Safety and Effectiveness of Lonapegsomatropin in the SkybriGHt Registry Study: Analysis of the First 100 Patients Enrolled With 1 Year of Follow-Up

Mako Sather,[1] Aristides Maniatis,[1] M Jennifer Abuzzahab,[2] Steven Chernausek,[3] Terri Lipman,[4] Daniel Flynn,[5] Gnanagurudasan Prakasam,[6] Alan Rogol,[7] Eric Huang,[8] Carol Zhao,[8] Philippe Backeljauw[9]

[1]Rocky Mountain Pediatric Endocrinology, PC, Centennial, CO, USA; [2]Children's Minnesota, Minneapolis, MN, USA; [3]University of Oklahoma Health Sciences Center, Oklahoma City, OK, USA; [4]University of Pennsylvania School of Nursing, Philadelphia, PA, USA; [5]St Luke's Boise Medical Center, Boise, ID, USA; [6]Sutter Memorial Hospital, Sacramento, CA, USA; [7]University of Virginia, Charlottesville, VA, USA; [8]Ascendis Pharma, Inc, Palo Alto, CA, USA; [9]Cincinnati Children's Hospital Medical Center, Cincinnati, OH, USA

2

Background

Lonapegsomatropin (SKYTROFA®; TransCon® hGH), a prodrug of somatropin, is approved for the once-weekly treatment of pediatric growth hormone deficiency (GHD) by the Food and Drug Administration (pediatric patients 1 year or older and weighing at least 11.5 kg),[1] and by the European Commission (pediatric patients aged 3 years to 18 years).[2]

The lonapegsomatropin phase 3 clinical trial program evaluated efficacy and safety outcomes in children with GHD.[3,4,5]

- Lonapegsomatropin demonstrated efficacy and safety in children with GHD who were treatment-naive in the 52-week heiGHt trial[3] and treatment-experienced in the 26-week fliGHt trial[4]
- The enliGHten open-label extension trial enrolled children from heiGHt and fliGHt to further evaluate long-term efficacy, safety, and tolerability, including children transitioning into puberty (more advanced pubertal stages)[5,6]

Real-world data extend our understanding of clinical trial findings by providing insights into disease management, treatment utilization, and long-term outcomes in routine clinical practice.

Aims

The objectives of the SkybriGHt study are to assess the safety and effectiveness of lonapegsomatropin in the post-marketing setting. We report outcomes in the first 100 enrolled patients who have reached 1-year follow-up.

Conclusions

This is the first prospective study evaluating real-world clinical experience with lonapegsomatropin treatment for pediatric GHD.

Among the first 100 patients enrolled with 12 months follow-up, the median and maximum lonapegsomatropin treatment duration at 1 year was 1.75 and 7.8 years, respectively.

Patient demographics reflected:

- A diverse age distribution
- 82% male, consistent with growth hormone (GH) prescription patterns[7]
- 59% of patients were previously treated with daily GH

Median height standard deviation score (SDS) increased over time and exceeded zero (CDC population mean height for age and sex)[8] at month 12 (0.207); median annualized height velocity (AHV) at month 12 was 7.82 cm/year.

IGF-I SDS remained stable over the 12 months analyzed (median change from enrollment at months 6 and 12 was 0).

There were no serious, serious related, severe, or severe related treatment-emergent adverse events (TEAEs), and none leading to treatment discontinuation.

Clinical Implication

At 1 year, data for the first 100 patients enrolled in the SkybriGHt registry demonstrated safety and effectiveness aligned with prior trial reports of lonapegsomatropin treatment for pediatric GHD.

