LB17

The Majority of Children With Pediatric Growth Hormone Deficiency Treated With Lonapegsomatropin for up to 6 Years Met or Exceeded Average Parental Height SDS: Final Results of enliGHten

Elpis Vlachopapadopoulou,[1] Paul S. Thornton,[2] Paul Hofman,[3] Ulhas M. Nadgir,[4] Paul Saenger,[5] Oleg Malievskiy,[6] Elena M. Aghajanova,[7] Maria Korpal-Szczyrska,[8] Meng Mao,[9] Sohair G. Abdelrahman,[9] Allison S. Komirenko,[9] Aimee D. Shu,[9] Aristides K. Maniatis[10]

[1]Children's Hospital. P. A. Kyriakou, Athens, Greece; [2]Cook Children's Medical Center, Fort Worth, TX, USA; [3]Liggins Institute, University of Auckland, Auckland, New Zealand; [4]Center of Excellence in Diabetes and Endocrinology, Sacramento, CA, USA; [5]NYU Langone Health, New York, NY, USA; [6]Bashkir State Medical University, Ufa, Russia; [7]Yerevan State Medical University, Yerevan, Armenia; [8]Klinika Pediatrii, Diabetologii i Endokrynologii Uniwersyteckie Centrum Kliniczne, Gdansk, Poland; [9]Ascendis Pharma, Palo Alto, CA, USA; [10]Rocky Mountain Pediatric Endocrinology, Centennial, CO, USA

BACKGROUND

- The lonapegsomatropin (SKYTROFA®; TransCon™ hGH) phase 3 trial program evaluated dosing, efficacy and safety outcomes in pediatric growth hormone deficiency (GHD) across a range of pubertal stages **(Figure 1)**
 - Lonapegsomatropin demonstrated safety and efficacy in both treatment-naïve and treatment-experienced children in the 52-week heiGHt trial[1] and 26-week fliGHt trial,[2] respectively
- The enliGHten open-label extension trial enrolled participants from heiGHt and fliGHt for longer term investigation of safety and efficacy including children transitioning into puberty (more advanced Tanner stages)[3]
- Lonapegsomatropin (SKYTROFA®; TransCon® hGH), a prodrug of somatropin, is approved for once-weekly treatment of pediatric GHD by the FDA (pediatric patients 1 year or older and weighing at least 11.5 kg),[4] and by the EC (pediatric patients aged 3 years to 18 years)[5] **(Figure 2)**

