TransCon PTH Enables Independence From Conventional Therapy While Maintaining Normal Serum Calcium in Adults With Chronic Postsurgical Hypoparathyroidism: Results From a Sub-Analysis of the PaTHway Phase 3 Trial

Mark Schneider,[1] Bart Clarke,[2] Aliya Khan,[3] Mishaela Rubin,[4] Peter Schwarz,[5] Dolores M Shoback,[6] Claudia Gagnon,[7] Andrea Palermo,[8] Lisa G Abbott,[9] Lynn Kohlmeier,[10] Filomena Cetani,[11] Elena Tsourdi,[12] Rajesh K Jain,[13] Xuebei An,[1*] Alden R Smith,[1] Bryant Lai,[1] Jenny Ukena,[1] Christopher T Sibley,[1] Aimee D Shu,[1] Lars Rejnmark[14]

[1]Ascendis Pharma Inc, Palo Alto, CA, USA; [2]Mayo Clinic E18-A, Rochester, MN, USA; [3]McMaster University, Hamilton, ON, Canada; [4]Columbia University, New York, NY, USA; [5]Rigshospitalet, Copenhagen, Denmark; [6]UCSF/VA Medical Center, San Francisco, CA, USA; [7]CHU de Québec-Université Laval Research Centre, Quebec City, QC, Canada and Department of Medicine, Université Laval, Quebec City, QC, Canada; [8]Unit of Metabolic Bone and Thyroid Disorders, Fondazione Policlinico Campus Bio-medico, Rome, Italy and Unit of Endocrinology and Diabetes, Campus Bio-medico University, Rome, Italy; [9]Northern Nevada Endocrinology, Reno, NV and University of Nevada, Reno, Reno, NV, USA; [10]Endocrinology and Spokane Osteoporosis, Spokane, WA, USA; [11]University Hospital of Pisa, Endocrine Unit, Pisa, Italy; [12]Uniklinikum Dresden, Medizinische Klinik III, Abteilung Endokrinologie, Dresden, Germany; [13]University of Chicago, Chicago, IL, USA; [14]Aarhus University Hospital, Aarhus N, Denmark

*Affiliation at time of abstract submission

224

BACKGROUND

- Hypoparathyroidism is a rare endocrine disease caused by inadequate levels of parathyroid hormone (PTH)
- Anterior neck surgery resulting in resection of or iatrogenic injury to the parathyroid glands accounts for ~75% of cases of hypoparathyroidism[1]
- An intact PTH axis maintains normal calcium and phosphate homeostasis[2,3]
 - PTH acts directly on bone and kidney, and indirectly, the intestine via endogenous calcitriol produced in the kidney[2,4]
 - PTH promotes normal nerve and muscle function[5]
- Conventional therapy for hypoparathyroidism (active vitamin D [eg, calcitriol, alfacalcidol] and elemental calcium) does not restore normal PTH physiology but aims to alleviate hypocalcemic symptoms
- Ideal PTH therapy for hypoparathyroidism should provide PTH levels within the physiological range and restore downstream calcitriol, promoting independence from conventional therapy and normalizing:
 - Serum and urine biochemistries
 - Skeletal health
 - Quality of life

Figure 1. TransCon® PTH (palopegteriparatide) Design

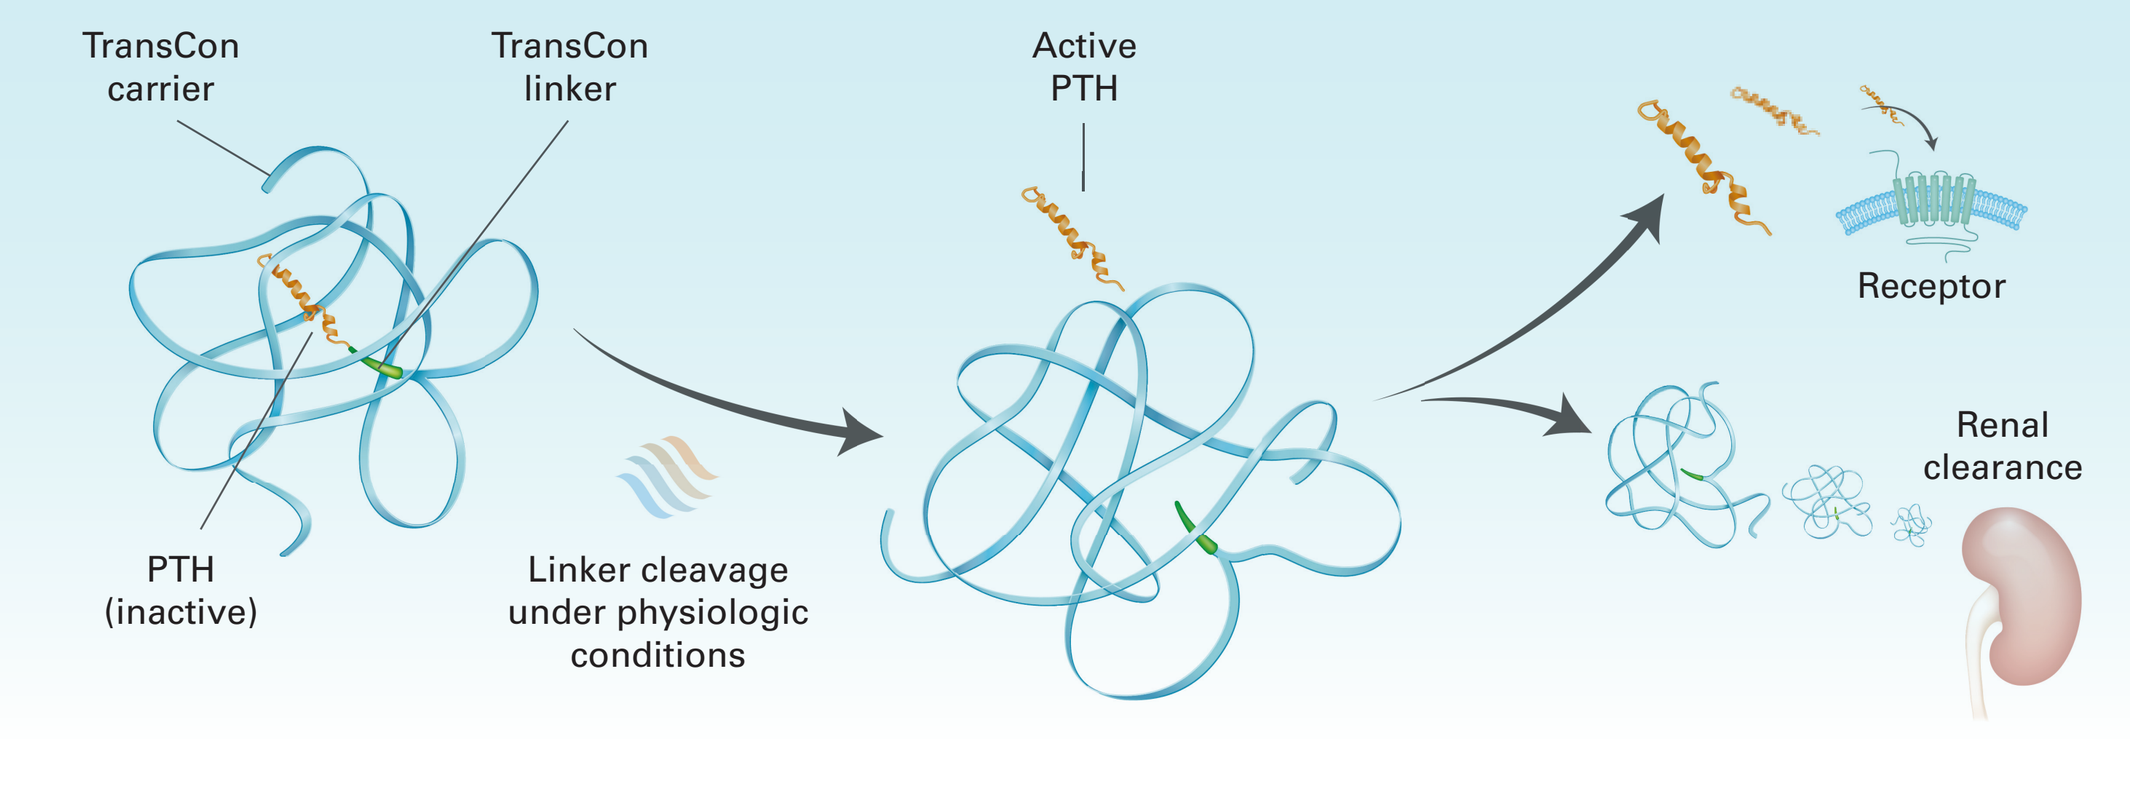

TransCon carrier
TransCon linker
Active PTH
Receptor
Renal clearance
PTH (inactive)
Linker cleavage under physiologic conditions