Methods

SkybriGHt Registry Study

- SkybriGHt aims to enroll 900 participants from the United States
- Patients who are prescribed lonapegsomatropin consistent with routine clinical practice are eligible
- Decision to treat with lonapegsomatropin is made prior to, and independently of, any potential study participation
- Enrollment will occur over a 5-year period with the study intended to conclude after a total of 10 years
- Clinical outcomes are evaluated through height SDS, annualized height velocity (AHV), and insulin-like growth factor I (IGF-I) SDS
- Safety is evaluated through adverse event (AE) reporting and laboratory test results

Statistical Analyses

- Data cut-off: October 15, 2024
- Analyses were based on the first 100 patients who had reached 12 months of follow-up after enrollment
 - Patients had to have the date of 1st dose entered in the electronic data capture (EDC) system
 - First 100 patients based on the chronological order of consent date
- Descriptive statistics were summarized for:
 - Demographics and baseline disease characteristics, prior GH therapy, and lonapegsomatropin treatment
 - Clinical outcomes assessed at enrollment and at 6 and 12 months following enrollment, with a 3-month window applied for the 6- and 12-month time points
 - AEs, including those that were serious, treatment-related, severe, or resulted in treatment discontinuation

Figure 1. **SkybriGHt Registry Study Design**

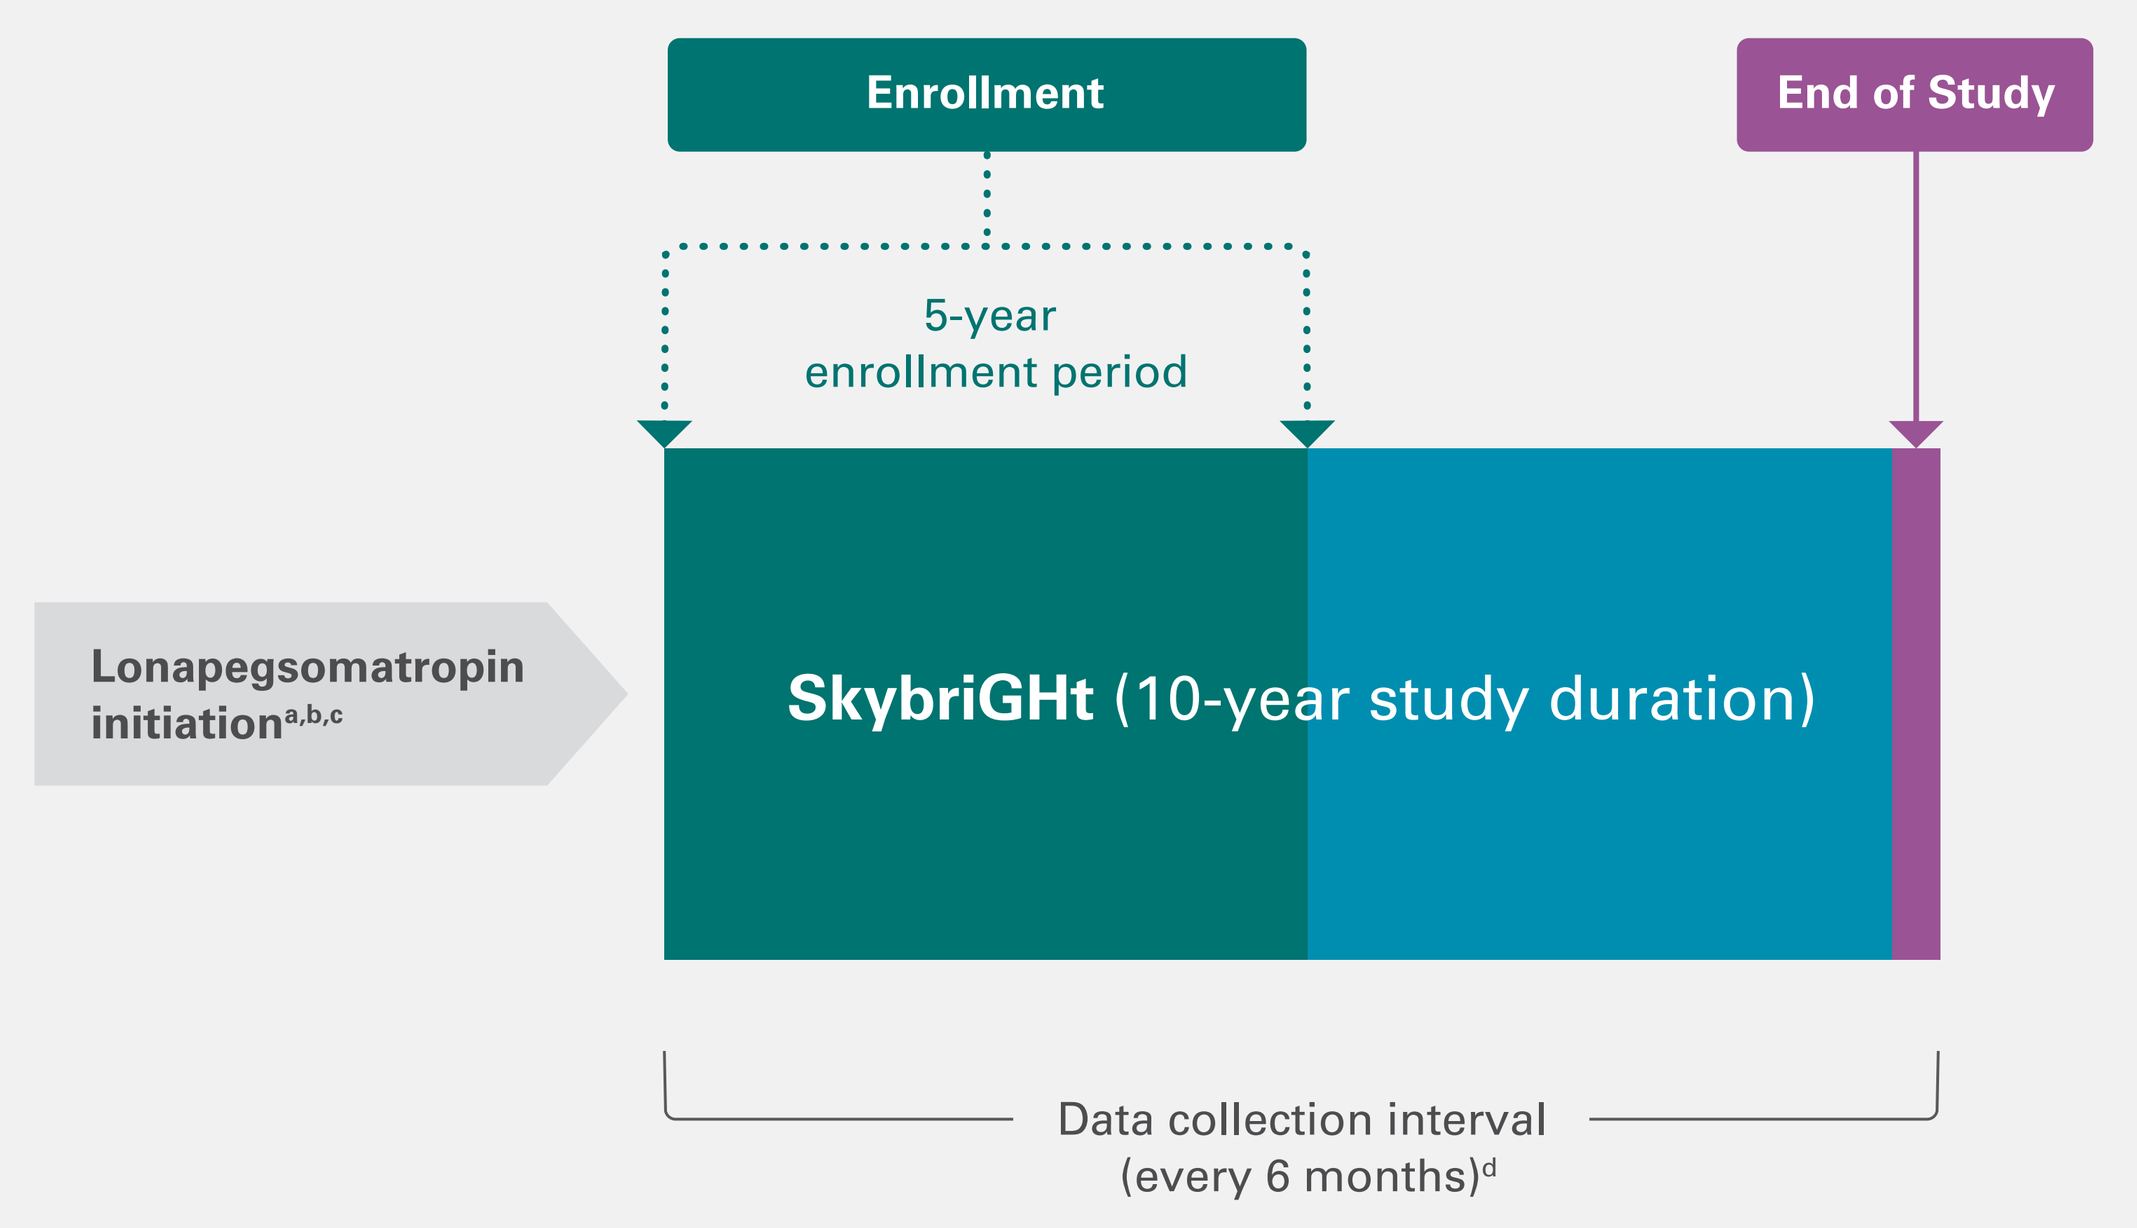
Enrollment
End of Study
5-year
enrollment period
Lonapegsomatropin initiation[a,b,c]
SkybriGHt (10-year study duration)
Data collection interval
(every 6 months)[d]