Figure 1. Lonapegsomatropin Phase 3 Clinical Trials Overview

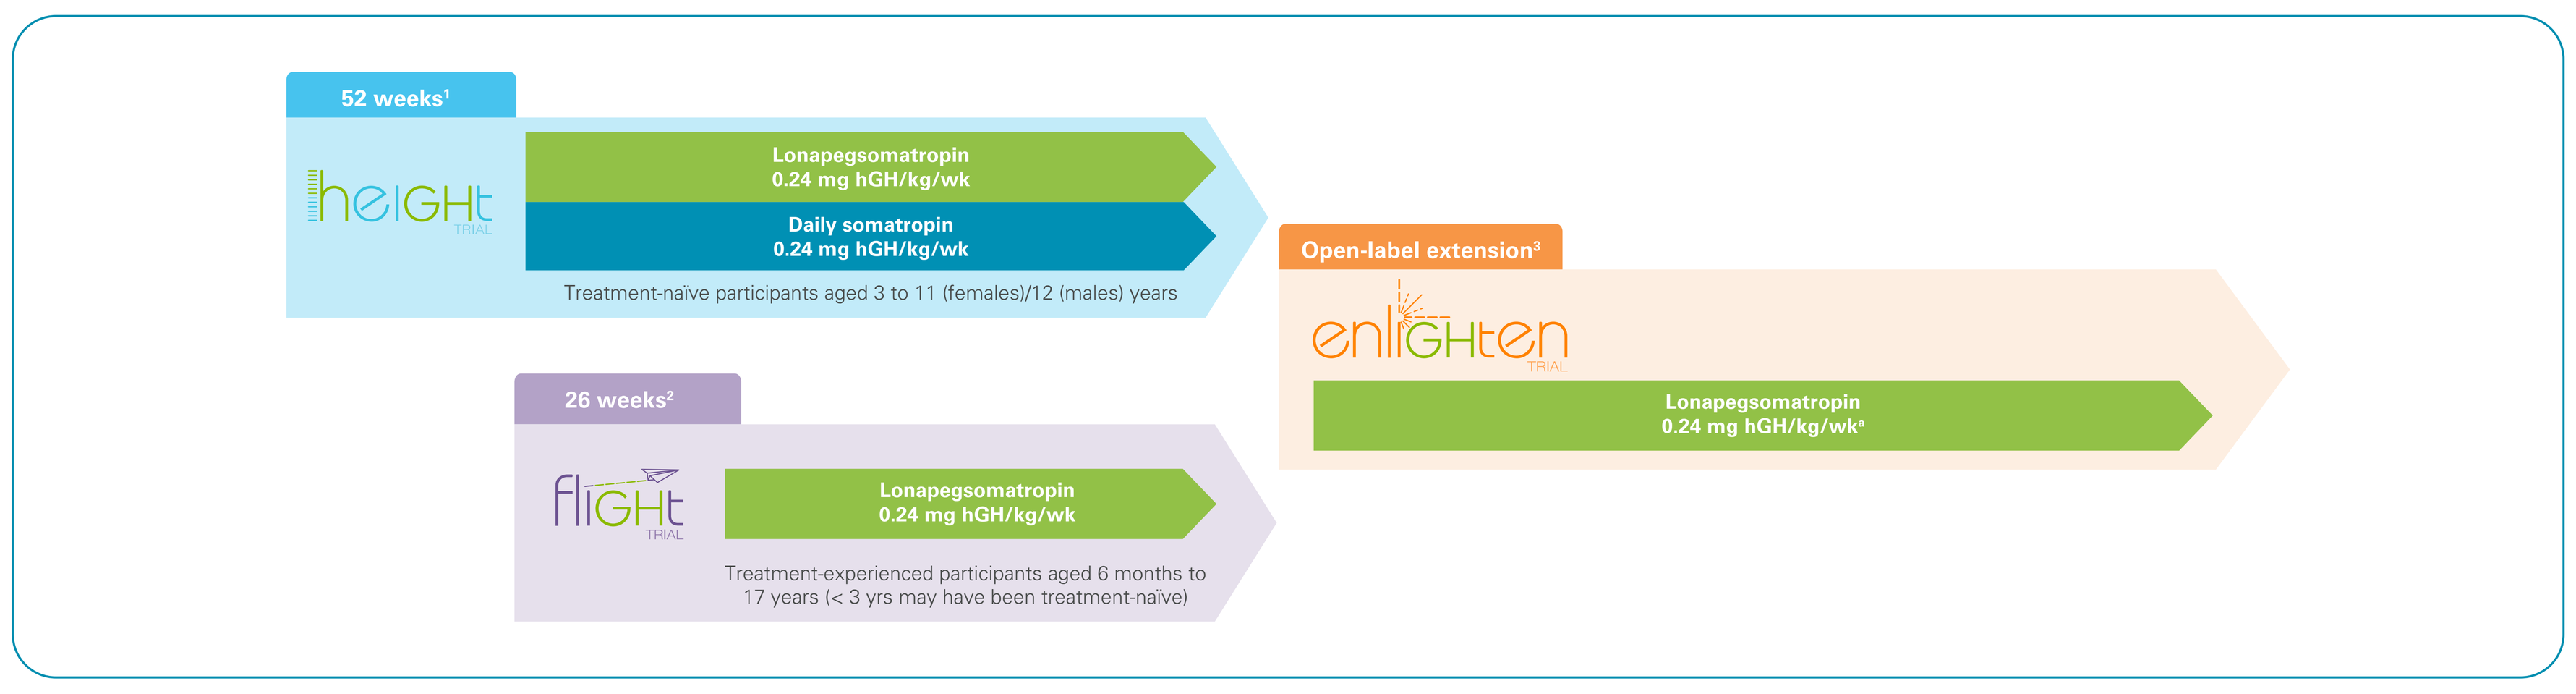

52 weeks[1]
heiGHt TRIAL
Lonapegsomatropin 0.24 mg hGH/kg/wk
Daily somatropin 0.24 mg hGH/kg/wk
Treatment-naïve participants aged 3 to 11 (females)/12 (males) years
Open-label extension[3]
enliGHten TRIAL
Lonapegsomatropin 0.24 mg hGH/kg/wk[a]
26 weeks[2]
fliGHt TRIAL
Lonapegsomatropin 0.24 mg hGH/kg/wk
Treatment-experienced participants aged 6 months to 17 years (< 3 yrs may have been treatment-naïve)

[a]Dose adjustments were allowed for weight change to next dosing bracket or to meet the goal of IGF-1 SDS between 0.0 and 2.0. If IGF-1 SDS was < 0.0 or > 2.0 (and clinically significant), dose could be increased or decreased to the next dose bracket, respectively, based on investigator or Medical Expert opinion.

Figure 2. Lonapegsomatropin (TransCon hGH) Design[3]

