Sustained Improvement in Renal Function With Palopegteriparatide in Adults With Chronic Hypoparathyroidism: Three-Year Results From the PaTH Forward and PaTHway Trials

Elvira Gosmanova,[1] Peter Schwarz,[2] Lars Rejnmark,[3] Aliya A Khan,[4] Bart Clarke,[5] Filomena Cetani,[6] Stuart M Sprague,[7] Dolores M Shoback,[8] Lynn Kohlmeier,[9] Mishaela R Rubin,[10] Andrea Palermo,[11] Claudia Gagnon,[12] Elena Tsourdi,[13] Carol Zhao,[14] Michael A Makara,[14] Jenny Ukena,[14] Bryant Lai,[14] Christopher T Sibley,[14] Aimee D Shu[14]

[1]Albany Medical College and Albany VAMC, Albany, NY, USA; [2]Rigshospitalet, Copenhagen, Denmark; [3]Aarhus University Hospital, Aarhus, Denmark; [4]McMaster University, Hamilton, ON, Canada; [5]Mayo Clinic, Rochester, MN, USA; [6]University of Pisa, Department of Clinical and Experimental Medicine, Endocrine Unit, Pisa, Italy; [7]NorthShore University Health System-University of Chicago Pritzker School of Medicine, Chicago, IL, USA; [8]University of California, San Francisco and VA Medical Center, San Francisco, CA, USA; [9]Spokane Osteoporosis and Endocrinology, Arthritis Northwest Research, Spokane, WA, USA; [10]Columbia University, New York, NY, USA; [11]Fondazione Policlinico Campus Bio-medico and Unit of Endocrinology and Diabetes, Campus Bio-medico University, Rome, Italy; [12]CHU de Québec-Université Laval Research Centre and Department of Medicine, Université Laval, Quebec City, QC, Canada; [13]Technische Universität Dresden Medical Center, Dresden, Germany; [14]Ascendis Pharma Inc, Palo Alto, CA, USA.

TH-PO0227

Conclusion

Clinically meaningful early improvements in eGFR with palopegteriparatide observed in PaTH Forward and PaTHway were sustained through Year 3.

The greatest increases in eGFR were observed in the first 6 months, with a continued upward trend thereafter.

Reductions in 24-hour urinary calcium excretion were observed and maintained within the normal range through Year 3.

Palopegteriparatide for the treatment of hypoparathyroidism maintained safety and efficacy through Year 3.

Background

- An intact PTH axis maintains normal serum and urine calcium and phosphate homeostasis[1,2,3]
- PTH is the primary regulator of calcium/phosphate balance, acting directly on bone and kidney, and indirectly on the intestine[4,5]
- Conventional therapy for hypoparathyroidism (active vitamin D (calcitriol) and oral calcium) aims to alleviate hypocalcemic symptoms but fails to restore normal PTH physiology[6]
- PTH replacement therapy for hypoparathyroidism should provide PTH levels within the physiological range and restore downstream calcitriol, promoting independence from conventional therapy and normalizing:
 - Serum and urine calcium and phosphate
 - Skeletal health
 - Quality of life

Figure 1: **PTH Therapy for Hypoparathyroidism[7]**

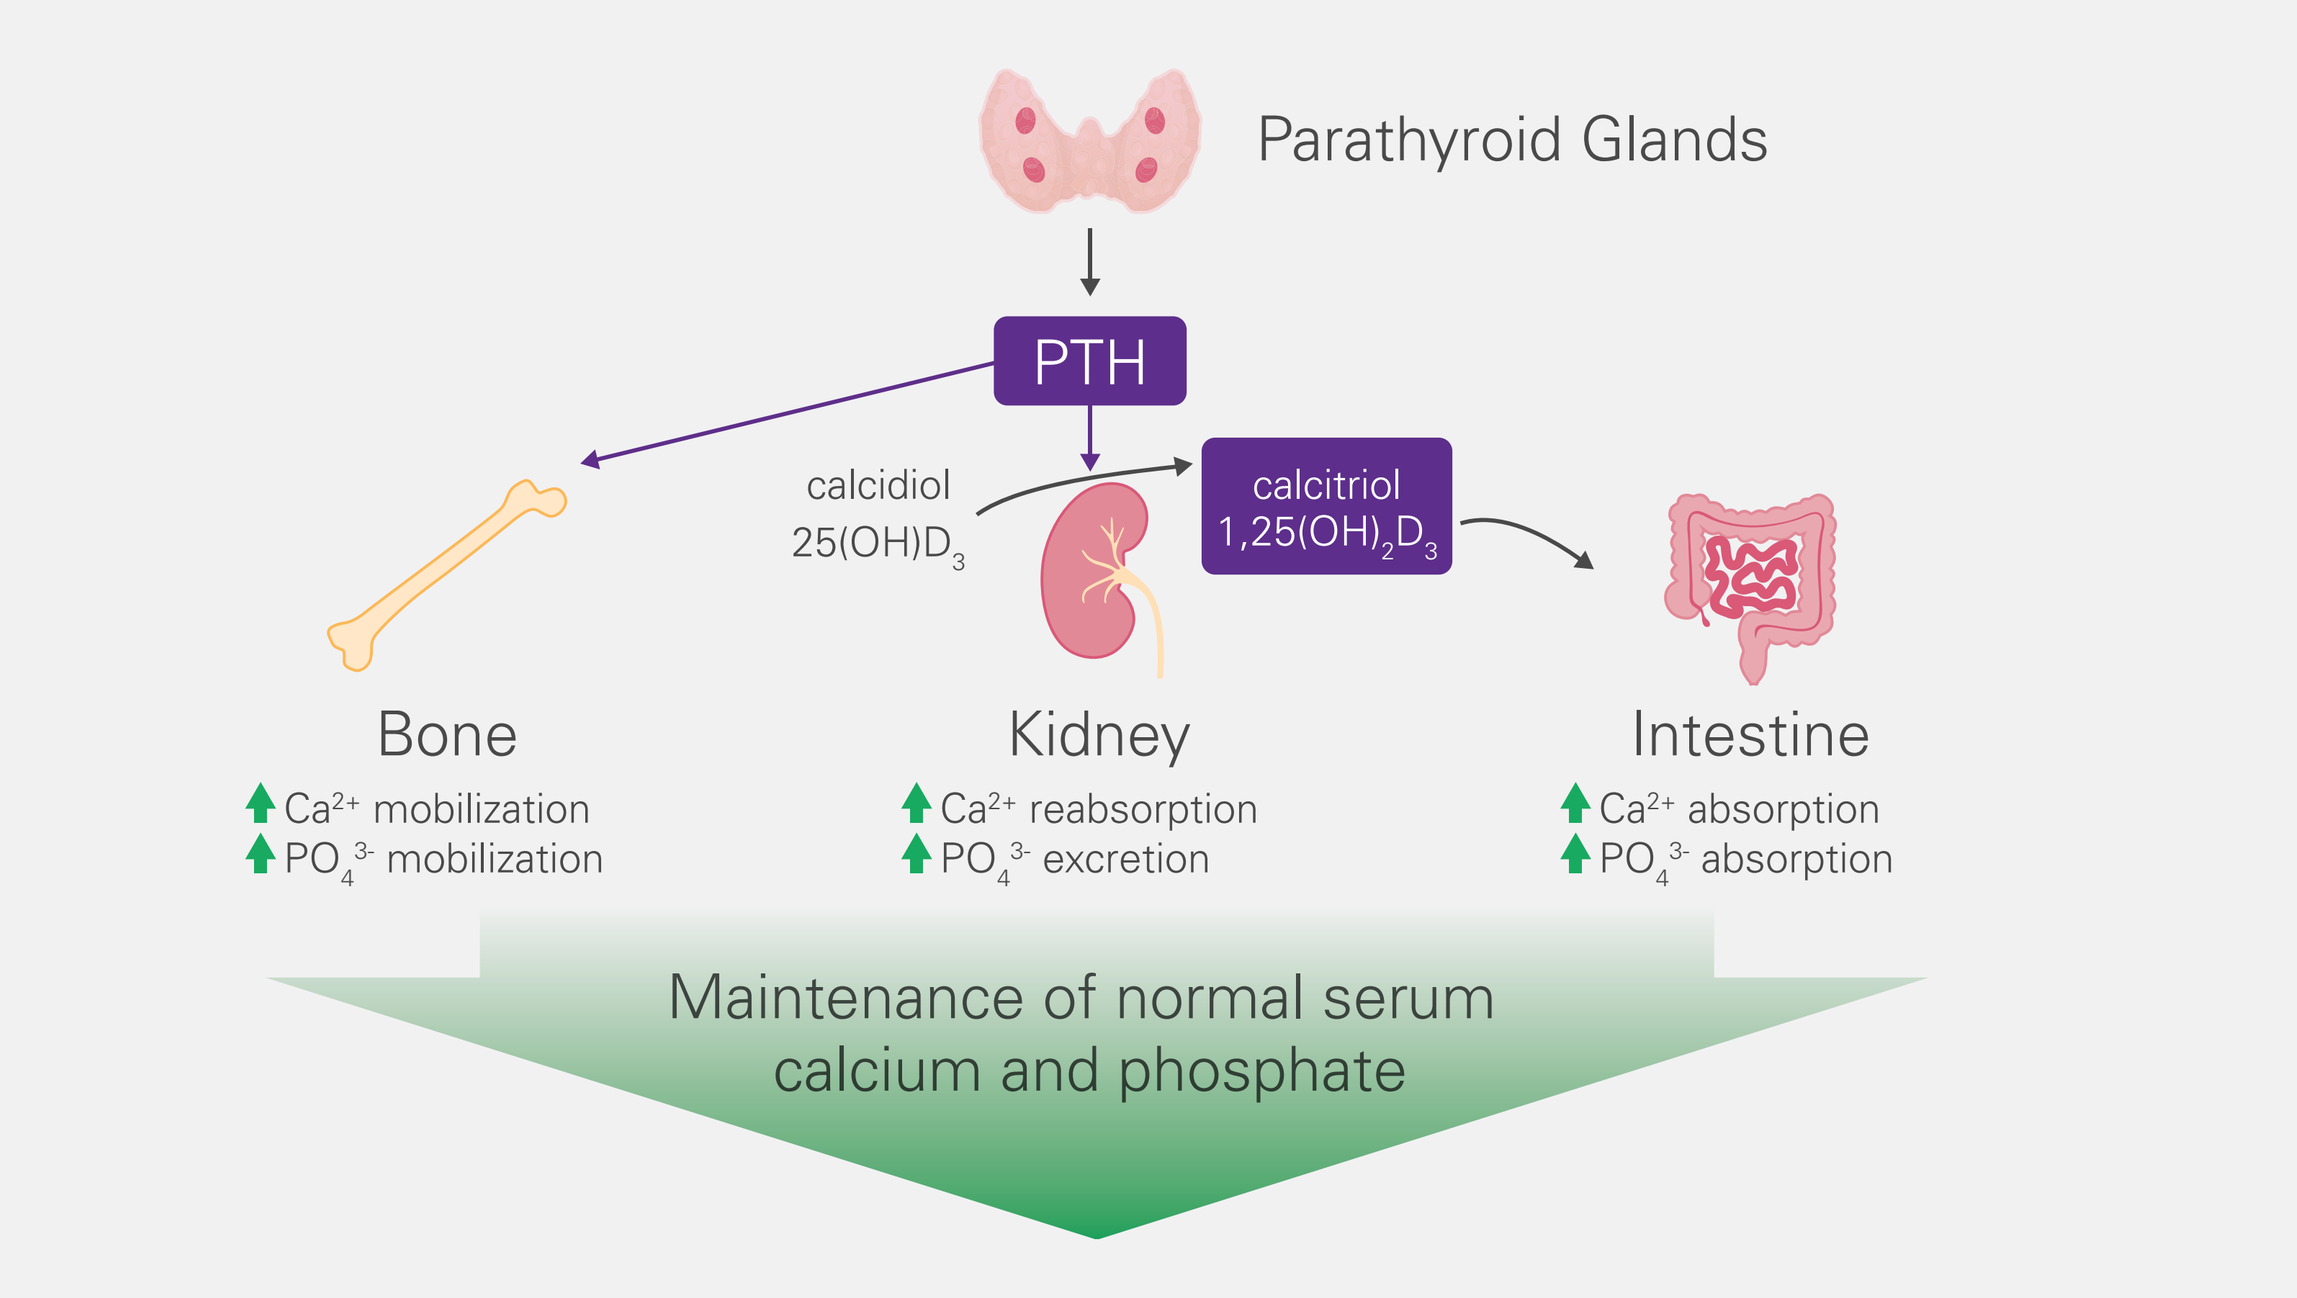
Parathyroid Glands
PTH
calcidiol
25(OH)D3
calcitriol
1,25(OH)2D3
Bone
Ca2+ mobilization
PO43- mobilization
Kidney
Ca2+ reabsorption
PO43- excretion
Intestine
Ca2+ absorption
PO43- absorption
Maintenance of normal serum calcium and phosphate