TransCon PTH (palopegteriparatide) is an investigational prodrug of PTH (1-34), administered subcutaneously once daily, with sustained release of active PTH that provides PTH levels in the physiological range for 24 hours/day in adults with hypoparathyroidism[6]

METHODS

Figure 2. TransCon PTH Phase 3 PaTHway Trial Design (NCT04701203)

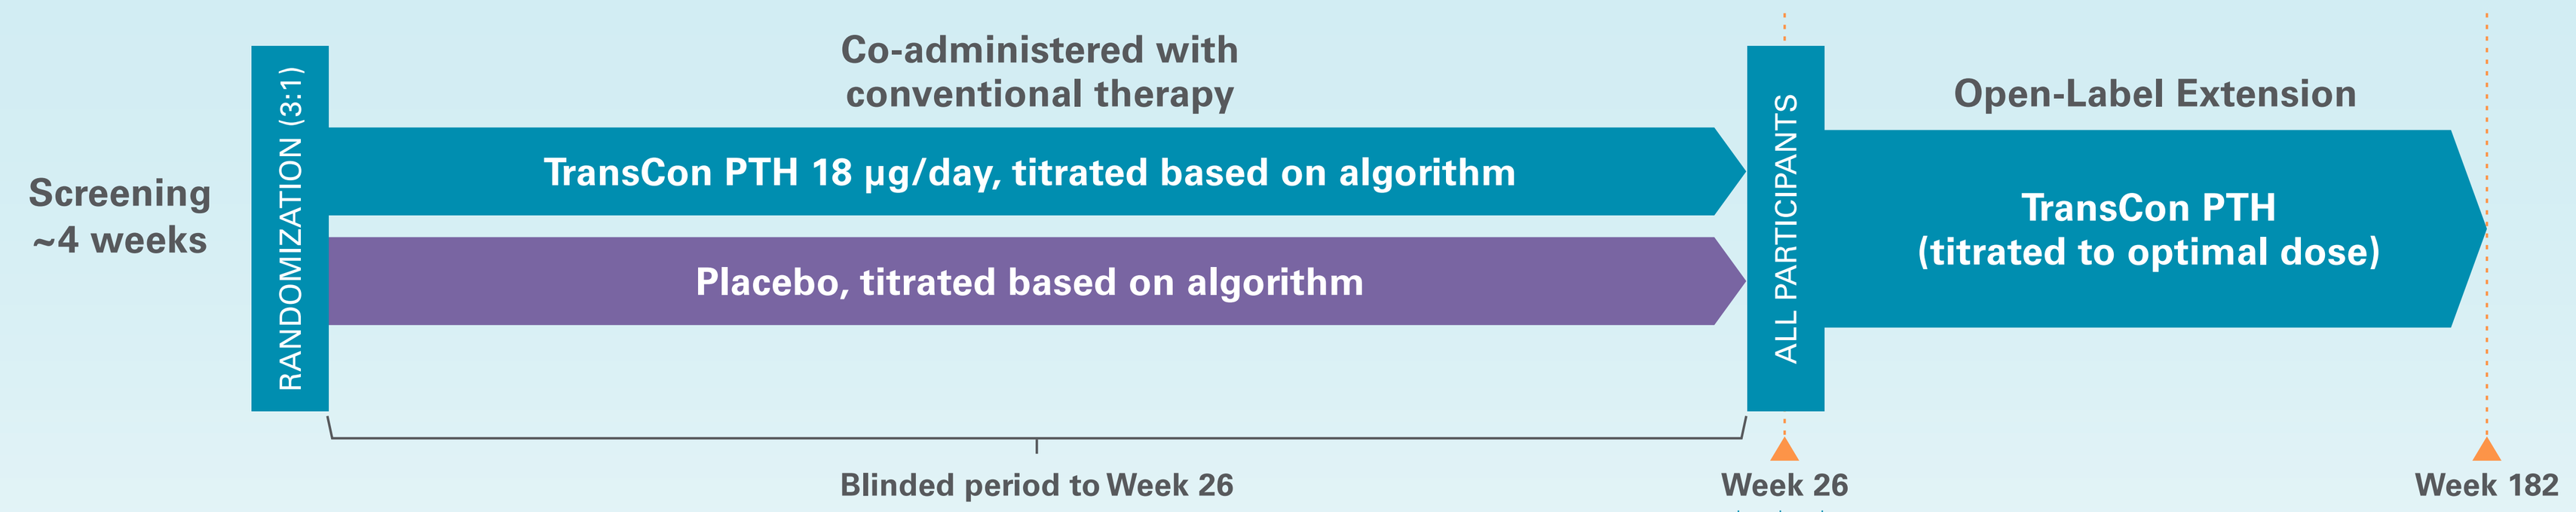

Screening ~4 weeks
RANDOMIZATION (3:1)
Co-administered with conventional therapy
TransCon PTH 18 µg/day, titrated based on algorithm
Placebo, titrated based on algorithm
ALL PARTICIPANTS
Open-Label Extension
TransCon PTH (titrated to optimal dose)
Blinded period to Week 26
Week 26
Week 182

Key Primary Endpoint
Proportion of participants with:[a]
- Serum calcium in the normal range (8.3–10.6 mg/dL); ***and***
- Independence from therapeutic doses of calcium;[b] ***and***
- Independence from active vitamin D[c]

Key Safety and Tolerability Endpoints
- HPES-Symptom physical and cognitive domain scores
- HPES-Impact physical functioning and daily life domain scores
- SF-36 physical functioning subscale score

Key Safety and Tolerability Endpoints
- 24-hour urine calcium excretion
- Incidence of TEAEs, SAEs, and treatment-related TEAEs

[a]No increase in prescribed study drug within 4 weeks prior to week 26 visit. [b]Taking ≤ 600 mg/day elemental calcium and use of any PRN doses ≤ 7 days within 4 weeks prior to week 26. [c]Not taking active vitamin D and use of any PRN doses ≤ 7 days within 4 weeks prior to the week 26 visit.