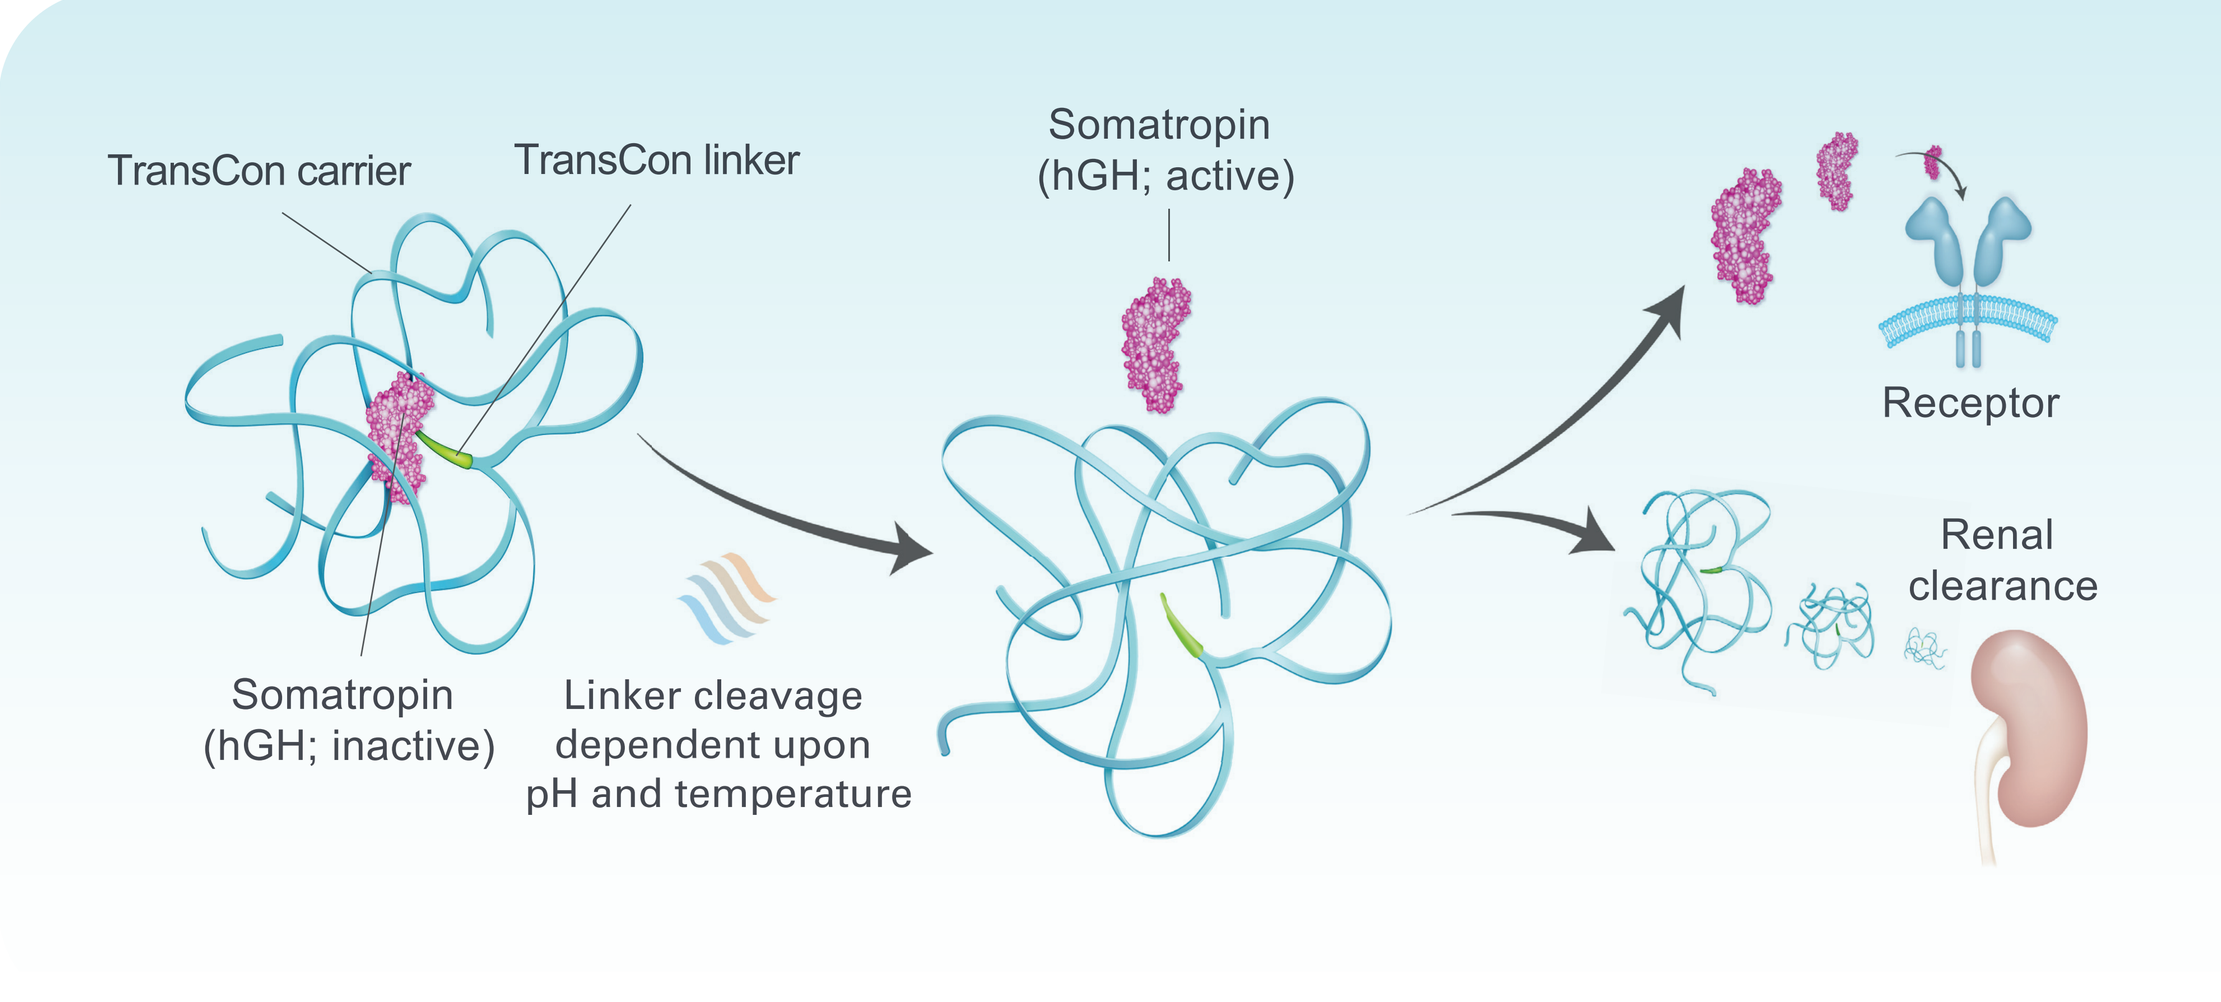

TransCon carrier
TransCon linker
Somatropin (hGH; active)
Receptor
Renal clearance
Somatropin (hGH; inactive)
Linker cleavage dependent upon pH and temperature

- Lonapegsomatropin is a prodrug of somatropin, administered once-weekly and designed to provide sustained release of active, unmodified somatropin[3,6]
- The unmodified, unbound somatropin released from lonapegsomatropin has the identical 191 amino-acid sequence and size (22 kDa) as endogenous growth hormone[3,6]

METHODS

OUTCOMES

- The primary objectives of enliGHten were to evaluate the long-term safety and efficacy of treatment with lonapegsomatropin
- enliGHten enrolled 298 participants from heiGHt and fliGHt
 - The full safety population includes all participants who enrolled in either heiGHt or fliGHt (n = 306)
- Results are reported through the participants' final visit (study end date: December 31, 2022), focusing on the subset (n = 81) that completed lonapegsomatropin treatment because it was determined by the investigator that treatment for pediatric GHD was no longer necessary ("treatment completers")

RESULTS

Table 1. enliGHten Baseline[a] Demographics

	Full Analysis Set (N = 298)	Treatment Completers (n = 81)
Age, years		
Mean (SD)	10.3 (3.4)	13.3 (1.8)
Min, max	1.7, 17.8	8.8, 17.4
Sex, male, n (%)	235 (78.9)	66 (81.5)
Height SDS, mean (SD)	-1.6 (0.9)	-1.6 (0.7)
Δ Average parental height SDS, mean (SD)[b]	-1.8 (1.2)	-1.2 (0.9)
IGF-1 SDS, mean (SD)	1.0 (1.3)	0.8 (1.5)
Tanner Stage, n (%)		
Stage I	214 (71.8)	22 (30.1)
Stage II	40 (13.4)	17 (23.3)
Stage III	25 (8.4)	28 (38.4)
Stage IV	16 (5.4)	6 (8.2)
Stage V	3 (1.0)	0

The trial of origin for treatment completers were:

- heiGHt (lonapegsomatropin arm), n = 8
- heiGHt (daily somatropin arm), n = 10
- fliGHt (lonapegsomatropin), n = 63

[a]Baseline indicates last non-missing measurement before first dose of lonapegsomatropin.
[b]Δ average parental height SDS is the difference between the patient's height SDS and the average parental height SDS where average parental height SDS = (height SDS^{mother} + height SDS^{father})/2.

