design**

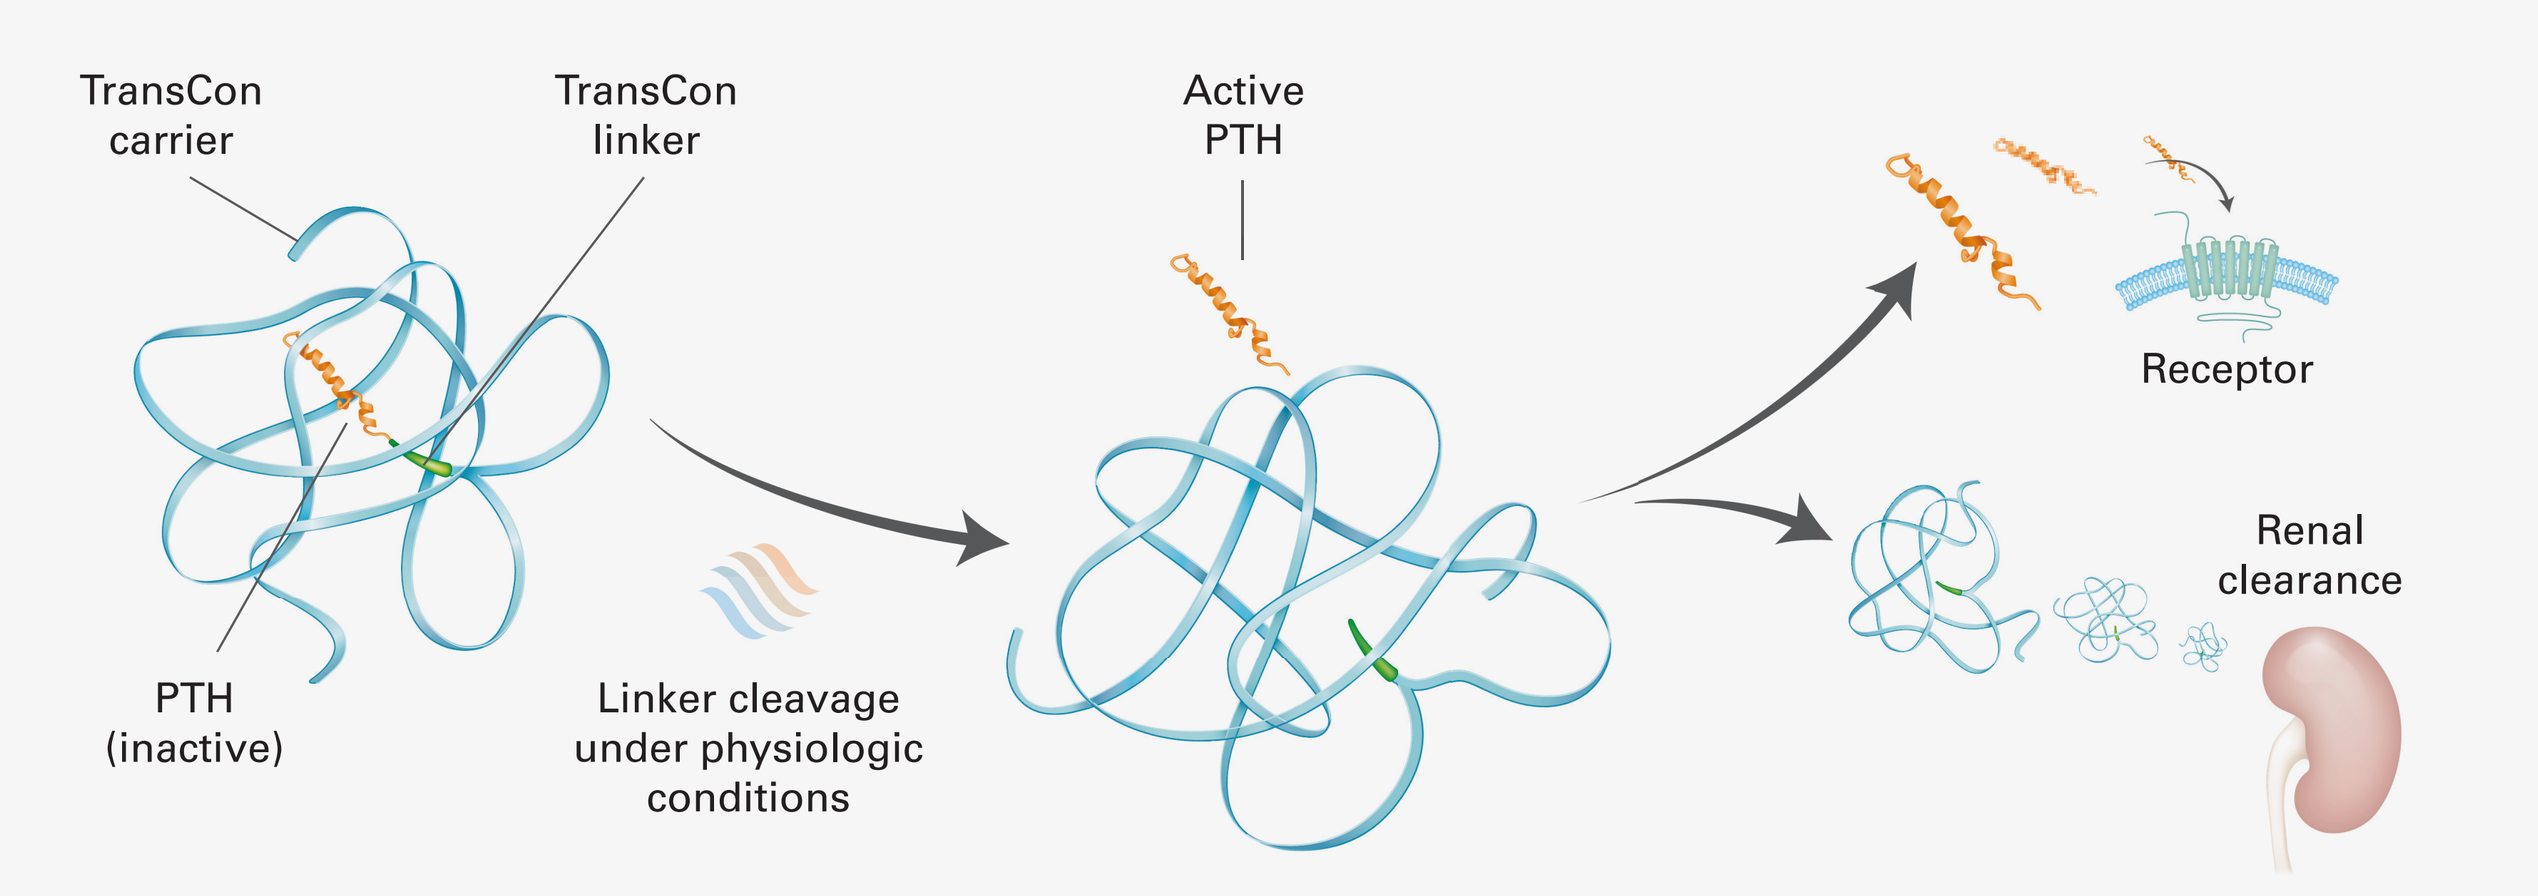

TransCon carrier
TransCon linker
Active PTH
PTH (inactive)
Linker cleavage under physiologic conditions
Receptor
Renal clearance

- Palopegteriparatide is a prodrug of PTH (1-34), administered once daily, that provides active PTH within the physiological range for 24 hours per day.[8,9]
- Palopegteriparatide has received regulatory approval in the US,[a] EU,[b] Japan,[a] and several other countries based on the results of the PaTH Forward and PaTHway trials.

[a]Indicated for the treatment of hypoparathyroidism in adults.
[b]Indicated for the treatment of adults with chronic hypoparathyroidism.