- Prespecified sub-analysis of participants with postsurgical hypoparathyroidism from the PaTHway trial of TransCon PTH
- PaTHway is a phase 3 trial with a 26-week randomized, double-blind, placebo-controlled period and ongoing 156-week open-label extension designed to evaluate the efficacy, safety, and tolerability of TransCon PTH in adults with hypoparathyroidism **(Figure 2)**
 - Adults (≥ 18 years) with chronic hypoparathyroidism of at least 6 months duration and reliant on calcitriol ≥ 0.5 µg/day or alfacalcidol ≥ 1.0 µg/day and therapeutic elemental calcium ≥ 800 mg/day for at least 12 weeks prior to screening were eligible for inclusion in the trial
 - A total of 106 participants were screened and 84 were randomized 3:1 to treatment with TransCon PTH (n = 63) or placebo (n = 21), both co-administered with conventional therapy; 82 participants received at least 1 dose of study drug (n = 61 TransCon PTH, n = 21 placebo)
- The multicomponent primary efficacy endpoint was defined as normocalcemia (8.3–10.6 mg/dL), independence from conventional therapy (no active vitamin D and ≤ 600 mg/day calcium), and no dose change within the 4 weeks prior to week 26
- Key secondary endpoints included components of the Hypoparathyroidism Patient Experience Scale (HPES) and 36-Item Short Form Survey (SF-36, version 2)
 - The HPES is a psychometrically validated, disease-specific measure designed to assess the symptoms and impacts associated with hypoparathyroidism; higher HPES scores indicate greater symptom frequency or impact.[7] Decreases in HPES scores indicate improvement
 - The SF-36 is a 36-item health questionnaire that assesses 8 dimensions of health; higher scores indicate a lower burden of disease.[8] Increases in SF-36 scores indicate improvement

RESULTS

BASELINE DEMOGRAPHICS AND CLINICAL CHARACTERISTICS

- Of the 82 participants who received study drug during the blinded period, 70 (52 TransCon PTH, 18 placebo) had postsurgical hypoparathyroidism and were included in the sub-analysis population **(Figure 3)**
- At baseline, participants with postsurgical hypoparathyroidism had a mean age of 51 years, 86% (60/70) were female, and 93% (65/70) were White **(Table 1)**

Figure 3. Hypoparathyroidism Etiology in the PaTHway Trial

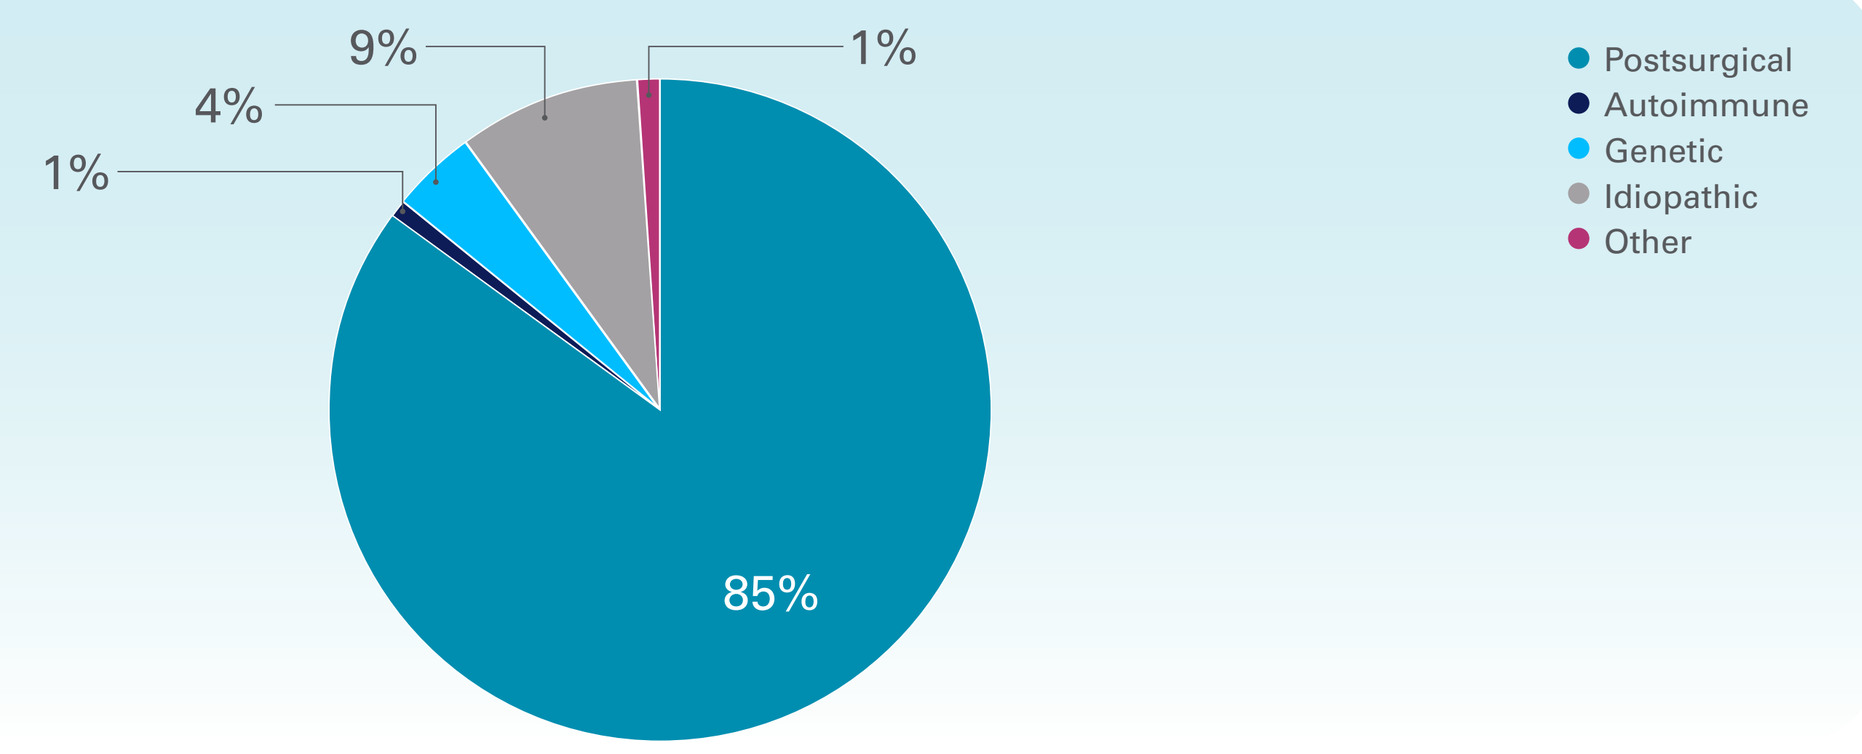

9%
4%
1%
1%
85%
Postsurgical
Autoimmune
Genetic
Idiopathic
Other

Table 1. Baseline Demographics and Clinical Characteristics of Participants With Postsurgical Hypoparathyroidism

Baseline Characteristics	Participants With Postsurgical Hypoparathyroidism (n = 70)	Baseline Characteristics	Participants With Postsurgical Hypoparathyroidism (n = 70)
Age (years), mean (SD)	51 (12)	**Duration of hypoparathyroidism (years)**	
		Mean (SD)	10.6 (9.9)
Sex, % female	86	Median (min,max)	7.0 (1,56)
Race, % White	93	**Conventional therapy, median TDD**	
Geographic region, %		Calcium (mg/day)	1763
North America	60	Calcitriol (µg/day)[a]	0.75
Europe	40	Alfacalcidol (µg/day)[b]	2.0

[a]n = 59 (84%) participants used calcitriol at baseline. [b]n = 11 (16%) participants used alfacalcidol at baseline.

MULTICOMPONENT PRIMARY EFFICACY ENDPOINT

- At week 26, 81% (42/52) of participants with postsurgical hypoparathyroidism treated with TransCon PTH met the multicomponent primary efficacy endpoint compared with 6% (1/18) who received placebo **(Table 2)**
 - By week 26, 94% (49/52) of participants treated with TransCon PTH and 6% (1/18) who received placebo achieved independence from conventional therapy (no active vitamin D and ≤ 600 mg/day calcium)
 - Albumin-adjusted serum calcium was within the normal range for 81% (42/52) of participants treated with TransCon PTH and 50% (9/18) who received placebo at week 26
 - Mean serum calcium levels were maintained within the normal range for participants receiving TransCon PTH at all study visits **(Figure 4)**

Table 2. Participants With Postsurgical Hypoparathyroidism Who Met the Multicomponent Primary Endpoint at Week 26

	Participants With Postsurgical Hypoparathyroidism	
	TransCon PTH (n = 52)	Placebo (n = 18)
Number of participants meeting the primary endpoint criteria at week 26 (responders)	42	1
Proportion, % (95% CI)	81 (68, 90)	6 (0, 27)
P value (TransCon PTH vs Placebo)[a]	< 0.0001	
Number of participants meeting each component, n (%)		
Albumin-adjusted serum calcium within the normal range[b]	42 (81)	9 (50)
Independence from active vitamin D[c]	51 (98)	5 (28)
Independence from therapeutic doses of calcium[d]	49 (94)	1 (6)
No increase in prescribed study drug[e]	50 (96)	10 (56)

[a]The *P* value is calculated from Cochran-Mantel-Haenszel test for response rates between TransCon PTH and placebo, controlling for the etiology of hypoparathyroidism. [b]The normal range for albumin-adjusted serum calcium is 8.3-10.6 mg/dL. [c]Not taking active vitamin D and use of any PRN doses ≤ 7 days within 4 weeks prior to the week 26 visit. [d]Taking ≤ 600 mg/day elemental calcium and use of any PRN doses ≤ 7 days within 4 weeks prior to week 26. [e]Within 4 weeks prior to the week 26 visit.