Background

Figure 2: **Palopegteriparatide (TransCon® PTH) Design**

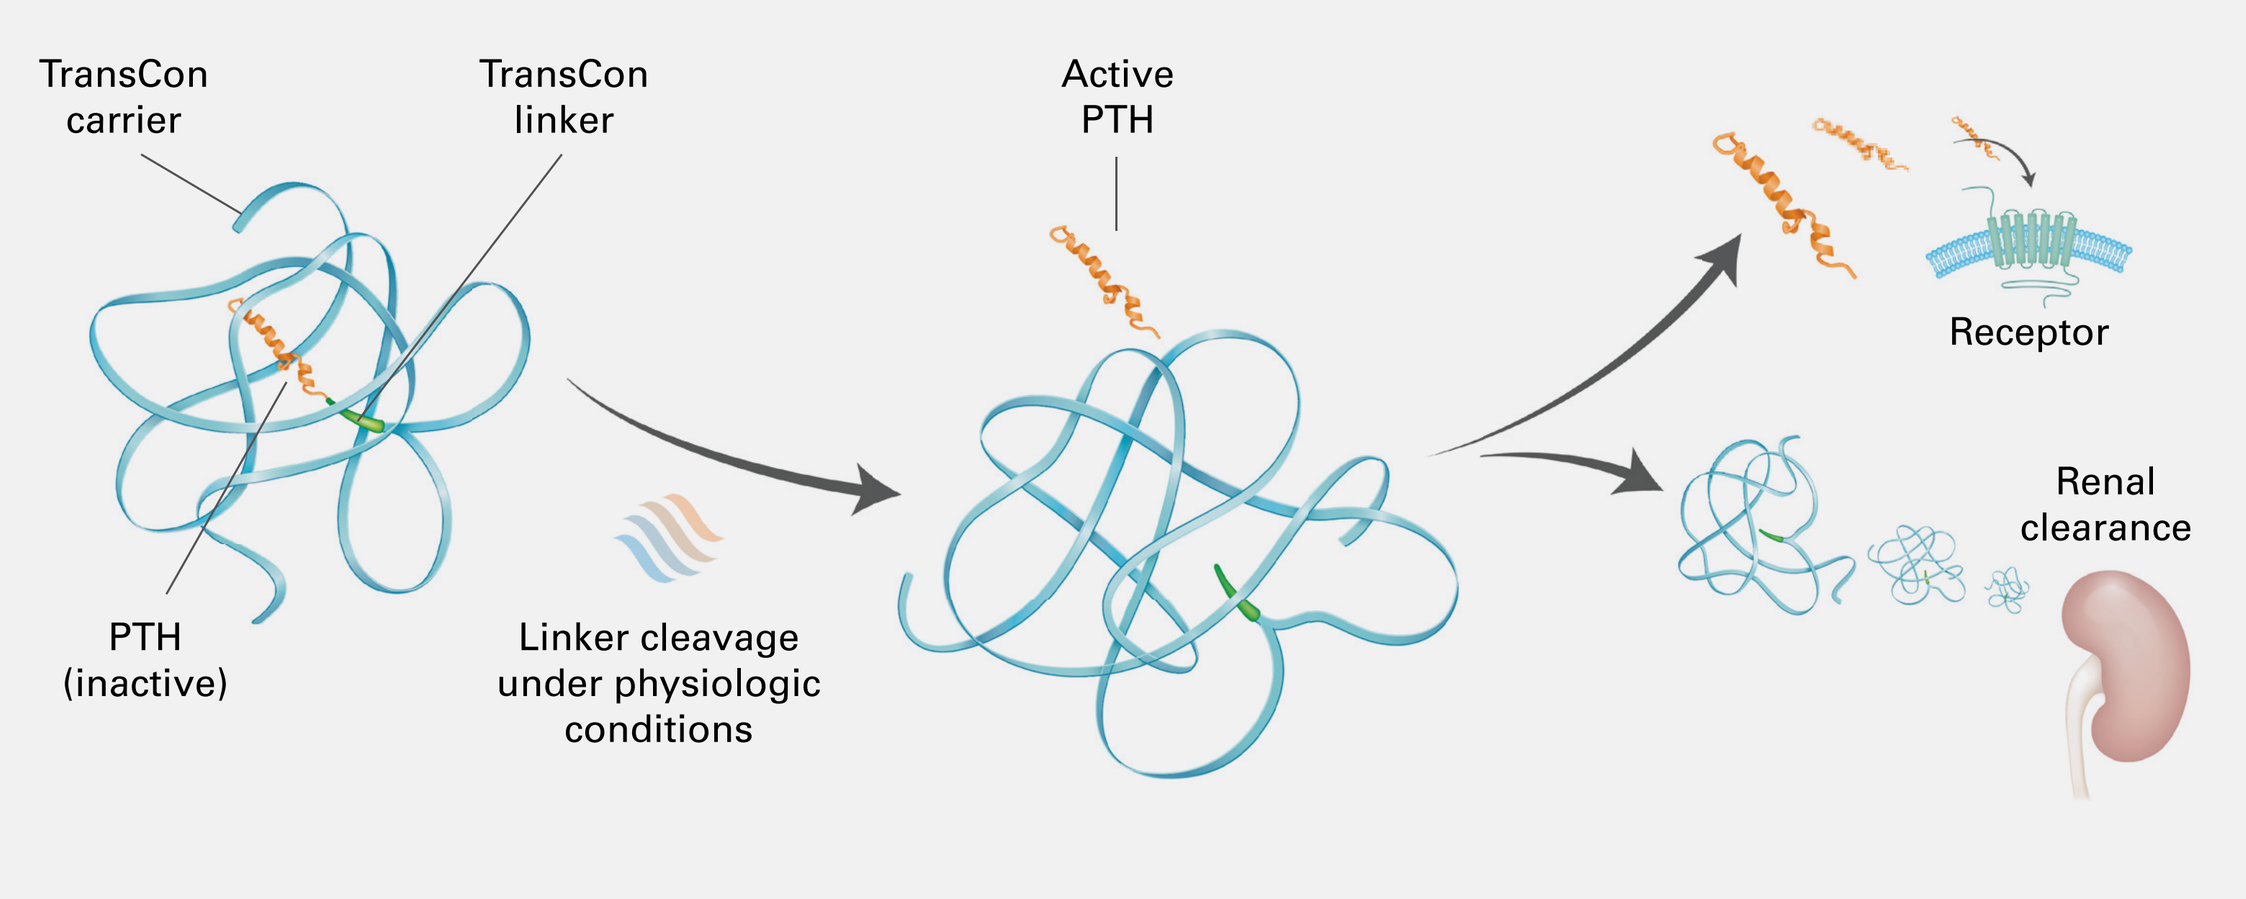
TransCon carrier
TransCon linker
Active PTH
Receptor
Renal clearance
PTH (inactive)
Linker cleavage under physiologic conditions