Methods

Figure 3. **Phase 3 PaTHway Trial Design (NCT04701203)**

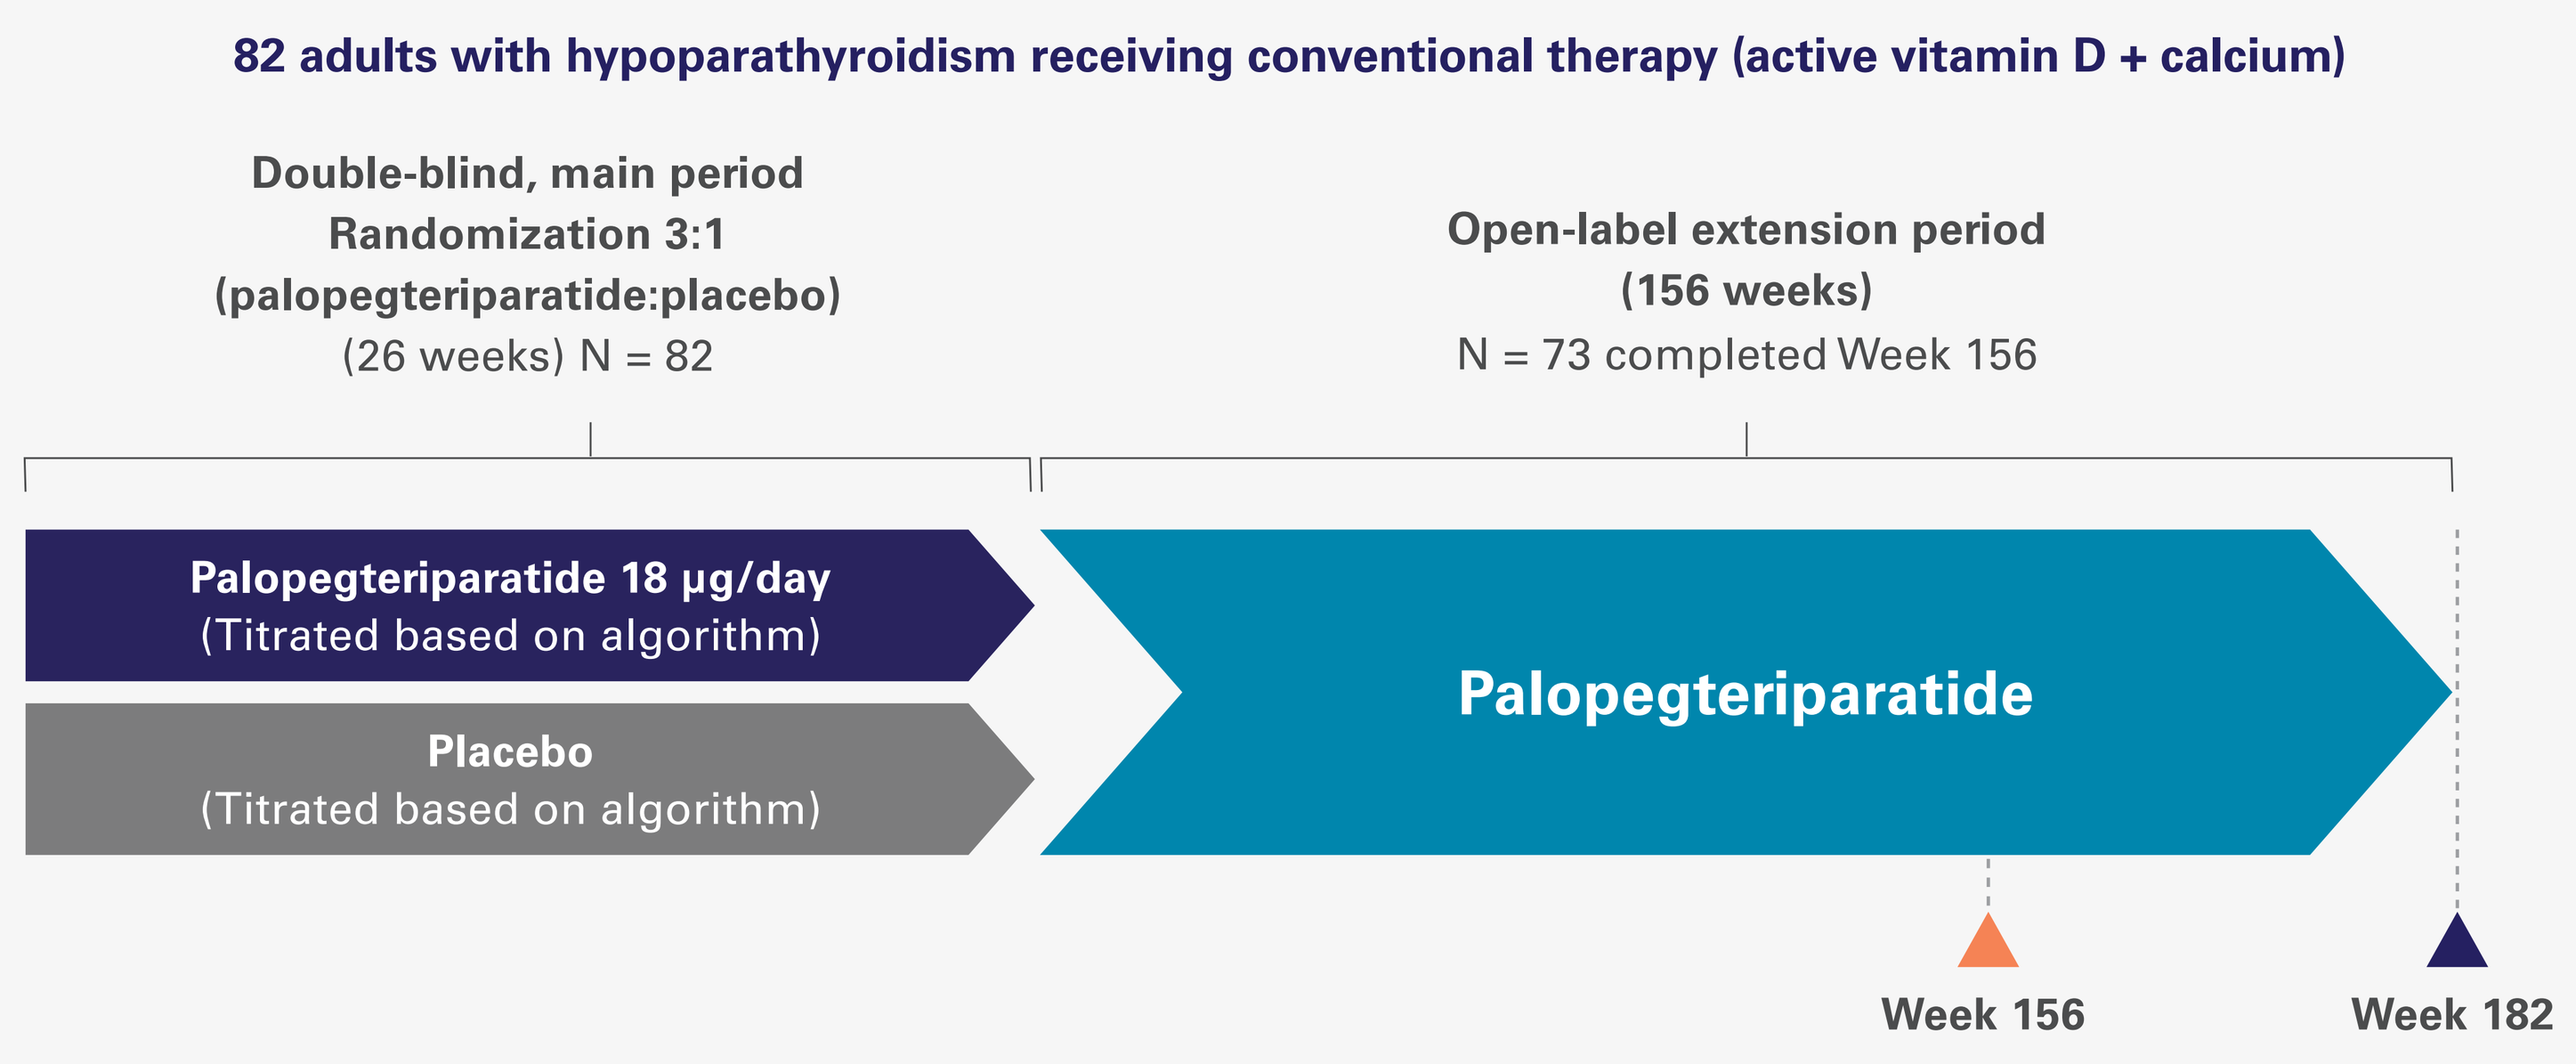

82 adults with hypoparathyroidism receiving conventional therapy (active vitamin D + calcium)
Double-blind, main period
Randomization 3:1
(palopegteriparatide:placebo)
(26 weeks) N = 82
Open-label extension period
(156 weeks)
N = 73 completed Week 156
Palopegteriparatide 18 µg/day
(Titrated based on algorithm)
Placebo
(Titrated based on algorithm)
Palopegteriparatide
Week 156
Week 182

Key efficacy and safety endpoints

- Levels of serum calcium (normocalcemia)
- Independence from conventional therapy (defined as taking no active vitamin D and ≤ 600 mg/day elemental calcium)
- Incidence of AEs, SAEs, TEAEs

Patient-reported outcomes assessments

- HPES-Symptom physical and cognitive domain scores and HPES-Impact physical functioning and daily life domain scores
- 36-Item short form survey (SF-36) scores
- **Work Limitations Questionnaire-25**

- At Week 156, 96% of PaTHway trial participants achieved independence from conventional therapy (no active vitamin D and ≤ 600 mg/day of calcium), and 88% achieved normal albumin-adjusted serum calcium levels (8.3-10.6 mg/dL).
- Palopegteriparatide was well-tolerated in the PaTHway trial, with most treatment-related adverse events (AEs) being mild to moderate in severity.
- This analysis assessed the impact of palopegteriparatide on hypoparathyroidism-related work limitations in the PaTHway trial.