Figure 4. Albumin-Adjusted Serum Calcium Levels

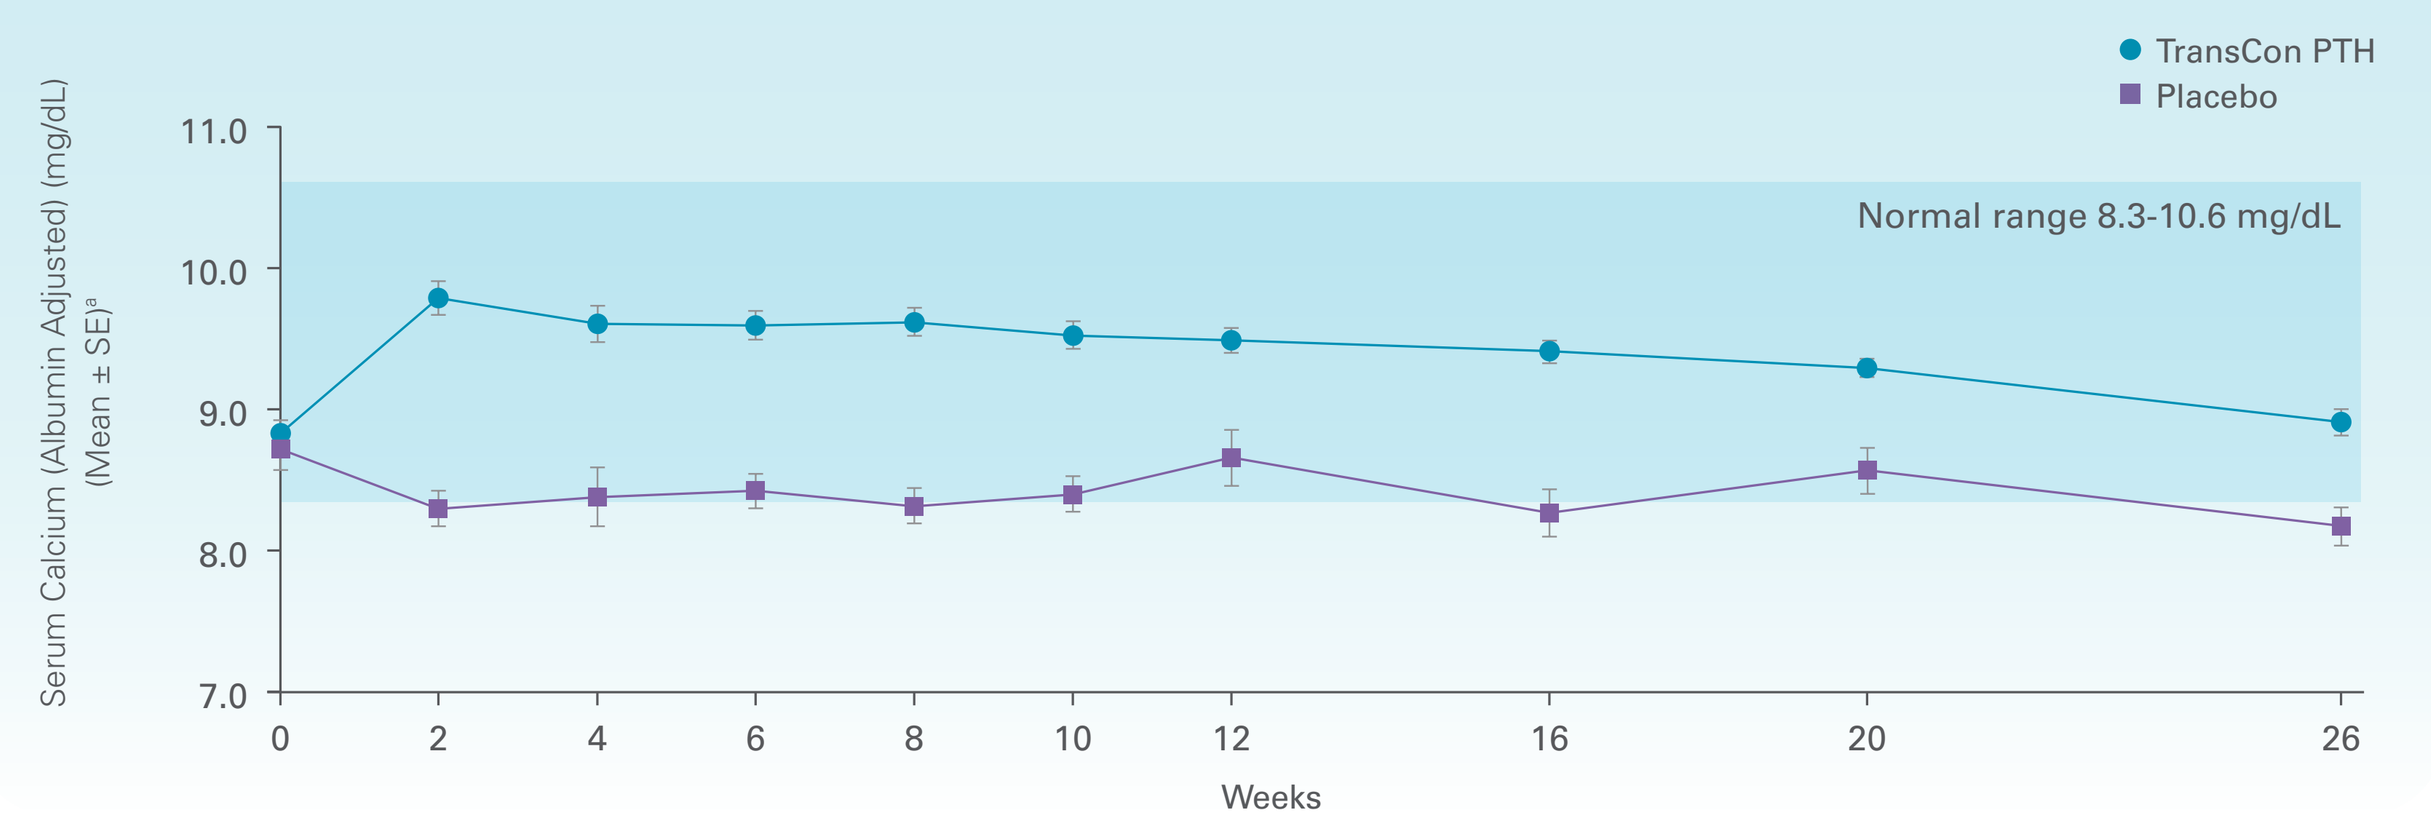

TransCon PTH
Placebo
Normal range 8.3-10.6 mg/dL
Serum Calcium (Albumin Adjusted) (mg/dL) (Mean ± SE)[a]
11.0
10.0
9.0
8.0
7.0
0 2 4 6 8 10 12 16 20 26
Weeks

[a]Data from the subpopulation of PaTHway trial participants with postsurgical hypoparathyroidism only (n = 70).