- Palopegteriparatide is a prodrug of PTH (1-34), administered once daily, that provides active PTH within the physiological range for 24 hours per day[8,9]
- Palopegteriparatide has received regulatory approval in the US, EU, and several other countries[a,b]

[a]Indicated for the treatment of hypoparathyroidism in adults.
[b]Indicated for the treatment of adults with chronic hypoparathyroidism.

Methods

Figure 3: **Schematics of Trial Designs Used in Combined Analysis**

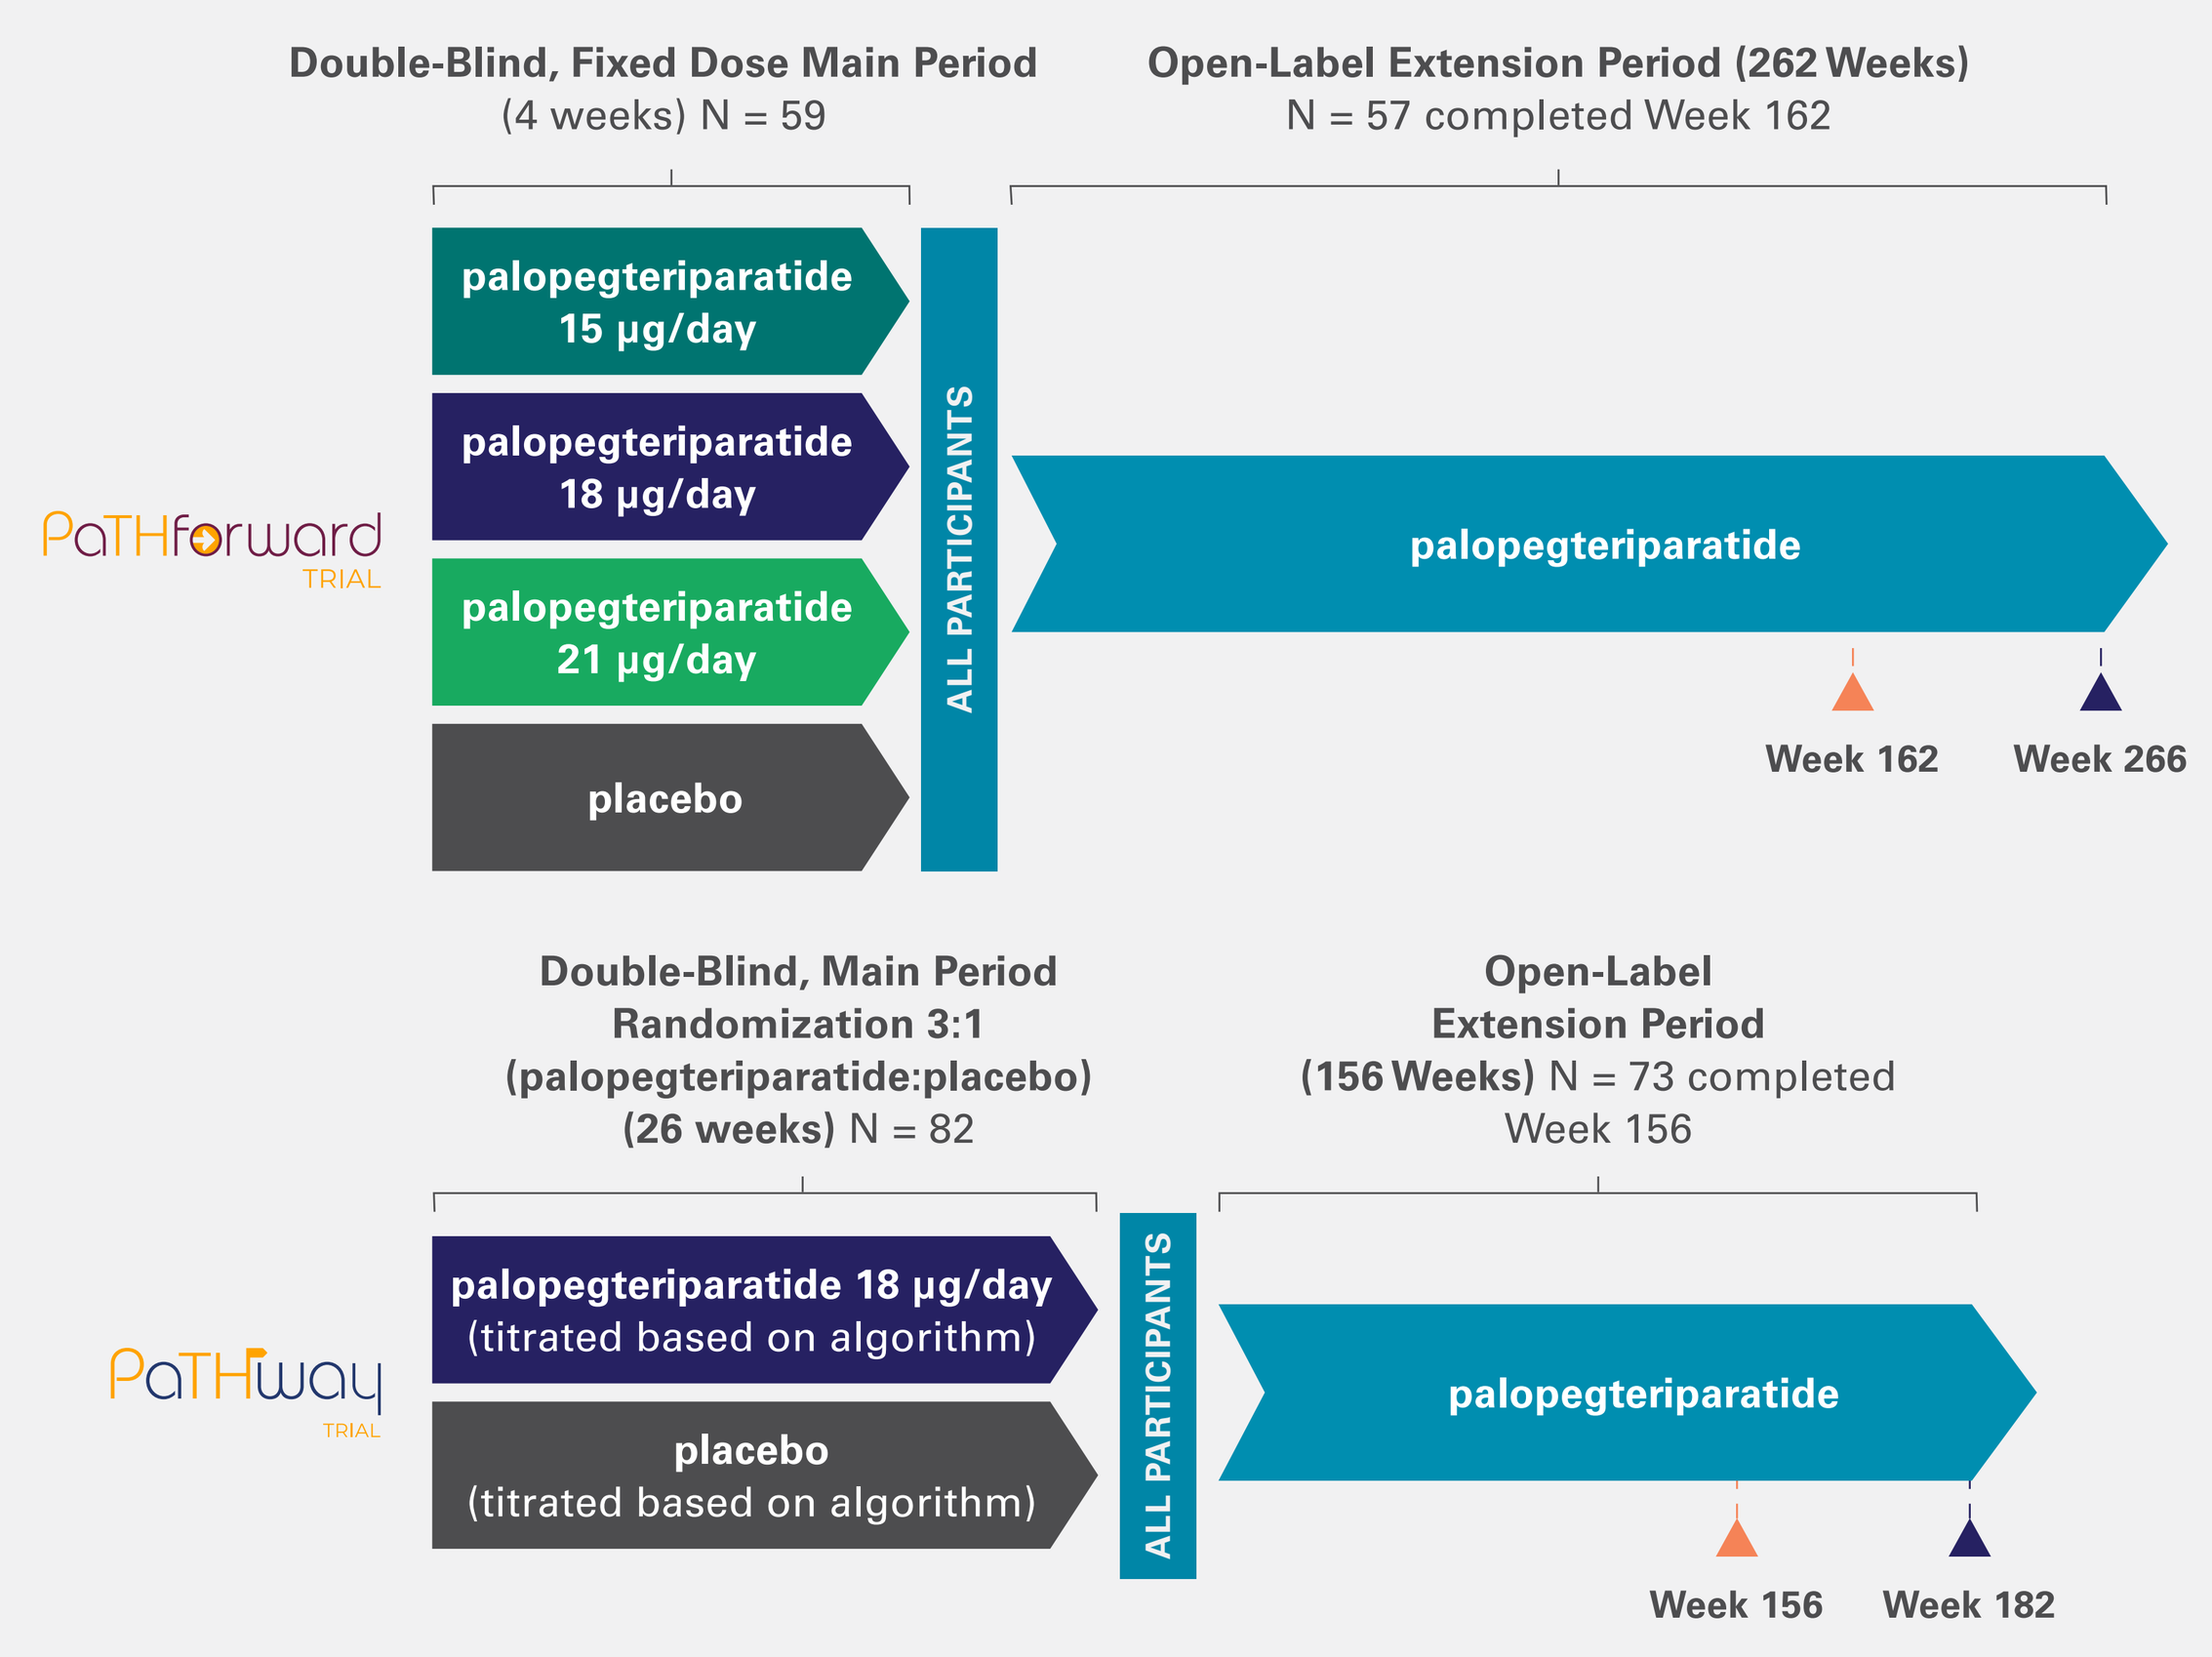
Double-Blind, Fixed Dose Main Period
(4 weeks) N = 59
Open-Label Extension Period (262 Weeks)
N = 57 completed Week 162
PaTHforward
TRIAL
palopegteriparatide 15 µg/day
palopegteriparatide 18 µg/day
palopegteriparatide 21 µg/day
placebo
ALL PARTICIPANTS
palopegteriparatide
Week 162
Week 266
Double-Blind, Main Period
Randomization 3:1
(palopegteriparatide:placebo)
(26 weeks) N = 82
Open-Label
Extension Period
(156 Weeks) N = 73 completed
Week 156
PaTHway
TRIAL
palopegteriparatide 18 µg/day
(titrated based on algorithm)
placebo
(titrated based on algorithm)
ALL PARTICIPANTS
palopegteriparatide
Week 156
Week 182