Work Limitations Questionnaire (WLQ)-25

Purpose of the WLQ

- The WLQ is a validated, self-reported instrument designed to quantify the **degree to which health problems interfere with job performance and productivity** in employed individuals.[10]
- The WLQ has strong reliability, differentiating between healthy and condition-affected employee groups.[10]
- It is widely used in clinical trials, occupational health research, and economic evaluations to assess on-the-job work functioning.[11,12]

Scoring approach[13]

- Each item is scored from **0 to 100**, where:
 - **0** = Limited none of the time
 - **100** = Limited all of the time
- Domain scores are calculated as the **mean of item scores** within that domain.
- **Higher scores** = greater limitation

WLQ-25 structure[14]

- The WLQ-25 consists of 25 items, grouped into 4 empirically derived domains that reflect different aspects of work functioning **(Table 1).**[10,14]

Table 1. **WLQ-25 Structure**[a]

Domain	Focus Area	Items
Time Management	Difficulties handling scheduling, being on time, staying focused	5
Physical Demands	Challenges with physical aspects of work (lifting, movement)	6
Mental-Interpersonal	Difficulty with concentration, problem-solving, social demands	9
Output Demands	Difficulties with quantity, quality, and speed of work	5

[a]Each item asks respondents to report how often, in the **past two weeks**, health problems limited their ability to meet work demands.[14]

Results

Table 2. **Baseline Demographics and Disease Characteristics**

Variable		Palopegteriparatide (N = 61)	Placebo (N = 21)
Age (years), mean (SD)		49 (13)	47 (11)
Sex, n (%) female		46 (75)	18 (86)
	Postmenopausal	19 (41)	3 (17)
Race, n (%) White		57 (93)	19 (90)
Geographic region, n (%)	North America	39 (64)	12 (57)
	Europe	22 (36)	9 (43)
Cause of hypoparathyroidism, n (%)	Acquired from neck surgery	52 (85)	18 (86)
	Genetic causes[a]	5 (8)	1 (5)
	Primary idiopathic disease	4 (7)	2 (10)
Duration of hypoparathyroidism (years), mean (SD)		12 (11.4)	11.1 (8.5)
Conventional therapy, mean TDD	Calcium (mg)	1748 (904)	2105 (1383)
	Calcitriol (µg)	0.76 (0.34), n = 53	0.69 (0.33), n = 17
	Alfacalcidol (µg)	2.5 (0.9), n = 8	2.0 (0.4), n = 4

[a]One participant in each arm had hypoparathyroidism, sensorineural deafness, and renal disease (HDR syndrome). In the palopegteriparatide arm, 2 participants had autoimmune polyglandular syndrome type 1 (APS-1), 1 participant had autosomal dominant hypocalcemia type 1 (ADH1), and 1 participant had DiGeorge syndrome. One participant previously reported as having idiopathic hypoparathyroidism was subsequently confirmed to have HDR syndrome. Percentages were calculated based on ITT population; the percentage of postmenopausal participants based on number of female patients.

Figure 4. **Time Management Scale**[a]

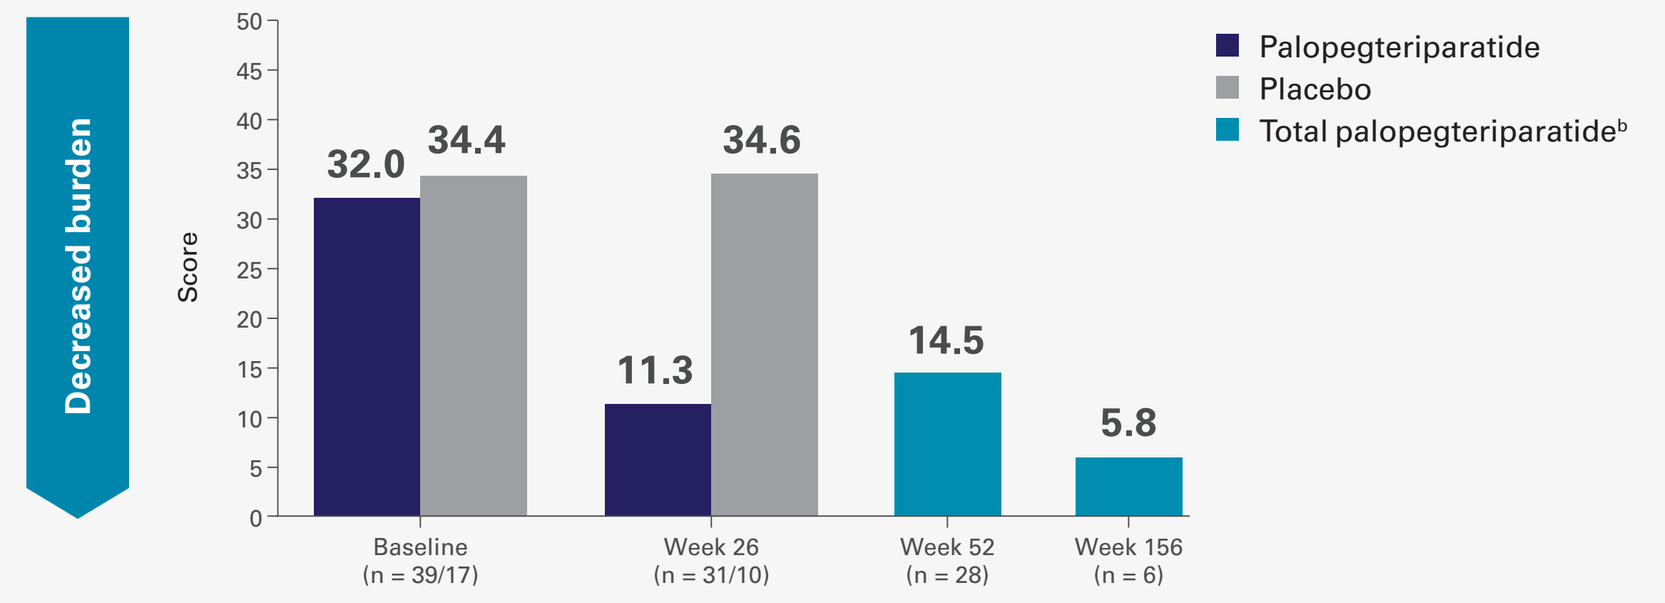

Decreased burden
Score
Palopegteriparatide
Placebo
Total palopegteriparatide[b]
32.0
34.4
11.3
34.6
14.5
5.8
Baseline (n = 39/17)
Week 26 (n = 31/10)
Week 52 (n = 28)
Week 156 (n = 6)

WLQ Time Management scores over time. Improvements observed with palopegteriparatide at Week 26 were maintained through long-term follow-up, including among participants who crossed over from placebo during the open-label extension.