KEY SECONDARY ENDPOINTS

- At week 26, 81% (42/52) of participants with postsurgical Participants with postsurgical hypoparathyroidism who received TransCon PTH demonstrated significant improvement compared to those on placebo in disease-related symptoms and the impact of hypoparathyroidism on physical functioning and daily life (all *P* < 0.01) **(Figure 5)**
- Health-related quality of life, as measured by the SF-36 physical functioning subscale score, improved significantly with TransCon PTH therapy compared to placebo in participants with postsurgical hypoparathyroidism (*P* = 0.0063) **(Figure 6)**

Figure 5. HPES Symptom and Impact Domain Scores

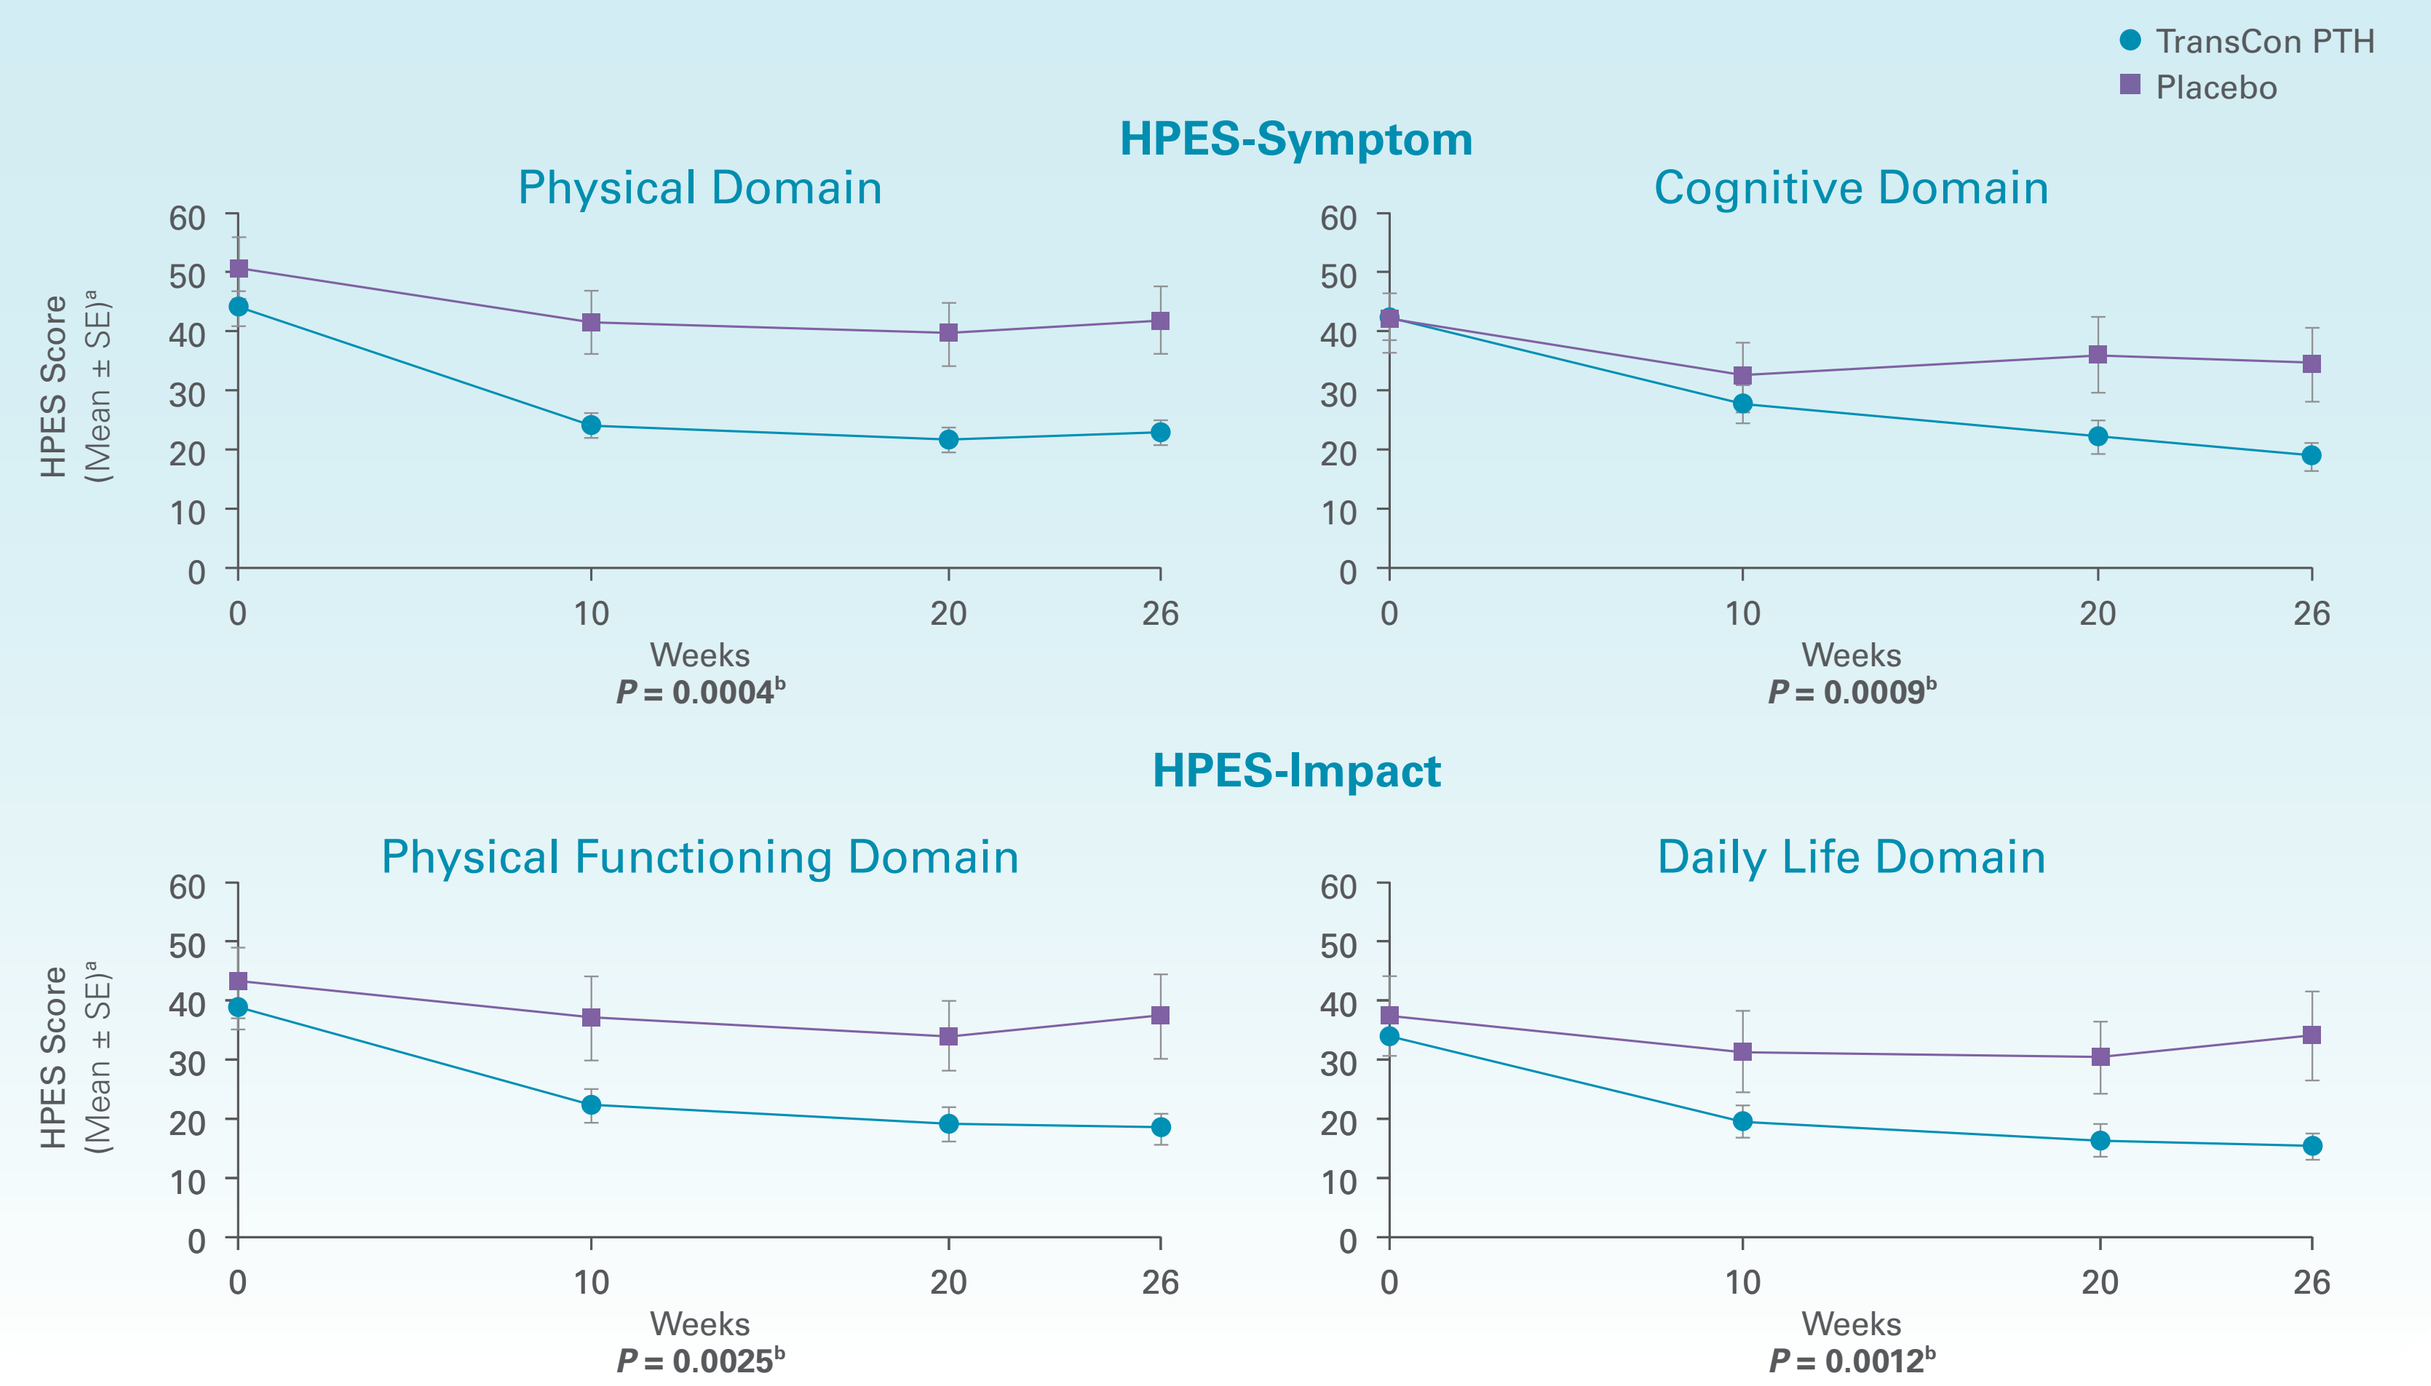

TransCon PTH
Placebo
HPES-Symptom
Physical Domain
Cognitive Domain
HPES Score (Mean ± SE)[a]
Weeks
P = 0.0004[b]
P = 0.0009[b]
HPES-Impact
Physical Functioning Domain
Daily Life Domain
P = 0.0025[b]
P = 0.0012[b]

[a]Data from the subpopulation of PaTHway trial participants with postsurgical hypoparathyroidism only (n = 70). [b]The p-values are from ANCOVA model with unequal variance testing the difference in change from baseline at week 26 between TransCon PTH and placebo, with etiology of hypoparathyroidism as fixed effects and baseline HPES domain scores as covariates.