Key Efficacy and Safety Endpoints
- Levels of serum calcium (normocalcemia)
- Independence from conventional therapy (defined as taking no active vitamin D and ≤ 600 mg/day elemental calcium)
- Incidence of AEs, SAEs, TEAEs

Combined Renal Analysis Study
- Changes in eGFR from baseline
- Proportion of participants with increase in eGFR ≥ 5 mL/min/1.73 m²
- Levels of 24-hour urinary calcium

Note: Analyses include only participants who received at least one dose of palopegteriparatide. Analyses were performed by time since the first dose of active drug.

Methods

Table 1: **Baseline Demographics and Disease Characteristics**

Variable	Statistic or Category	Baseline GFR < 60 mL/min/1.73 m²	Baseline GFR ≥ 60 mL/min/1.73 m²	All Participants
		(N = 41)	(N = 98)	(N = 139)
Mean age, years (SD)		52 (12)	48 (12)	49 (12)
Sex, n (%)	Female	33 (80)	78 (80)	111 (80)
	Postmenopausal	13 (39)	26 (33)	39 (35)
Race, n (%)	White	39 (95)	89 (91)	128 (92)
Geographic region, n (%)	North America	28 (68)	60 (61)	88 (63)
	Europe	13 (32)	38 (39)	51 (37)
Mean eGFR, mL/min/1.73 m² (SD)*		51 (8)	76 (13)	69 (17)
Cause of hypoparathyroidism, n (%)	Acquired from neck surgery	33 (80)	83 (85)	116 (84)
	Autoimmune disease	0 (0)	2 (2)	2 (1)
	Idiopathic disease	6 (15)	11(11)	17(12)
	Intrinsic genetic defects of parathyroid glands	1 (2)	2 (2)	3 (2)
	Other	1 (2)	0 (0)	1 (1)
Median duration of hypoparathyroidism, years (range)		13 (1-56)	8 (1-39)	9 (1-56)
Baseline hypoparathyroidism supplements, mean TDD	Elemental calcium (mg)	2091.9 (n = 38)	1769.8 (n = 96)	1861.2 (n = 134)
	Calcitriol (µg)	0.8 (n = 33)	0.8 (n = 81)	0.8 (n = 114)
	Alfacalcidol (µg)	2.4 (n = 8)	2.4 (n = 17)	2.4 (n = 25)
	Cholecalciferol/ vitamin D3 (µg)	67.3 (n = 20)	64.2 (n = 59)	65.0 (n = 79)
	Magnesium (mg)	666.4 (n = 13)	587.6 (n = 37)	608.1 (n = 50)

Note: Numbers may not add to 100% due to rounding.
*Calculated according to the Modified Diet in Renal Disease Equation (MDRD): eGFR (mL/min/1.73 m²) = 175 × (serum creatinine mg/dL) -1.154 × (age) -0.203 × 0.742 [if female] × 1.212 [if Black race].