[a]Greater scores correspond with more self-reported difficulties at work.
[b]Participants randomized to placebo crossed over to palopegteriparatide after Week 26.

KEY INTERPRETATION

Baseline assessment
- Participants in the palopegteriparatide and placebo groups had comparable mean baseline limitation scores (32.0 and 34.4, respectively).

Double-blind period (Baseline – Week 26)
- At the end of the double-blind period (Week 26), participants receiving palopegteriparatide experienced rapid improvement, with the mean score decreasing to 11.3 (indicating reduced work limitation for Time Management).
- No improvement was observed for participants in the placebo group.
- *P* value for treatment arm comparison from Week 0 to Week 26: < 0.0001

Open-label extension (Week 26 – Week 156)
- In the open-label extension period, improvements in Time Management limitations were maintained among participants treated with palopegteriparatide through Week 156.

Figure 5. **Physical Demands Scale**[a]

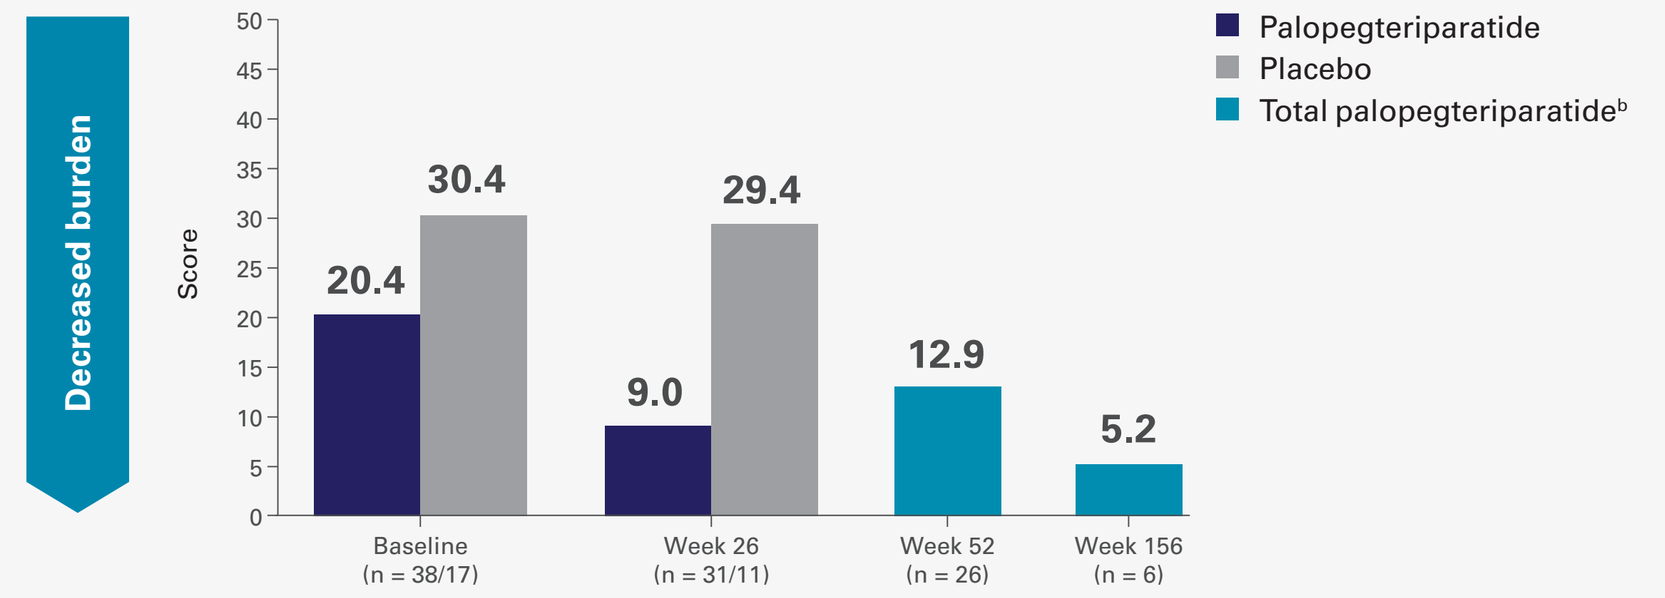

Decreased burden
Score
Palopegteriparatide
Placebo
Total palopegteriparatide[b]
20.4
30.4
9.0
29.4
12.9
5.2
Baseline (n = 38/17)
Week 26 (n = 31/11)
Week 52 (n = 26)
Week 156 (n = 6)

WLQ Physical Demands scores over time. Palopegteriparatide treatment resulted in a quick improvement by Week 26 with sustained improvement through Week 156, whereas placebo showed no improvement during the double-blind period.

[a]Greater scores correspond with more self-reported difficulties at work.
[b]Participants randomized to placebo crossed over to palopegteriparatide after Week 26.

KEY INTERPRETATION

Baseline assessment
- A slight imbalance was observed at baseline, with participants randomized to placebo exhibiting greater Physical Demands limitations.

Double-blind period (Baseline – Week 26)
- Participants receiving palopegteriparatide showed rapid improvement, with scores dropping to a mean value of 9.0 at Week 26.
- Scores in the group randomized to placebo remained high with a mean value of 29.4 at Week 26
- *P* value for treatment arm comparison from Week 0 to Week 26: 0.0008

Open-label extension (Week 26 – Week 156)
- During the open-label extension, participants continued to demonstrate reduction in Physical Demands limitations, remaining substantially improved through Week 156.