Figure 6. SF-36 Physical Functioning Subscale Scores

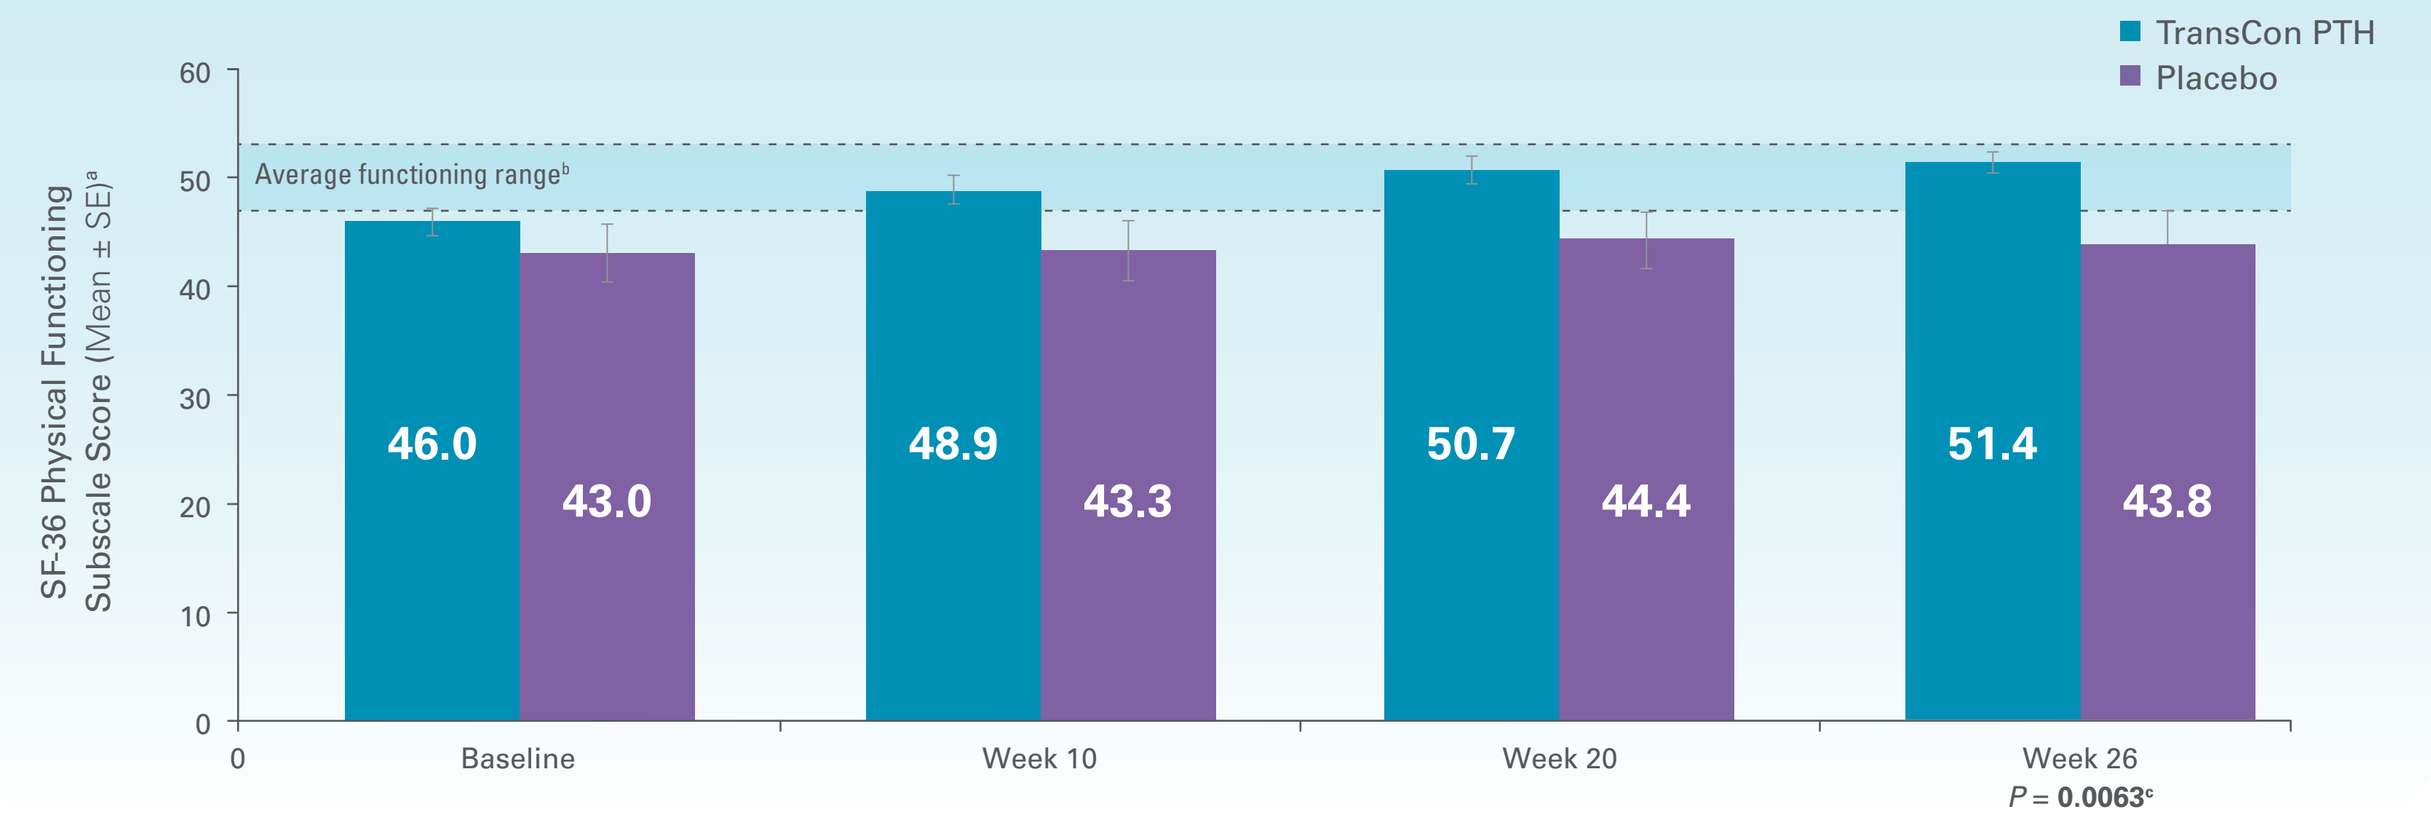

TransCon PTH
Placebo
Average functioning range[b]
SF-36 Physical Functioning Subscale Score (Mean ± SE)[a]
46.0
43.0
48.9
43.3
50.7
44.4
51.4
43.8
Baseline
Week 10
Week 20
Week 26
P = 0.0063[c]

[a]Data from the subpopulation of PaTHway trial participants with postsurgical hypoparathyroidism only (n = 70). [b]The dashed lines (—) indicate the upper (53) and lower (47) bounds of T scores considered to be in the range of average functioning for the U.S. general population of group-level data; the MID threshold for between-group comparisons of physical functioning is 3.0 points.[8] [c]The *P* value is from the ANCOVA model with unequal variance testing the difference in change from baseline at week 26 between TransCon PTH and placebo, with etiology of hypoparathyroidism as fixed effects and baseline SF-36 subscale score as a covariate.