Figure 3. Height SDS by Visit for Treatment Completers

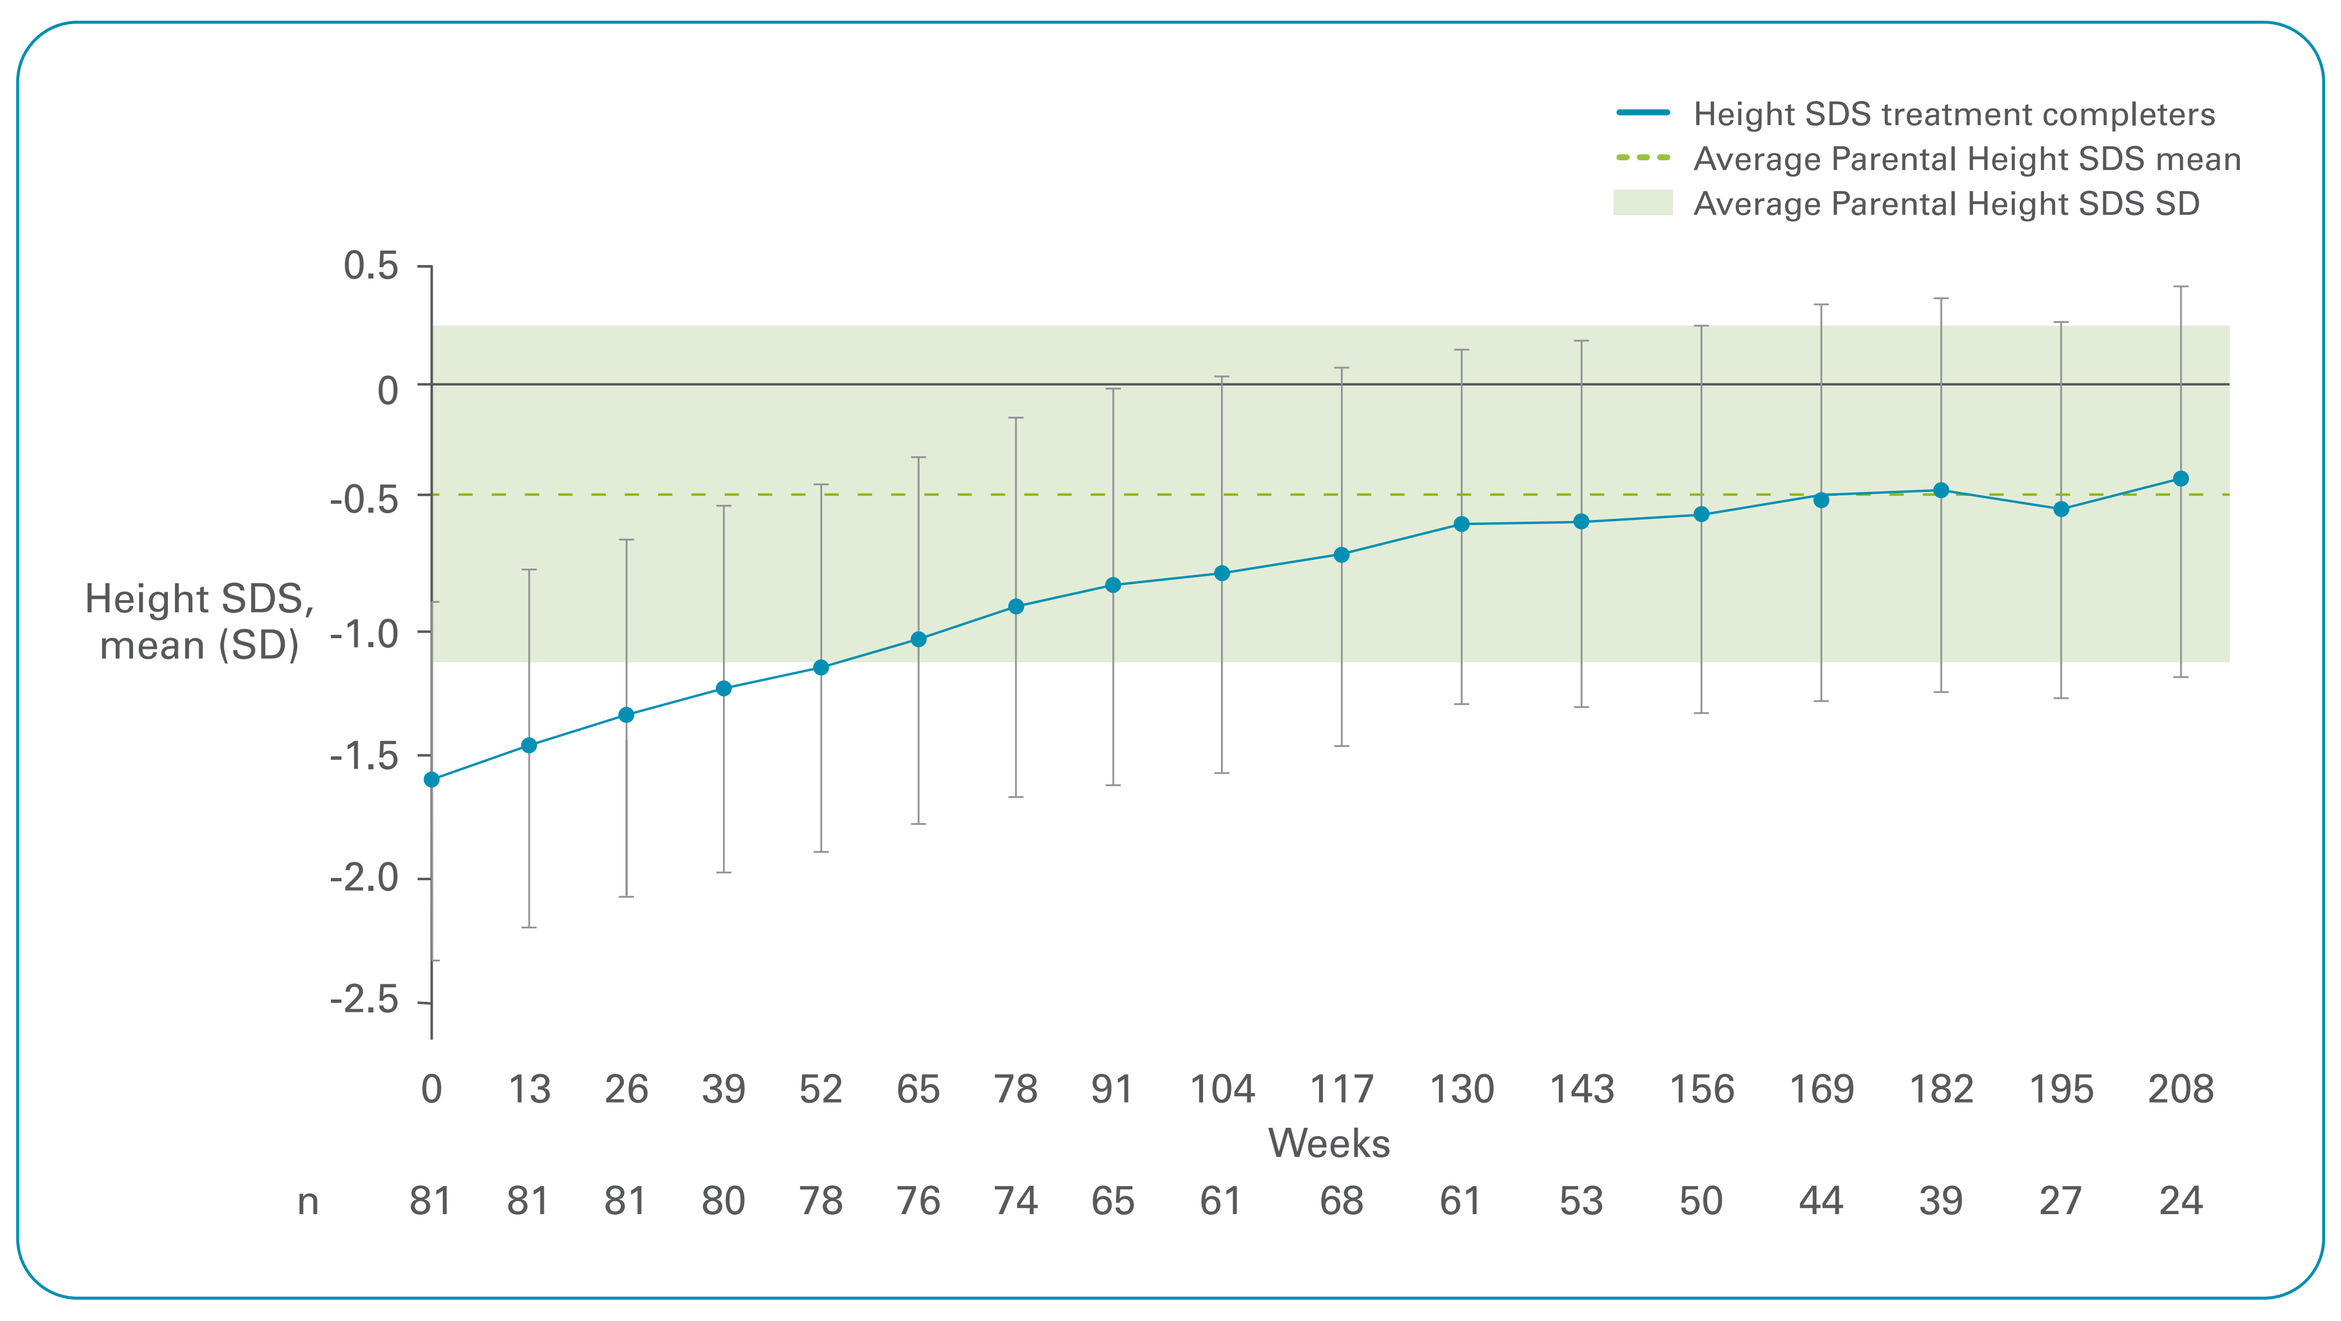

Height SDS treatment completers
Average Parental Height SDS mean
Average Parental Height SDS SD
Height SDS, mean (SD)
0.5
0
-0.5
-1.0
-1.5
-2.0
-2.5
0 13 26 39 52 65 78 91 104 117 130 143 156 169 182 195 208
Weeks
n 81 81 81 80 78 76 74 65 61 68 61 53 50 44 39 27 24