Figure 6. **Mental-Interpersonal Scale**[a]

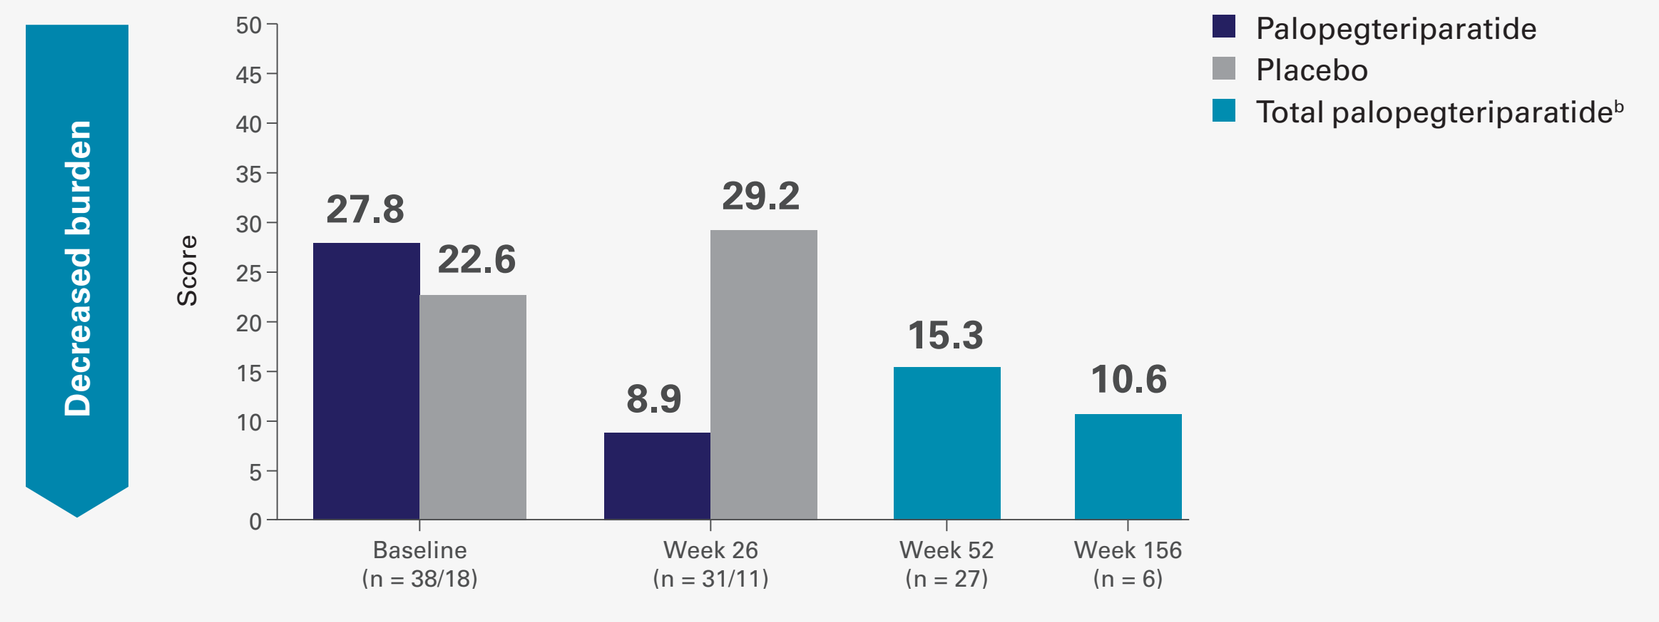

Decreased burden
Score
Palopegteriparatide
Placebo
Total palopegteriparatide[b]
27.8
22.6
8.9
29.2
15.3
10.6
Baseline (n = 38/18)
Week 26 (n = 31/11)
Week 52 (n = 27)
Week 156 (n = 6)

WLQ Mental-Interpersonal scores over time. Palopegteriparatide treatment resulted in improvement by Week 26 with continued improvement through Week 156, whereas placebo slightly worsened during the double-blind period.

[a]Greater scores correspond with more self-reported difficulties at work.
[b]Participants randomized to placebo crossed over to palopegteriparatide after Week 26.

KEY INTERPRETATION

Baseline assessment
- Both groups started with Mental-Interpersonal limitations including challenges with concentration, problem-solving, and social demands.
- Participants randomized to palopegteriparatide had slightly worse baseline limitation (mean score 27.8) compared to those randomized to placebo (mean score 22.6).

Double-blind period (Baseline – Week 26)
- At the end of the double-blind period (Week 26), participants treated with palopegteriparatide showed a rapid improvement, indicating reduced mental-interpersonal work limitations.
- In contrast, participants randomized to placebo slightly worsened during the double-blind period, with the mean score increasing to 29.2.
- *P* value for treatment arm comparison from Week 0 to Week 26: < 0.0001

Open-label extension (Week 26 – Week 156)
- In the open-label extension period, participants demonstrated continued improvement in Mental-Interpersonal limitations.

Figure 7. **Output Demands Scale**[a]