Results

Figure 4: **Improvements in eGFR: Combined Analysis**

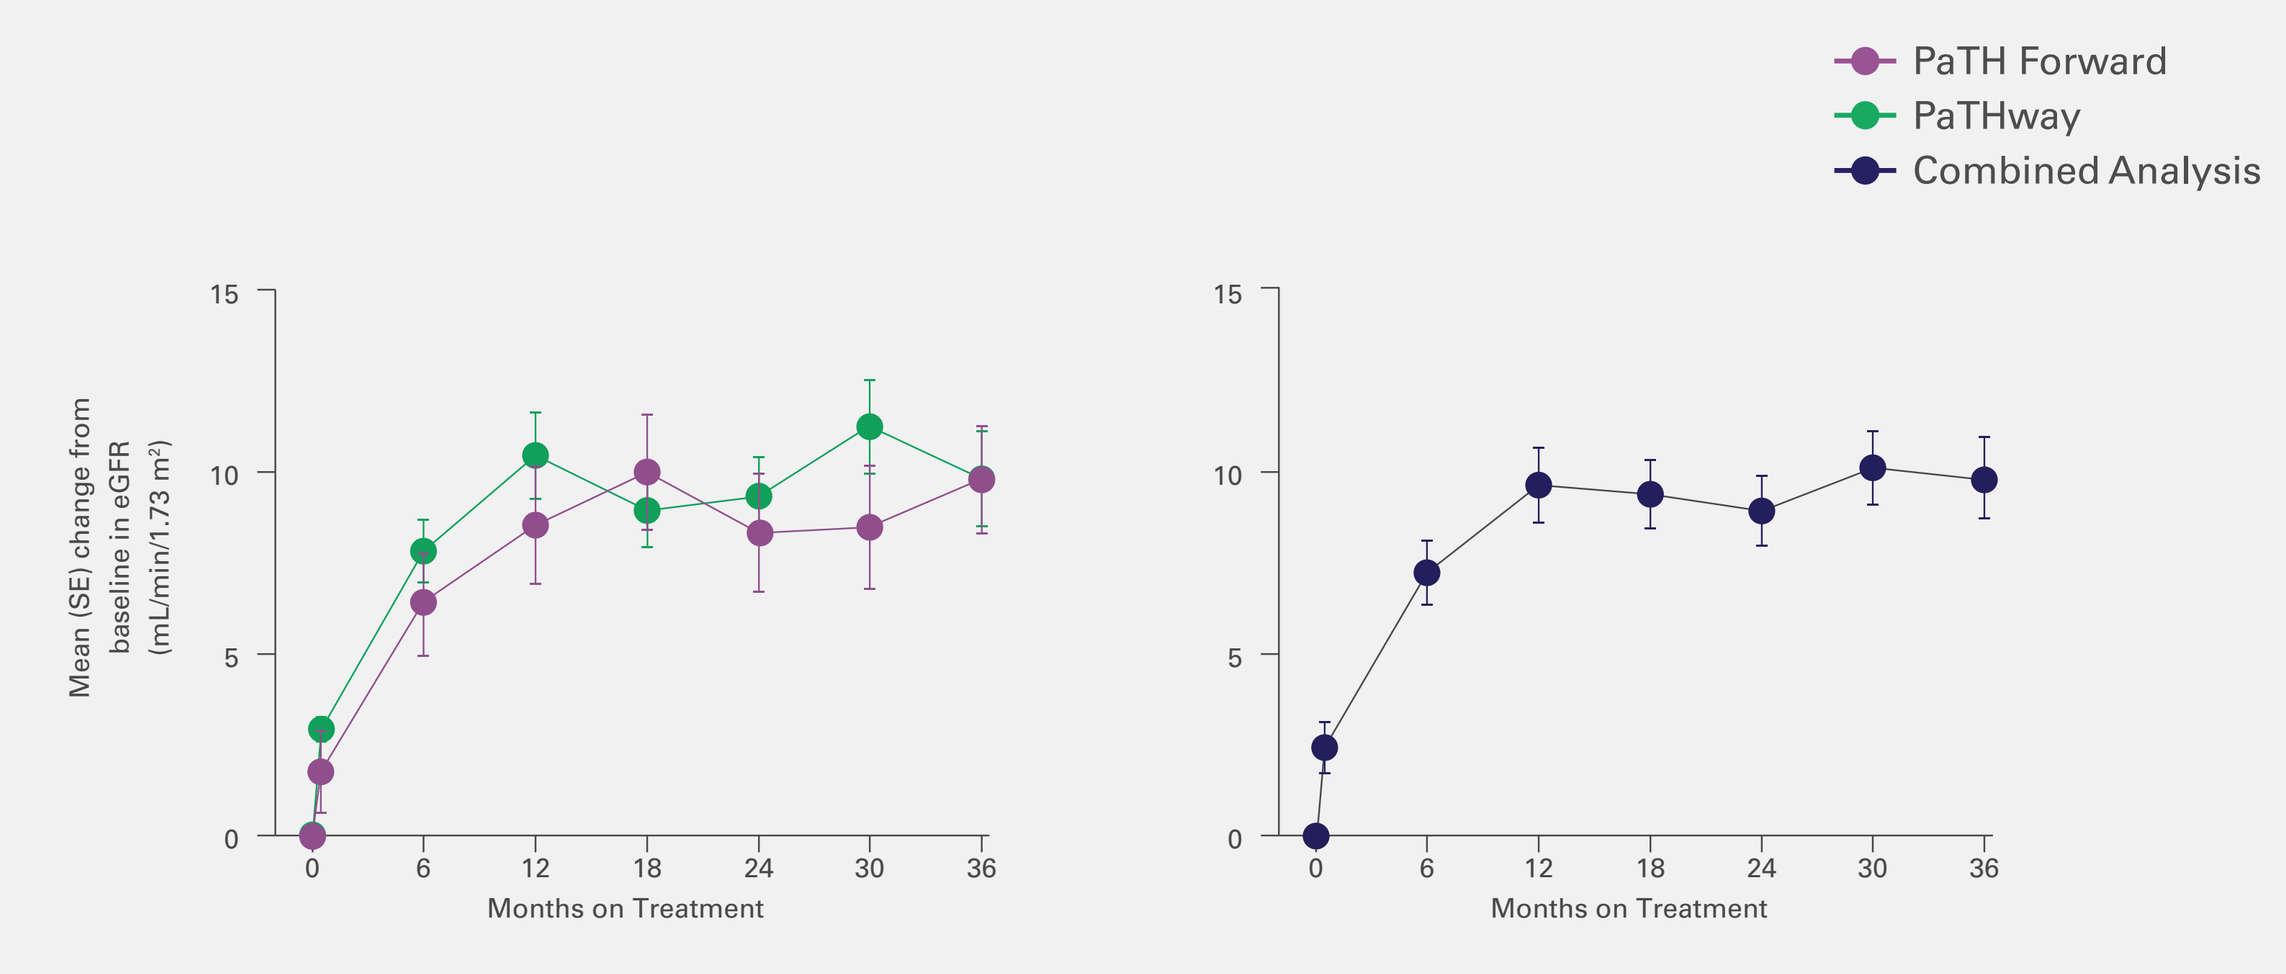
PaTH Forward
PaTHway
Combined Analysis
Mean (SE) change from baseline in eGFR (mL/min/1.73 m²)
Months on Treatment

PaTH Forward
eGFR Slope Change (mL/min/1.73 m²/year):
- 0-6 Months: **12.47**
- 6-36 Months: **0.99**

PaTHway
eGFR Slope Change (mL/min/1.73 m²/year)
- 0-6 Months: **15.64**
- 6-36 Months: **0.86**

Combined Analysis
eGFR Slope Change (mL/min/1.73 m²/year):
- 0-6 Months: **14.33**
- 6-36 Months: **0.89**