Figure 4. Average IGF-1 SDS by Visit for Treatment Completers

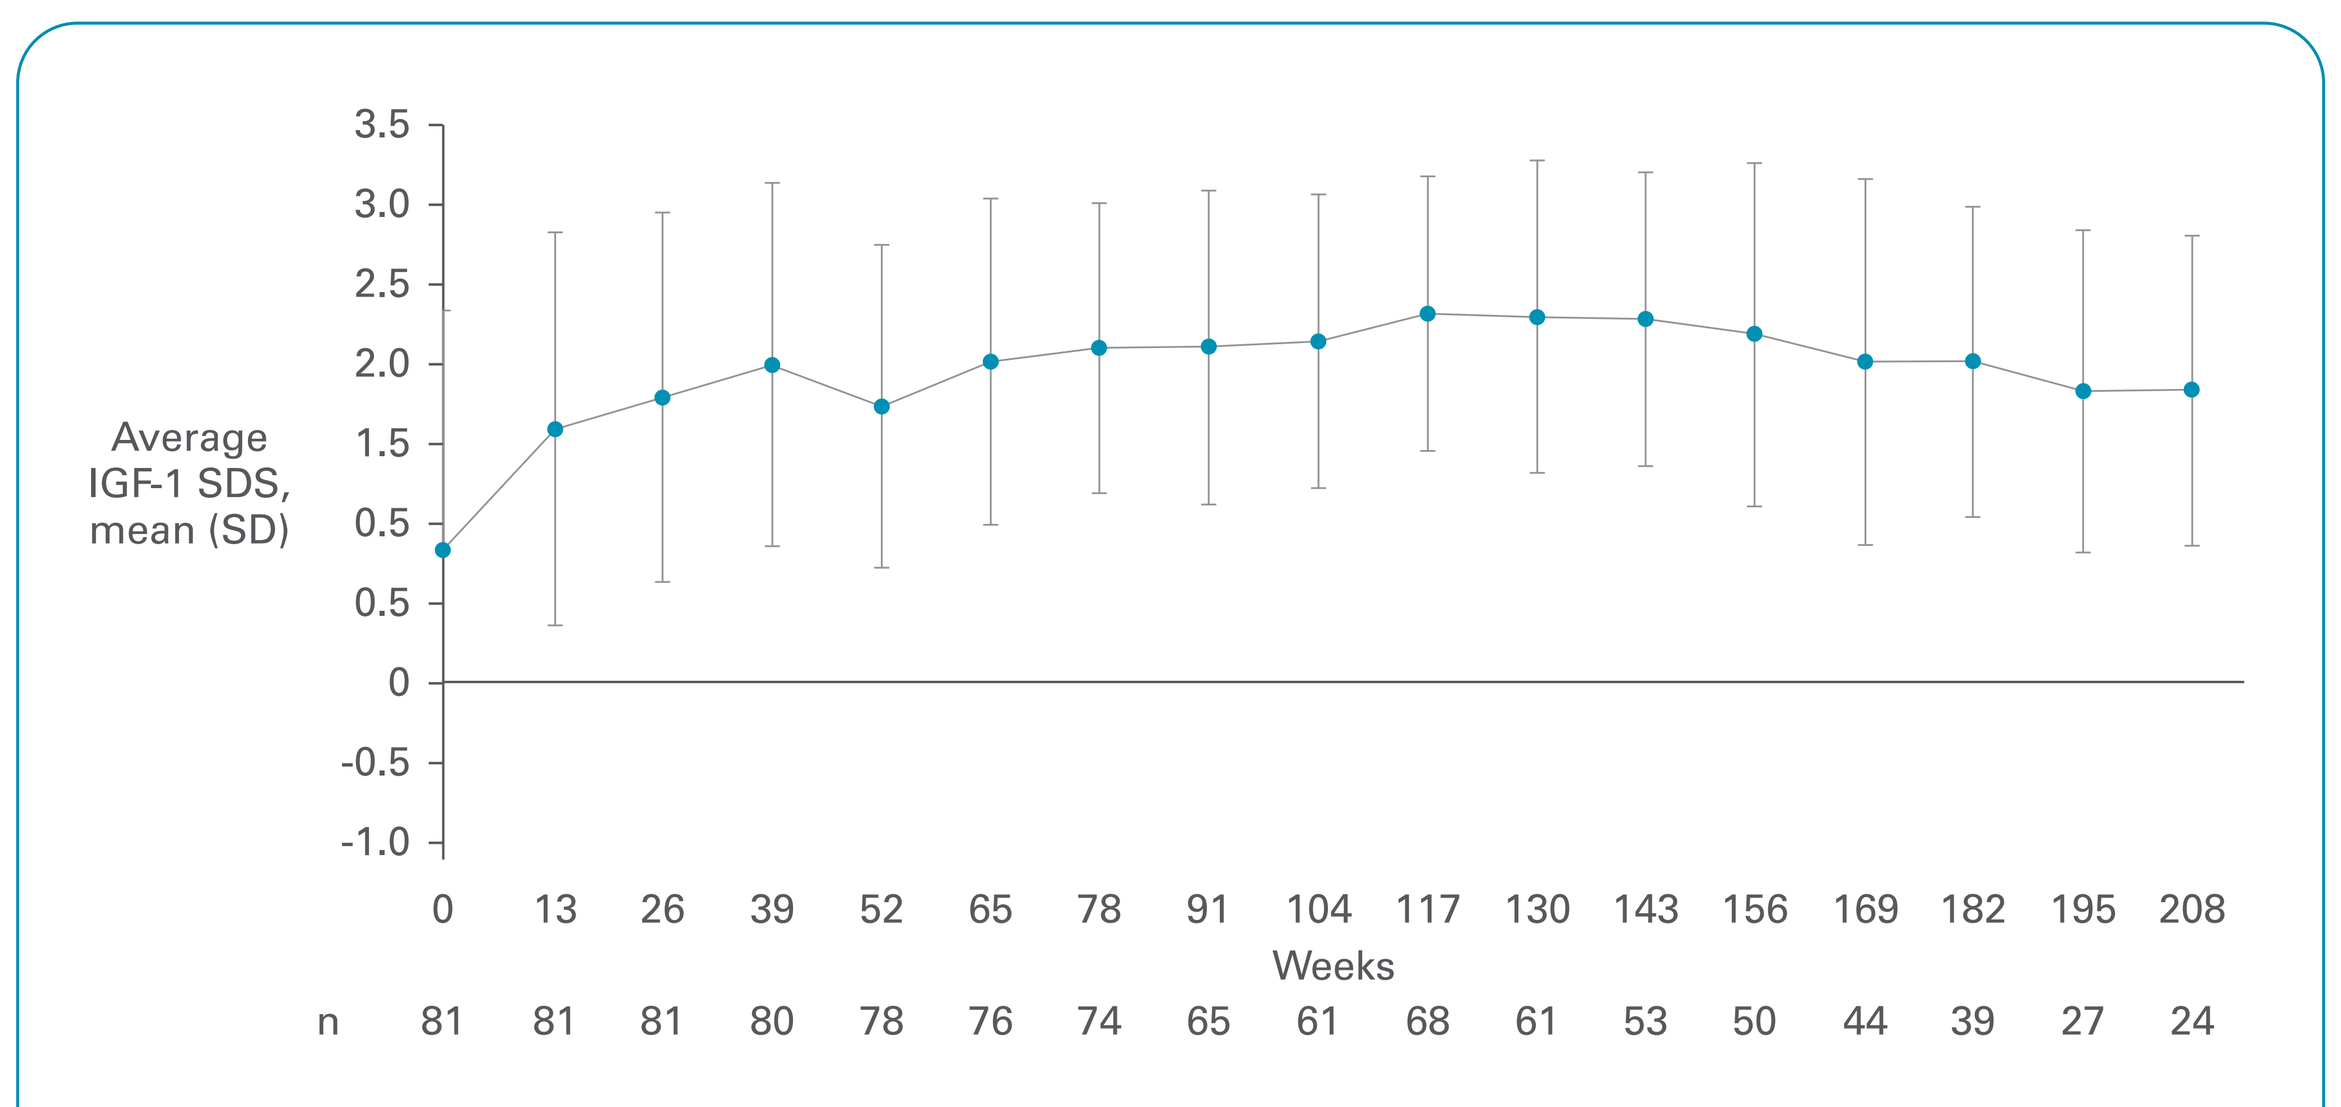

Average IGF-1 SDS, mean (SD)
3.5
3.0
2.5
2.0
1.5
0.5
0.5
0
-0.5
-1.0
0 13 26 39 52 65 78 91 104 117 130 143 156 169 182 195 208
Weeks
n 81 81 81 80 78 76 74 65 61 68 61 53 50 44 39 27 24

- IGF-1 SDS remained stable over time in a population that included older participants in more advanced Tanner stages

Table 2. Key Data at Last Visit

Growth Outcomes for Treatment Completers at Last Visit	Treatment Completers (n = 81)
Height SDS, mean (SD)	-0.4 (0.7)
Δ Average parental height SDS, mean (SD)[a]	-0.1 (0.7)
% met or exceeded average parental height	59.3
Additional Data for Treatment Completers at Last Visit	**Treatment Completers (n = 81)**
Age, years, mean (SD)	16.5 (1.4)
Min, max	13.0, 18.6
Female (n = 15), mean (SD)	14.5 (1.0)
Male (n = 66), mean (SD)	16.8 (1.0)
Bone age, years, mean (SD)	15.7 (1.3)
Min, max	13.0, 17.0
Female (n = 15), mean (SD)	14.2 (0.8)
Male (n = 66), mean (SD)	16.0 (1.1)
Duration of lonapegsomatropin Treatment, mean (SD)	3.2 (1.1)
Min, max	0.5, 5.3
Average IGF-1 SDS, mean (SD)	1.8 (1.1)
Tanner Stage, n (%)	
Stage I	0
Stage II	0
Stage III	0
Stage IV	23 (28.4)
Stage V	58 (71.6)

[a]Δ average parental height SDS is the difference between the patient's height SDS and the average parental height SDS where average parental height SDS = (height SDS^{mother} + height SDS^{father})/2.

- The majority of treatment completers (59.3%) had met or exceeded average parental height at last visit and all were in Tanner stage IV or V