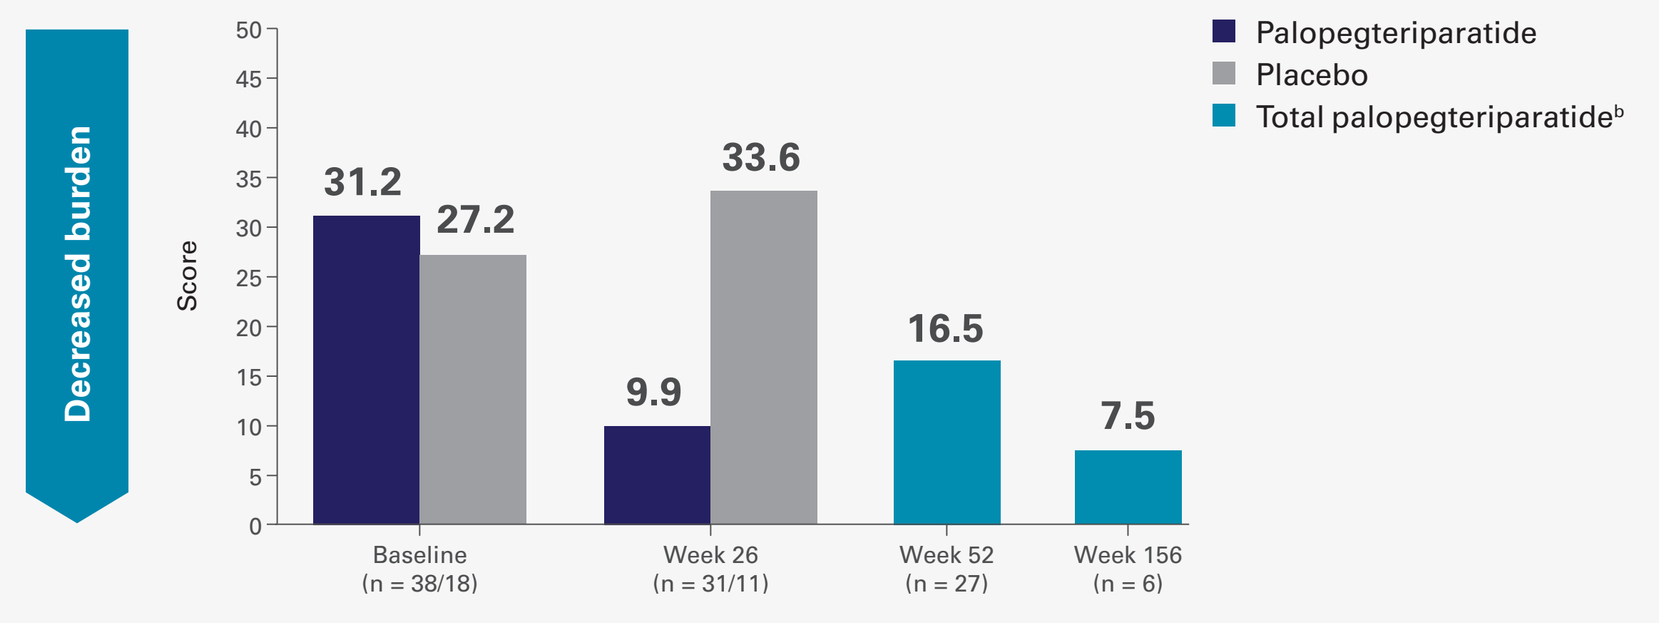

Decreased burden
Score
Palopegteriparatide
Placebo
Total palopegteriparatide[b]
31.2
27.2
9.9
33.6
16.5
7.5
Baseline (n = 38/18)
Week 26 (n = 31/11)
Week 52 (n = 27)
Week 156 (n = 6)

WLQ Output Demands scores over time. Palopegteriparatide treatment resulted in improvement by Week 26 with continued improvement through Week 156, whereas participants randomized to placebo slightly worsened through the double-blind period.

[a]Greater scores correspond with more self-reported difficulties at work.
[b]Participants randomized to placebo crossed over to palopegteriparatide after Week 26.

KEY INTERPRETATION

Baseline Assessment
- Participants in the groups randomized to palopegteriparatide and placebo had mean baseline scores of 31.2 and 27.2, respectively, indicating some degree of limitation in work quantity and quality.

Double-blind period (Baseline – Week 26)
- Participants randomized to palopegteriparatide showed rapid improvement, with a mean score of 9.9 by the end of the double-blind period (Week 26), whereas scores slightly worsened for participants randomized to placebo (rising to a mean value of 33.6).
- *P* value for treatment arm comparison from Week 0 to Week 26: 0.0007

Open-label extension (Week 26 – Week 156)
- By Week 156 during the open-label extension, participants showed further improvement in Output Demands.

Figure 8. **At-Work Productivity Improvement in Percentage**[a]