Results

Figure 5: **Sustained Improvements in eGFR From Baseline**

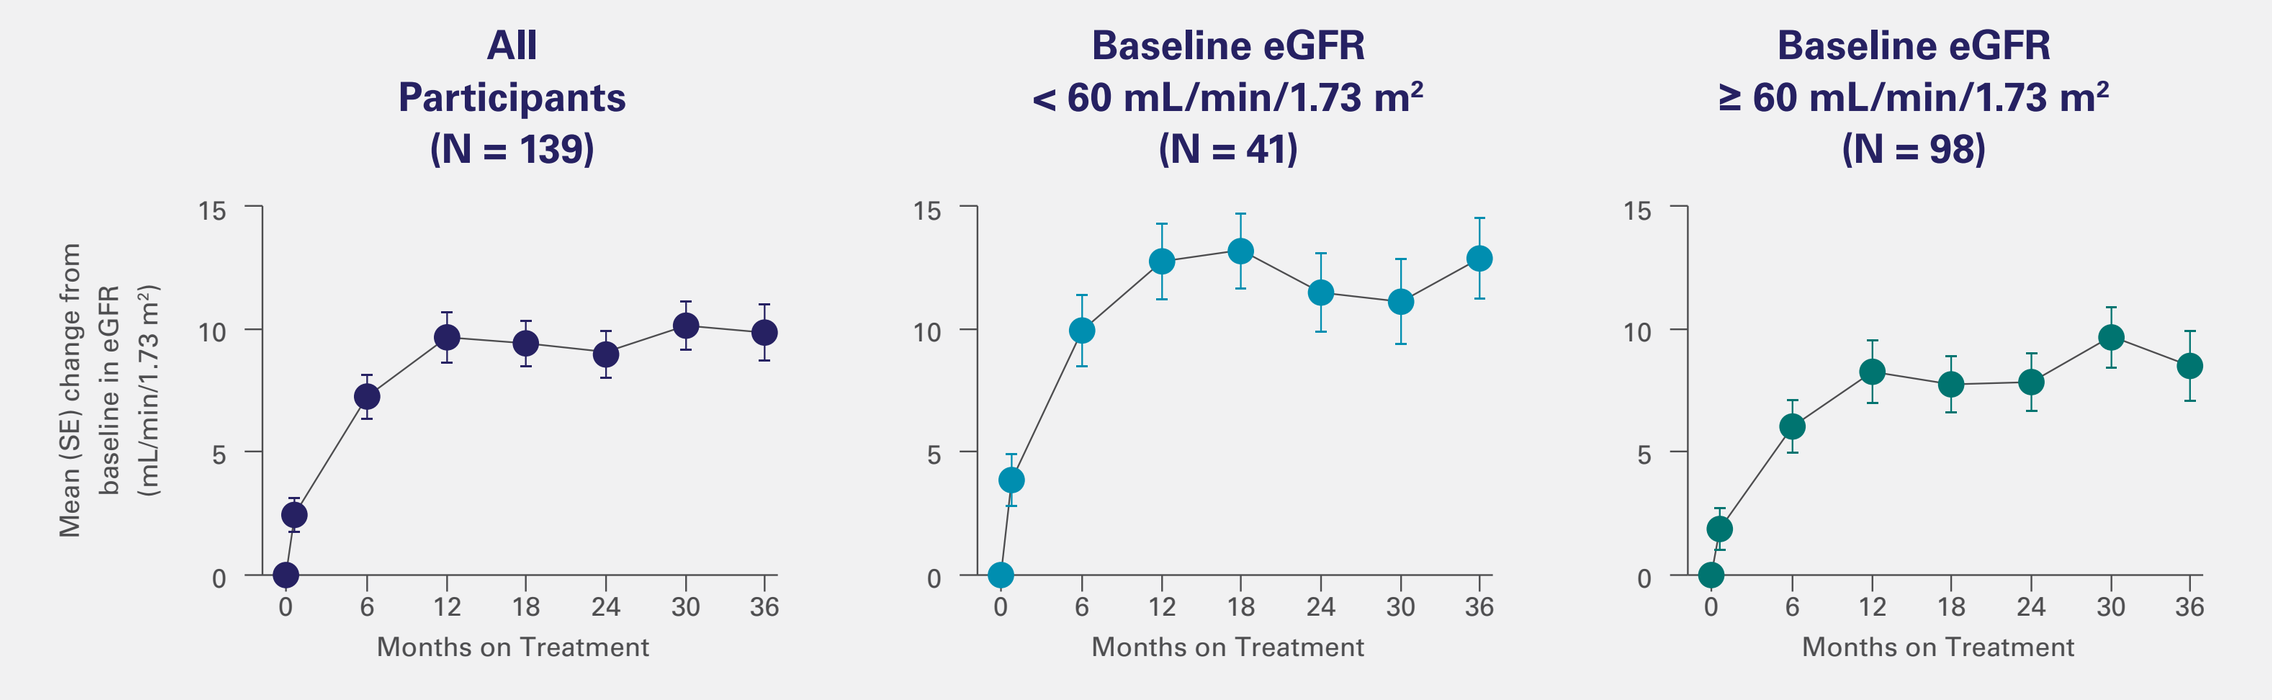
All Participants (N = 139)
Baseline eGFR < 60 mL/min/1.73 m² (N = 41)
Baseline eGFR ≥ 60 mL/min/1.73 m² (N = 98)
Mean (SE) change from baseline in eGFR (mL/min/1.73 m²)
Months on Treatment

A numerically greater improvement was observed in participants with lower baseline eGFR at month 36.

All Participants
eGFR Slope Change (mL/min/1.73 m²/year):
- 0-6 Months: **14.33**
- 6-36 Months: **0.89**

Baseline eGFR < 60 mL/min/1.73 m²
eGFR Slope Change (mL/min/1.73 m²/year):
- 0-6 Months: **19.93**
- 6-36 Months: **0.57**

Baseline eGFR ≥ 60 mL/min/1.73 m²
eGFR Slope Change (mL/min/1.73 m²/year):
- 0-6 Months: **11.98**
- 6-36 Months: **1.06**

Figure 6: **Proportion of Participants With Clinically Meaningful Increases in eGFR**

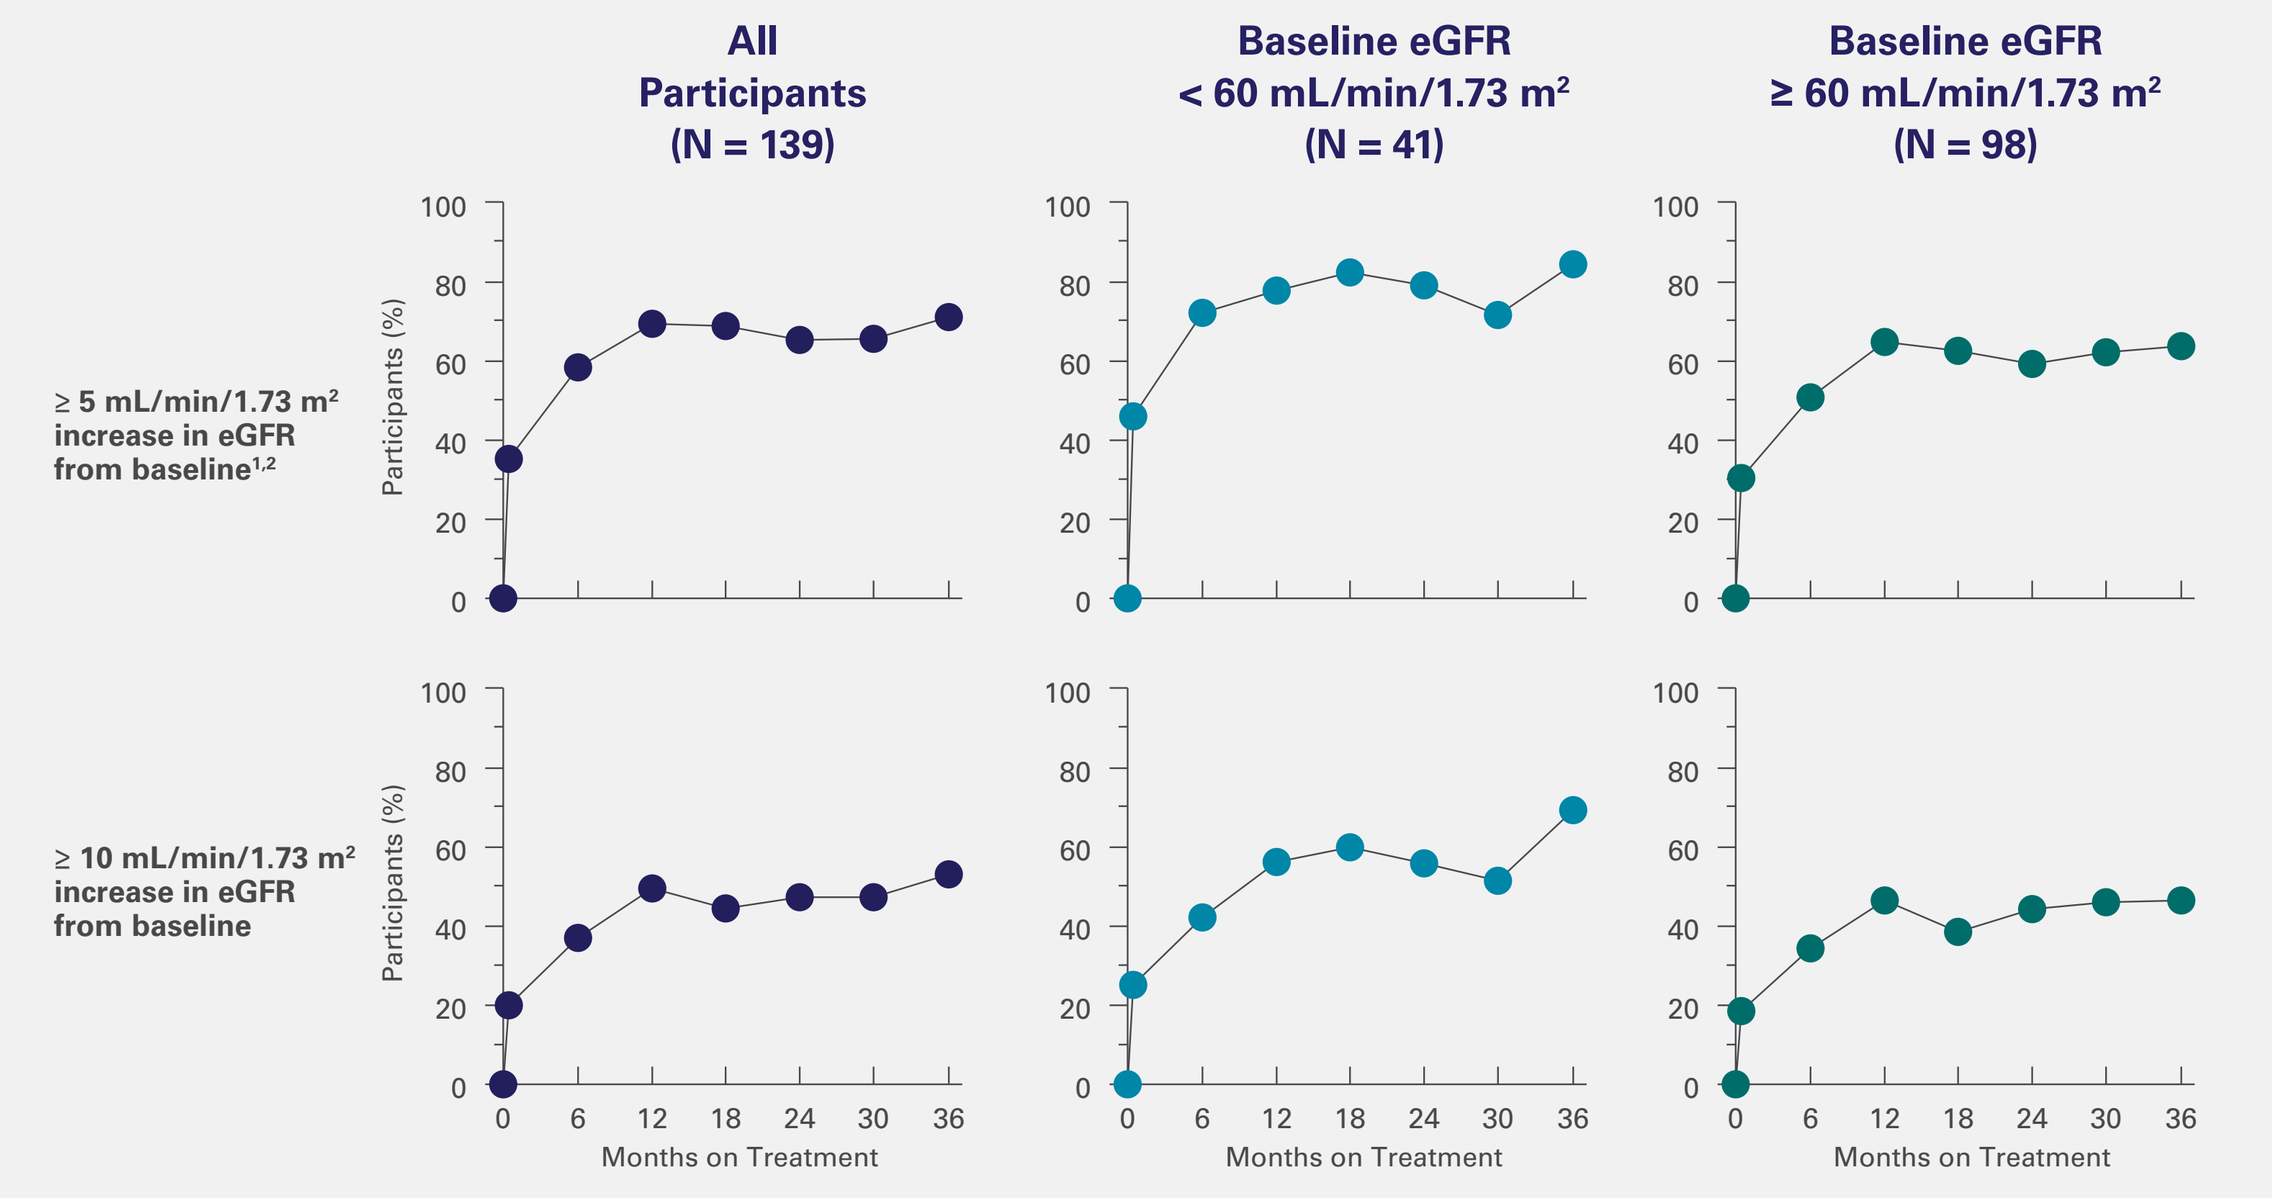
All Participants (N = 139)
Baseline eGFR < 60 mL/min/1.73 m² (N = 41)
Baseline eGFR ≥ 60 mL/min/1.73 m² (N = 98)
≥ 5 mL/min/1.73 m² increase in eGFR from baseline[1,2]
≥ 10 mL/min/1.73 m² increase in eGFR from baseline
Participants (%)
Months on Treatment