SAFETY AND TOLERABILITY

- Participants with postsurgical hypoparathyroidism treated with TransCon PTH showed a normalization and greater reduction in urine calcium excretion than those on placebo from baseline to week 26 **(Figure 7)**
- Most treatment-emergent adverse events (TEAEs) were mild or moderate in severity and no treatment-related TEAEs led to discontinuation of treatment or the trial **(Table 3)**

Figure 7. 24-Hour Urine Calcium Excretion

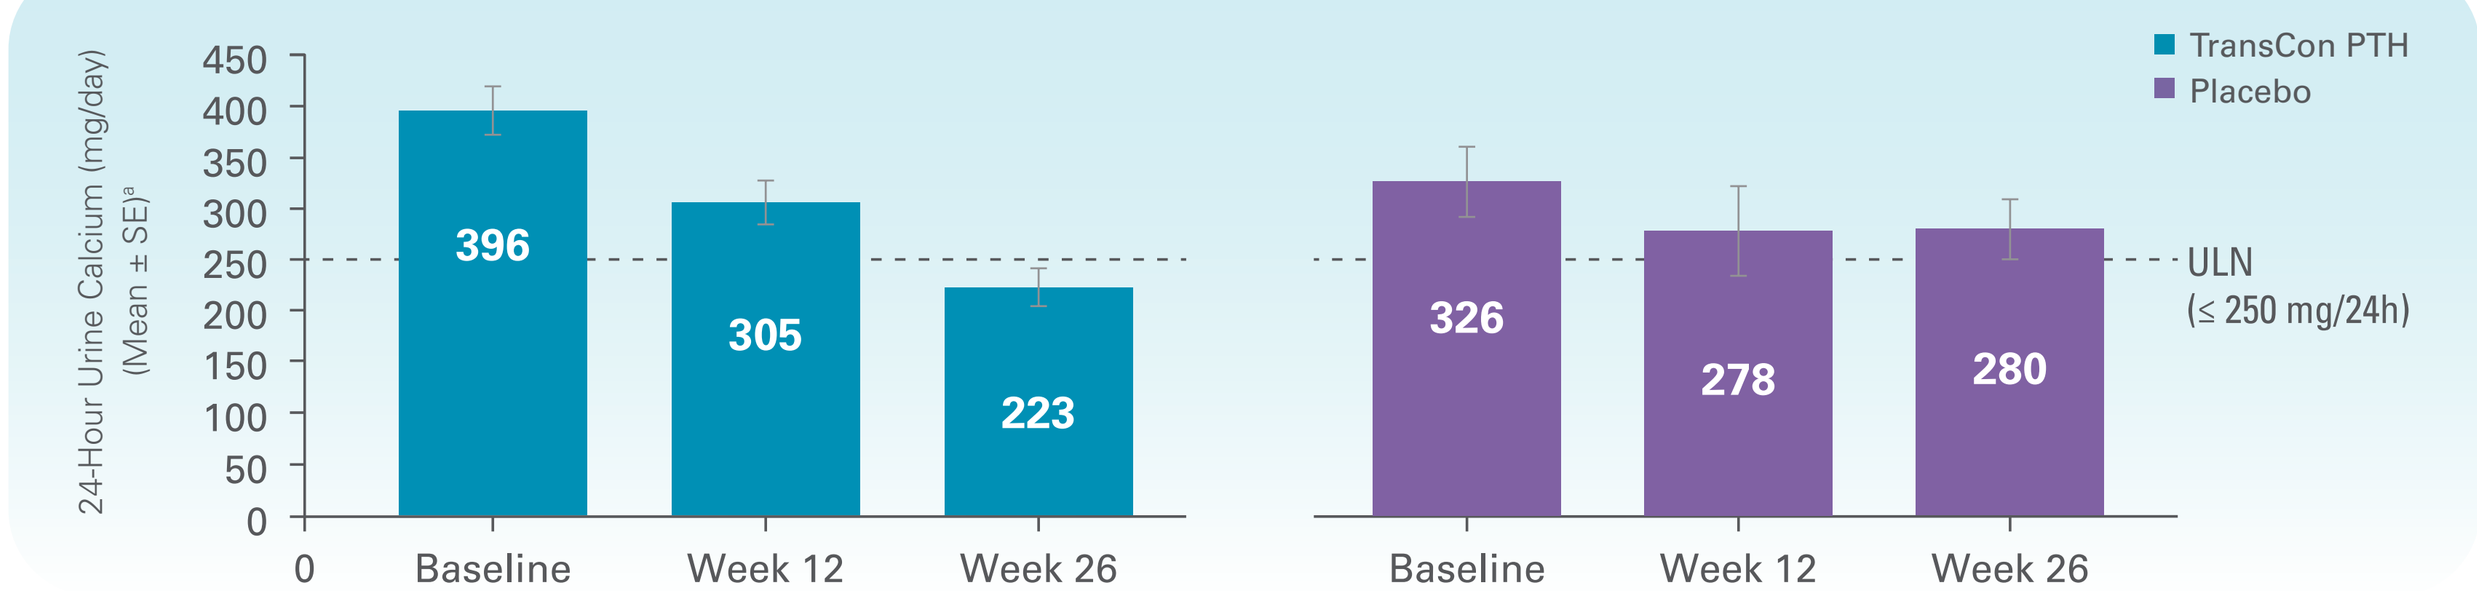

TransCon PTH
Placebo
24-Hour Urine Calcium (mg/day) (Mean ± SE)[a]
396
305
223
326
278
280
Baseline
Week 12
Week 26
ULN (≤ 250 mg/24h)

[a]Data from the subpopulation of PaTHway trial participants with postsurgical hypoparathyroidism only (n = 70).