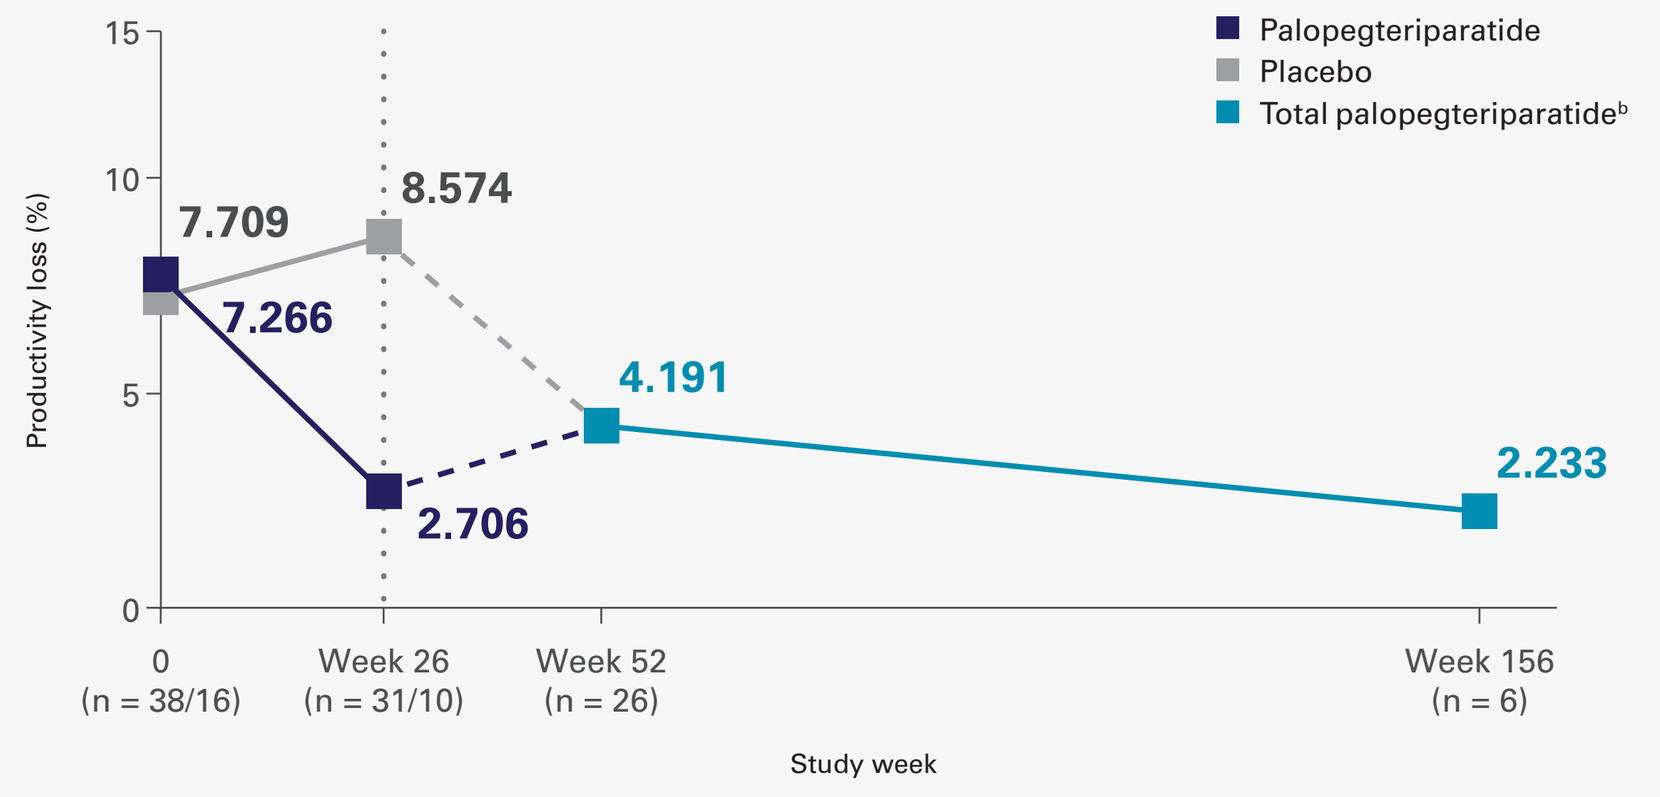

Palopegteriparatide
Placebo
Total palopegteriparatide[b]
Productivity loss (%)
7.709
7.266
8.574
2.706
4.191
2.233
0 (n = 38/16)
Week 26 (n = 31/10)
Week 52 (n = 26)
Week 156 (n = 6)
Study week

[a]Higher percentage corresponds with higher work productivity loss. WLQ At-Work Productivity Loss is expressed as the mean percentage of productivity loss over the past 2 weeks, derived from the four WLQ domain scores.
[b]Participants randomized to placebo crossed over to palopegteriparatide after Week 26.

KEY INTERPRETATION

Baseline assessment
- At baseline, both groups demonstrated similar levels of at-work productivity loss (approximately 7-8%).

Double-blind period (Baseline – Week 26)
- By the end of the double-blind period (Week 26), a substantial improvement was observed among participants treated with palopegteriparatide, with mean productivity loss improving to 2.7%, representing an approximately 65% reduction from baseline.
- No improvement was observed in the placebo group.
- *P* value for treatment arm comparison from Week 0 to Week 26: 0.0001

Open-label extension (Week 26 – Week 156)
- During the open-label extension, at-work productivity loss remained substantially improved among participants receiving palopegteriparatide, with the lowest levels observed at Week 156.

Abbreviations

AE = adverse event; **DBP** = double-blind period; **EU** = European Union; **HEOR** = health economics and outcomes research; **HPES** = hypoparathyroidism patient experience scales; **n** = number; **PTH** = parathyroid hormone; **SAE** = serious adverse event; **SD** = standard deviation; **SF-36** = 36-item short form survey; **TDD** = total daily dose; **TEAE** = treatment emergent adverse event; **TransCon** = transient conjugation; **US** = United States; **Wk** = week; **WLQ** = Work Limitations Questionnaire

References

1. Khan AA, et al. *J Bone Miner Res.* 2022;37:2568-2585. **2.** Shoback DM, et al. *J Clin Endocrinol Metab.* 2016;101(6):2300-2312. **3.** Bilezikian JP, et al. *J Clin Endocrinol Metab.* 2016;101(6):2313-2324. **4.** Mannstadt M, et al. *Nat Rev Dis Primers.* 2017;3:17055. **5.** Brandi ML, et al. *J Clin Endocrinol Metab.* 2016;101(6):2273-2283. **6.** Khan AA, et al. *Eur J Endocrinol.* 2019;180(3):R33-63. **7.** Shoback D. *N Engl J Med.* 2008;359:391-403. **8.** Karpf DB, et al. *J Bone Miner Res.* 2020;35(8):1430-1440. **9.** Holten-Andersen L, et al. *J Bone Miner Res.* 2019;34(11):2075-2086. **10.** Munir F. *Occup Med (Lond.).* 2008;58(4):310-311. **11.** Lerner D, et al. *J Occup Environ Med.* 2009;51(5):515-524. **12.** Cangelosi MJ, et al. *J Occup Environ Med.* 2012;54(7):826-833. **13.** Allaire SH. *Arthritis Rheum.* 2003;49(5 Suppl): S85-S89. **14.** Lerner D, et al. *QoL Newsletter.* 2002;28:9-10.

Disclosures and Funding

Ascendis Pharma and the authors thank the participants, study sites, and investigators who participated in this clinical trial.

Ascendis Pharma Bone Diseases A/S funded this trial and participated in the trial design, research, analysis, data collection, interpretation of the data, and the review and approval of the publication. All authors had access to relevant data and participated in the drafting, review, and approval of this publication. No honoraria or payments were made for authorship. Medical writing support was provided by Robert Geist, MD, of Ascendis Pharma.

Financial arrangements of the authors with companies whose products may be related to this presentation are listed as declared by the authors: **WN, CZ, ARS, CTS,** and **ADS:** Full-time employees of Ascendis Pharma.

Ascendis®

MED-US-TC-PTH-2600055
07/2026