In 70.3% of all participants, sustained, clinically meaningful improvement in eGFR was observed at month 36.[10,11]

Note: Only 1 participant exceeded the decline in eGFR outcome with a reduction ≥ 30% from baseline of ≥ 2 consequent visits for at least 3 months (90 days) apart. This was secondary to nephrectomy due to malignancy.
[a]Clinically meaningful increases in eGFR were those ≥ 5 mL/ min / 1.73 m².

Table 2: **High Proportion of Participants Independent From Conventional Therapy at 3 Years**

Variable	All Participants
Number of participants with complete data at Year 3	**129**
Met multi-component efficacy endpoint criteria, n (%)	**110 (85%)**
Normal albumin-adjusted serum calcium[a]	118 (91%)
Independence from active vitamin D[b]	128 (99%)
Independence from therapeutic doses of calcium[b]	121 (94%)

[a]Normal calcium range: 8.3-10.6 mg/dL.
[b]Independence from conventional therapy defined as no active vitamin D and ≤ 600 mg/day elemental calcium.

Figure 7: **Normalization of 24-Hour Urinary Calcium Through Year 3 of PaTH Forward and PaTHway**

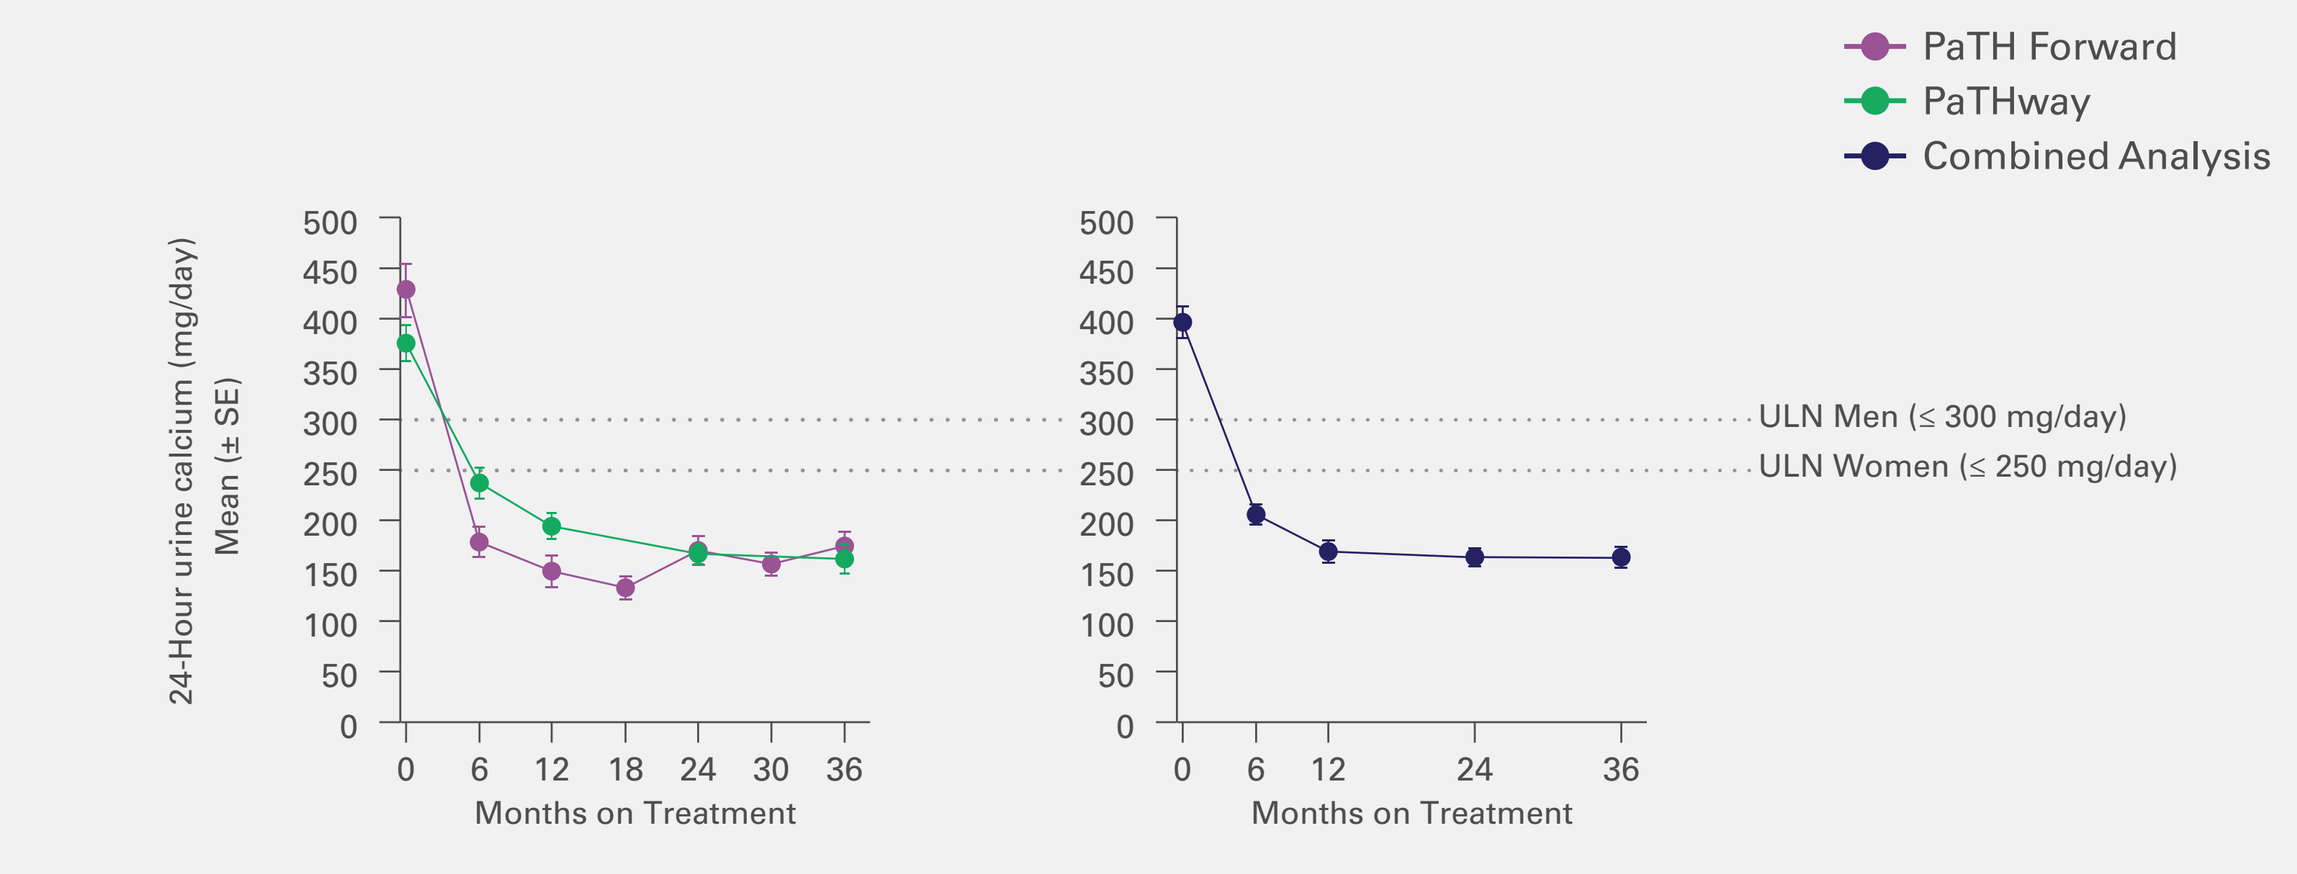
PaTH Forward
PaTHway
Combined Analysis
24-Hour urine calcium (mg/day) Mean (± SE)
ULN Men (≤ 300 mg/day)
ULN Women (≤ 250 mg/day)
Months on Treatment