Figure 5. TEAEs for Treatment Completers

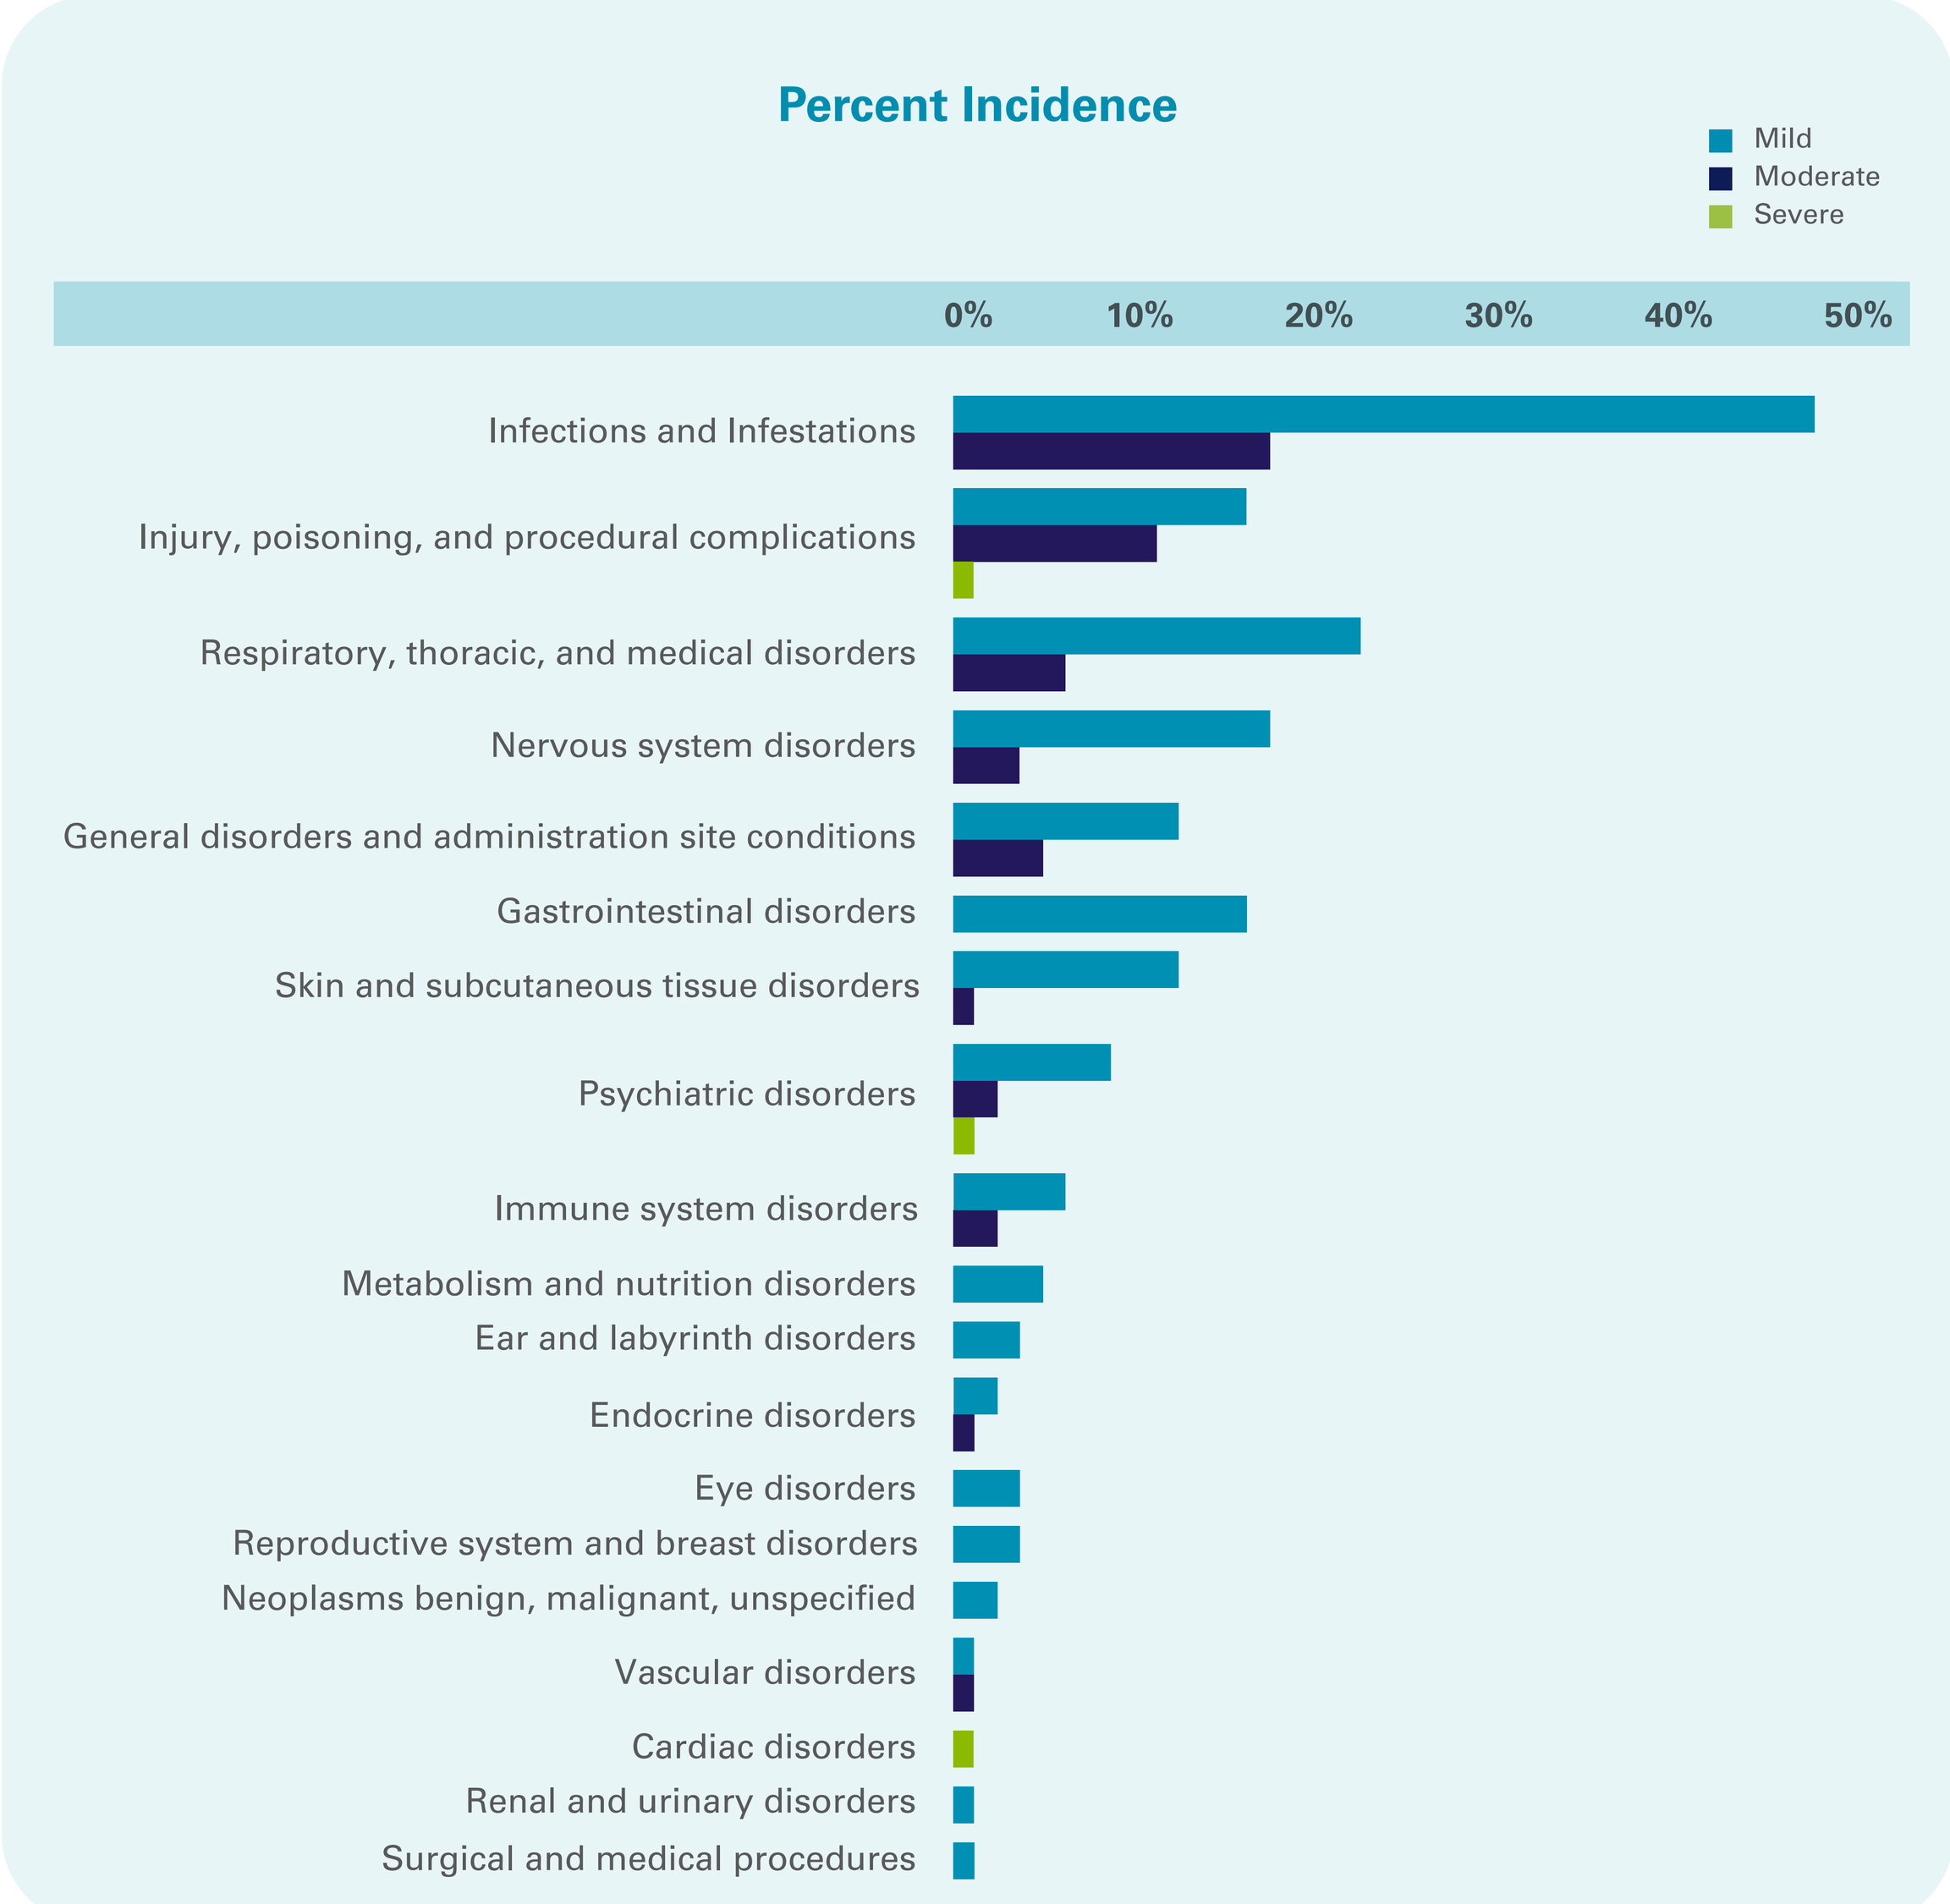

Percent Incidence
Mild
Moderate
Severe
0% 10% 20% 30% 40% 50%
Infections and Infestations
Injury, poisoning, and procedural complications
Respiratory, thoracic, and medical disorders
Nervous system disorders
General disorders and administration site conditions
Gastrointestinal disorders
Skin and subcutaneous tissue disorders
Psychiatric disorders
Immune system disorders
Metabolism and nutrition disorders
Ear and labyrinth disorders
Endocrine disorders
Eye disorders
Reproductive system and breast disorders
Neoplasms benign, malignant, unspecified
Vascular disorders
Cardiac disorders
Renal and urinary disorders
Surgical and medical procedures

Table 3. Adverse Event Summary in Treatment Completers

	n = 81
Treatment emergent adverse events (TEAE)	67 (82.7)
Related TEAE	7 (8.6)
Serious TEAEs	2 (2.5)
Serious Related TEAEs	0
Severe TEAEs	3 (3.7)
Severe Related TEAEs	0
TEAEs leading to discontinuation of study drug	0

- There were no TEAEs that led to discontinuation of study drug in treatment completers (n = 81) or in the full safety population (n = 306)

Table 4. Overview of Outcomes for Full enliGHten Population at Final Visit

	n = 298
Lonapegsomatropin treatment duration, years, mean (SD)	4.1 (1.1)
Min, max	0.5, 6.0
Duration 4 - < 5 years, n (%)	140 (47.0)
Duration 5 - < 6 years, n (%)	52 (17.5)
% met or exceeded average parental height	53.4
Tanner Stage, n (%)	
Stage I	89 (29.9)
Stage II	32 (10.7)
Stage III	28 (9.4)
Stage IV	63 (21.1)
Stage V	86 (28.9)

CONCLUSIONS

- Long-term safety was demonstrated in participants treated with lonapegsomatropin for pediatric GHD for up to 6 years
- The majority of all participants (n = 298), including those who by study end date had completed treatment with lonapegsomatropin (n = 81), met or exceeded average parental height SDS without a mean dose increase over time

ABBREVIATIONS

GHD = growth hormone deficiency; **hGH** = human growth hormone; **SD** = standard deviation; **SDS** = standard deviation score; **TEAE** = treatment-emergent adverse event

REFERENCES:

1. Thornton PS, Maniatis AK, Aghajanova E, et al. Weekly Lonapegsomatropin in Treatment-Naive Children With Growth Hormone Deficiency: The Phase 3 heiGHt Trial. *J Clin Endocrinol Metab.* 2021;106(11):3184-3195. **2.** Maniatis AK, Nadgir U, Saenger P, et al. Switching to Weekly Lonapegsomatropin from Daily Somatropin in Children with Growth Hormone Deficiency: The fliGHt Trial. *Horm Res Paediatr.* 2022;95(3):233-243. **3.** Maniatis AK, Casella SJ, Nadgir UM, et al. Safety and Efficacy of Lonapegsomatropin in Children With Growth Hormone Deficiency: enliGHten Trial 2-Year Results. *J Clin Endocrinol Metab.* 2022;107(7):e2680-e2689. **4.** SKYTROFA. US Prescribing Information (USPI). Ascendis Pharma; 2022. Accessed March 29, 2023. https://ascendispharma.us/products/pi/skytrofa/skytrofa_pi.pdf. **5.** SKYTROFA. SmPC. EPAR product information. EMA. https://www.ema.europa.eu/en/medicines/human/EPAR/skytrofa-previously-lonapegsomatropin-ascendis-pharma. Accessed March 29, 2023. **6.** Sprogoe K, Mortensen E, Karpf DB, Leff JA. The rationale and design of TransCon Growth Hormone for the treatment of growth hormone deficiency. *Endocr Connect.* 2017;6(8):R171-R181.

DISCLOSURES AND FUNDING

This study was sponsored by Ascendis Pharma Endocrinology Division A/S.

EV: Research Funding: Ascendis Pharma, OPKO, and Janssen; Honoraria: Sandoz, Eli Lilly, and Novo Nordisk.

PH: Honoraria: Novo Nordisk and Eli-Lilly.

PST: Research Funding: Ascendis, Pfizer, Opko, Zealand, Rezolute, Spruce; Expert Testimony: Johnson and Johnson; Travel, Accomodations, Expenses; Rezolute.

EMA: Travel, Accommodations, Expenses: Berlin Hemi Menarini pharmaceutical company, conference attendance.

MKS: Research Funding: Ascendis Pharma.

AKM: Honoraria: Ascendis, Novo Nordisk, Pfizer; Advisory Board: Ascendis, Novo Nordisk, Pfizer; Consulting: Ascendis, Novo Nordisk; Speaker Bureau: Ascendis, Novo Nordisk; Research Funding: Ascendis, Novo Nordisk, OPKO, Pfizer.

UMN, PS: Nothing to disclose.

MM, CZ, SGA, ASK, and **ADS** are employees of Ascendis Pharma Inc.

Medical writing support was provided by Cindy Gode, PhD, an employee of Ascendis Pharma, Inc.

Ascendis, Ascendis Pharma, Ascendis Pharma logo, the company logo and TransCon are trademarks owned by the Ascendis Pharma group. © September 2023 Ascendis Pharma A/S

MED-US-TC-PGHD-2500029
November 2025