Table 3. Summary of TEAEs Through Week 26

	Participants With Postsurgical Hypoparathyroidism	
TEAE, n (%)	**TransCon PTH (n = 52)**	**Placebo (n = 18)**
Any TEAE	42 (80.8)	18 (100.0)
Serious TEAE	4 (7.7)	2 (11.1)
Severity[a]		
Grade 1	21 (40.4)	11 (61.1)
Grade 2	19 (36.5)	6 (33.3)
Grade 3	1 (1.9)	1 (5.6)
Grade 4	1 (1.9)	0
Related TEAE	25 (48.1)	7 (38.9)
Serious related TEAE	1 (1.9)	0
TEAE related to hypercalcemia or hypocalcemia leading to ER/urgent care visit and/or hospitalization	2 (3.8)	1 (5.6)
TEAE leading to discontinuation of study drug[b]	1 (1.9)	1 (5.6)
TEAE leading to death[b]	1 (1.9)	0
Related TEAEs in ≥ 2% of all participants by preferred term, n (%)		
Injection site reaction	16 (30.8)	0
Headache	6 (11.5)	0
Nausea	4 (7.7)	1 (5.6)
Hypercalcemia	4 (7.7)	0
Hypocalcemia	0	3 (16.7)
Injection site bruising	2 (3.8)	1 (5.6)
Abdominal discomfort	2 (3.8)	0
Constipation	2 (3.8)	0
Photosensitivity reaction	2 (3.8)	0

[a]In the severity categories, participants are displayed for the highest severity only. TEAEs occurring prior to the first dose of open-label treatment are included. AE severity is assessed by WHO toxicity grading scale. [b]One participant had a TEAE (fatal cardiac arrest assessed as unrelated to study drug) leading to discontinuation of the study drug and death.

CONCLUSIONS

81% of participants with postsurgical hypoparathyroidism treated with TransCon PTH achieved independence from conventional therapy while maintaining normocalcemia vs 6% treated with placebo in this prespecified sub-analysis of the pivotal PaTHway phase 3 trial

- Across multiple evaluated parameters in participants with postsurgical hypoparathyroidism, TransCon PTH therapy enabled:
 - Independence from conventional therapy (no active vitamin D and ≤ 600 mg/day calcium)
 - Improvement of hypoparathyroidism-specific symptoms and their impact on daily life and physical functioning as measured by the HPES and SF-36
 - Normalization of mean 24-hour urine calcium excretion
- Most TEAEs were mild or moderate, and no participants discontinued study treatment or the trial due to a treatment-related TEAE
- These findings are comparable to those of the overall PaTHway trial population (N = 82) demonstrating consistency in improved outcomes with TransCon PTH compared to conventional therapy across etiologies of hypoparathyroidism

ABBREVIATIONS
ER = emergency room; **HPES** = hypoparathyroidism patient experience scale; **MID** = minimal important difference; **PTH** = parathyroid hormone; **SAE** = serious adverse event; **SE** = standard error; **SF-36** = 36-item short form survey; **TDD** = total daily dose; **TEAE** = treatment-emergent adverse event; **TransCon** = transient conjugation; **ULN** = upper limit of normal; **WHO** = World Health Organization

REFERENCES:
1. Clarke BL, et al. *J Clin Endocrinol Metab.* 2016;101(6):2284-99. **2.** Brandi ML, et al. *J Clin Endocrinol Metab.* 2016;101(6):2273-2283. **3.** Shoback DM, et al. *J Clin Endocrinol Metab.* 2016;101(6):2300-2312. **4.** Bilezikian JP, et al. *J Clin Endocrinol Metab.* 2016;101(6):2313-2324. **5.** Mannstadt M, et al. *Nat Rev Dis Primers.* 2017; 3:17055. **6.** Karpf DB, et al. *J Bone Miner Res.* 2020;35(8):1430-1440. **7.** Brod M, et al. *J Patient Rep Outcomes.* 2021;5(1):70. **8.** Maruish, M. E. (Ed.). User's manual for the SF-36v2 Health Survey (3rd ed.). Lincoln, RI: Quality Metric Incorporated.

DISCLOSURES AND FUNDING
This study was sponsored by Ascendis Pharma Bone Diseases A/S.

Financial arrangements of the authors with companies whose products may be related to this presentation are listed as declared by the authors: **M Schneider** is a full-time employee of Ascendis Pharma. **B Clarke:** Research funding and industry grants from Ascendis Pharma and Takeda; advisory board member, consultant, honoraria from Ascendis Pharma, Takeda, Entera-Bio, Extend-Bio, Amolyt. **A Khan:** Research funding and/or grants from Amolyt, Ascendis Pharma, Chugai, Radius, Takeda, Ultragenyx; advisory board member, honoraria from Amgen, Alexion, Ascendis Pharma, Takeda, Ultragenyx; consultant and/or speakers bureau for Amgen, Alexion, Amolyt, Ascendis Pharma, Ultragenyx. **M Rubin:** Institutional research funding from Chugai Pharma. **P Schwarz:** Stock ownership Novo Nordisk, Genmab. **D Shoback:** Research/salary funding from Bone Health Tech. **C Gagnon:** Research funding, advisory board for Novo Nordisk; honoraria from Amgen; industry grants from Ascendis Pharma, Shire, Takeda. **A Palermo:** Consultant for Theramex, Bruno Farmaceutici, Amgen; research funding from Amgen, Shire, Ascendis Pharma; speakers bureau UCB, Amgen; industry grant from Amgen. **L Abbott:** Advisory board, honoraria Ascendis Pharma; speakers bureau Abbott, Clarus, research funding from Ascendis Pharma, Shire/Takeda. **L Kohlmeier:** Research funding Ascendis; speakers bureau, honoraria from Amgen, Radius, Ascendis, MediMaps; advisory board, consultant for Radius, Alexion, Ascendis. **F Cetani:** Nothing to disclose; **E Tsourdi:** Honoraria from Amgen, UCB, Takeda; advisory board member for Amgen, UCB. **R Jain:** Research funding from the Amgen Foundation; honoraria ExtendBio; consultant for Ascendis Pharma. **L Rejnmark:** Research funding Takeda, Kyowa Kirin International, Ascendis Pharma, Calcilytix; honoraria Calcilytix Therapeutics; advisory board Takeda, Amolyt. **X An** was an employee of Ascendis Pharma during the trial and analysis. **A Smith, B Lai, J Ukena, C Sibley,** and **A Shu** are full-time employees of Ascendis Pharma.

Medical writing support was provided by Kristin Hirahatake, PhD, an employee of Ascendis Pharma, Inc.

TransCon PTH is investigational. Intended for education and scientific exchange only. Not for use in promotion or product commercialization.

Ascendis, Ascendis Pharma, Ascendis Pharma logo, the company logo and TransCon are trademarks owned by the Ascendis Pharma group.
© August 2023 Ascendis Pharma A/S.

MED-US-TC-PTH-2500142
November 2025