Note: Values reflect total study population with data available from baseline and corresponding subsequent visits.

Table 3: **Summary of Adverse Events Through Year 3**

Treatment Emergent Adverse Events (TEAEs), n (%)	All Participants
	N = 139
Any TEAE	133 (95.7%)
Serious TEAE	26 (18.7%)
Related TEAE	72 (51.8%)
Serious related TEAE	2 (1.4%)
TEAE related to hyper- or hypocalcemia leading to ER/urgent care visit and/or hospitalization	6 (4.3%)
TEAE leading to discontinuation of study drug[a]	3 (2.2%)
TEAE leading to discontinuation of trial[b]	1 (0.7%)
TEAE leading to death[b]	1 (0.7%)

Treatment-related TEAEs occurring at a rate of ≥ 5% among all participants (N = 139) included:
- Injection site reaction (15.1%)
- Hypercalcemia (10.8%)
- Headache (9.4%)
- Hypocalcemia (7.9%)
- Nausea (7.9%)

Most TEAEs were classified as mild or moderate.[c]

[a]Includes TEAEs occurring on or after the first dose of palopegteriparatide in the Safety Analysis Population (participants who received ≥1 dose of palopegteriparatide): TEAEs leading to treatment discontinuation were deemed unrelated to study drug.
[b]One participant had a TEAE (fatal cardiac arrest unrelated to study drug) leading to discontinuation of the trial and death during blinded treatment.
[c]Classified using the World Health Organization toxicity grading scale (1=mild, 2=moderate, 3=severe, 4=life-threatening)

Abbreviations

AE = adverse event; **eGFR** = estimated glomerular filtration rate; **EU** = European Union; **MDRD** = Modified Diet in Renal Disease equation; **N** = number; **PTH** = parathyroid hormone; **SAE** = serious adverse event; **SD** = standard deviation; **SE** = standard error; **TDD** = total daily dose; **TEAE** = treatment emergent adverse event; **TransCon** = transient conjugation; **US** = United States

References

1. Khan AA, et al. *J Bone Miner Res.* 2022;37:2568-2585. **2.** Shoback DM, et al. *J Clin Endocrinol Metab.* 2016;101(6):2300-2312. **3.** Bilezikian JP, et al. *J Clin Endocrinol Metab.* 2016;101(6):2313-2324. **4.** Mannstadt M, et al. *Nat Rev Dis Primers.* 2017; 3:17055. **5.** Brandi ML, et al. *J Clin Endocrinol Metab.* 2016;101(6):2273-83. **6.** Khan AA, et al. *Eur J Endocrinol.* 2019;180(3):R33-63. **7.** Shoback D. *N Engl J Med.* 2008;359:391-403. **8.** Karpf DB, et al. *J Bone Miner Res.* 2020;35(8):1430-1440. **9.** Holten-Andersen L, et al. *J Bone Miner Res.* 2019;34(11):2075-2086. **10.** Mayne TJ, et al. *Clin Transplant.* 2021;35(7):e14326. **11.** Ku E, et al. *J Am Soc Nephrol.* 2016;27(7):2196-204. **12.** Saha S, et al. *J Clin Endocrinol Metab.* 2021;106(7):2092-2102.

Disclosures and Funding

Ascendis Pharma and the authors thank the participants, study sites, and investigators who participated in this clinical trial.

Ascendis Pharma Bone Diseases A/S funded this trial and participated in the trial design, research, analysis, data collection, interpretation of the data, and the review and approval of the publication. All authors had access to relevant data and participated in the drafting, review, and approval of this publication. No honoraria or payments were made for authorship. Medical writing support was provided by Robert Geist, MD, of Ascendis Pharma.

Financial arrangements of the authors with companies whose products may be related to this presentation are listed as declared by the authors: **EG:** An employee of the US Department of Veterans Affairs. Opinions expressed in this paper are those of the authors and do not necessarily represent the opinion of the Department of Veterans Affairs. Advisory board, speakers bureau, travel, accommodations, and expenses from Ascendis Pharma; industry-sponsored grants from AstraZeneca. **PS:** Stock ownership Novo Nordisk, Genmab. **LR:** Research funding from Takeda, Kyowa Kirin International, Ascendis Pharma, and Calcilytix; honoraria from Calcilytix Therapeutics; advisory board for Takeda and Amolyt. **AAK:** Research funding and/or industry grants from Amolyt, Ascendis Pharma, Chugai, Radius, and Takeda; honoraria from and advisory board member for Amgen, Alexion, Ascendis Pharma, and Takeda; travel, accommodations, and expenses from Ascendis Pharma; consulting role for Amgen, Alexion, Amolyt, and Ascendis Pharma; speakers bureau participation for Amgen. **BC:** Research funding and industry grants from Ascendis and Takeda; advisory board member, consultant, honoraria from Ascendis, Takeda, Entera-Bio, Extend-Bio, Amolyt. FC: Study investigator for Ascendis Pharma, Amolyt, and Calcilytix. **SMS:** Research funding from Amgen, Amolyt, Ascendis, Ardelyx, Fresenius, and OPKO; consultant Amgen, Ardelyx, Bayer, Fresenius, Horizon, OPKO, and Shire. **DMS:** Research/salary funding from Bone Health Tech; research funding from Ascendis Pharma. **LK:** Research funding from Alexion/Amolyt and Ascendis Pharma; speakers bureau, honoraria from Amgen and Ascendis Pharma; advisory board, consultant for Alexion and Ascendis Pharma. **MRR:** Study investigator for Takeda, Ascendis Pharma, Amolyt, and Calcilytix; advisory board for Ascendis Pharma, speakers bureau for Ascendis Pharma; consulting for MBX. **AP:** Consultant for Theramex, Bruno Farmaceutici, Amgen; research funding from Amgen, Shire, Ascendis Pharma; speakers bureau UCB, Amgen; industry grant from Amgen. **CG:** Honoraria from Pendopharm; research funding from Ascendis Pharma, Amolyt, Shire, and Takeda. **ET:** Advisory role for Ascendis Pharma and Kyowa Kirin; honoraria from Alexion, Ascendis Pharma, KKI, Takeda and UCB. **CZ, MAM, BL, JU, CTS,** and **ADS:** Full-time employees of Ascendis Pharma.

Ascendis, Ascendis Pharma, Ascendis Pharma logo, the company logo and TransCon are trademarks owned by the Ascendis Pharma group. © October 2025 Ascendis Pharma A/S

MED-US-TC-PTH-2600035
March 2026