[a]**Lonapegsomatropin initiaion**

- Lonapegsomatropin treatment may have been initiated at varying times and of varying duration prior to enrollment
- Patients may have participated in lonapegsomatropin clinical trials prior to SkybriGHt
- Initiation of lonapegsomatropin may have been preceded by other GH therapies such as daily somatropin or another long-acting growth hormone

[b]**Data collection interval**

- Included baseline and medical history at enrollment and data entry for outcomes and AEs every 6 months following enrollment

Results

Table 1. **Disposition**

Variable	Number of Patients (%)
All patients enrolled	100
Initiated lonapegsomatropin > 60 days prior to enrollment	86 (86)
Participated in heiGHt, fliGHt, and/or enliGHten clinical trial(s)	17 (17)
Withdrew from treatment[a]	17 (17)

[a]Reasons included: reached adult height/bone age maturation, change in insurance coverage, withdrawal by patient, other.

Table 2. **Demographics**

Variable	Category	Total (N = 100%)
Sex, n (%)	Male	82 (82)
	Female	18 (18)
Age, years	Median	10.0
	Q1, Q3	8.0, 12.0
Age category, n (%)	< 3 years	0
	≥ 3 and < 6 years	5 (5)
	≥ 6 to < 11 years for girls, ≥ 6 to < 12 years for boys	47 (47)
	≥ 11 years for girls, ≥ 12 years for boys	48 (48)
Race, n (%)	White	86 (86)
	Asian	6 (6)
	Not reported	4 (4)
	Black or African American	3 (3)
	Other	1 (1)
Ethnicity, n (%)	Not Hispanic or Latino	88 (88)
	Hispanic or Latino	9 (9)
	Unknown	2 (2)
	Not reported	1 (1)
Peak stimulated GH at diagnosis, n (%)[a]	≤ 3 ng/mL	4 (6)
	> 3 ng/mL to ≤ 7 ng/mL	37 (56)
	> 7 ng/mL[b]	25 (38)

All 100 participants in the analysis set had a diagnosis of pediatric GHD.
[a]Percentages were calculated based on patients with available test results (n = 66).
[b]All participants were < 10 ng/mL except for 1 patient at 10.2 ng/mL.

Figure 2. **Prior Growth Hormone Therapy**

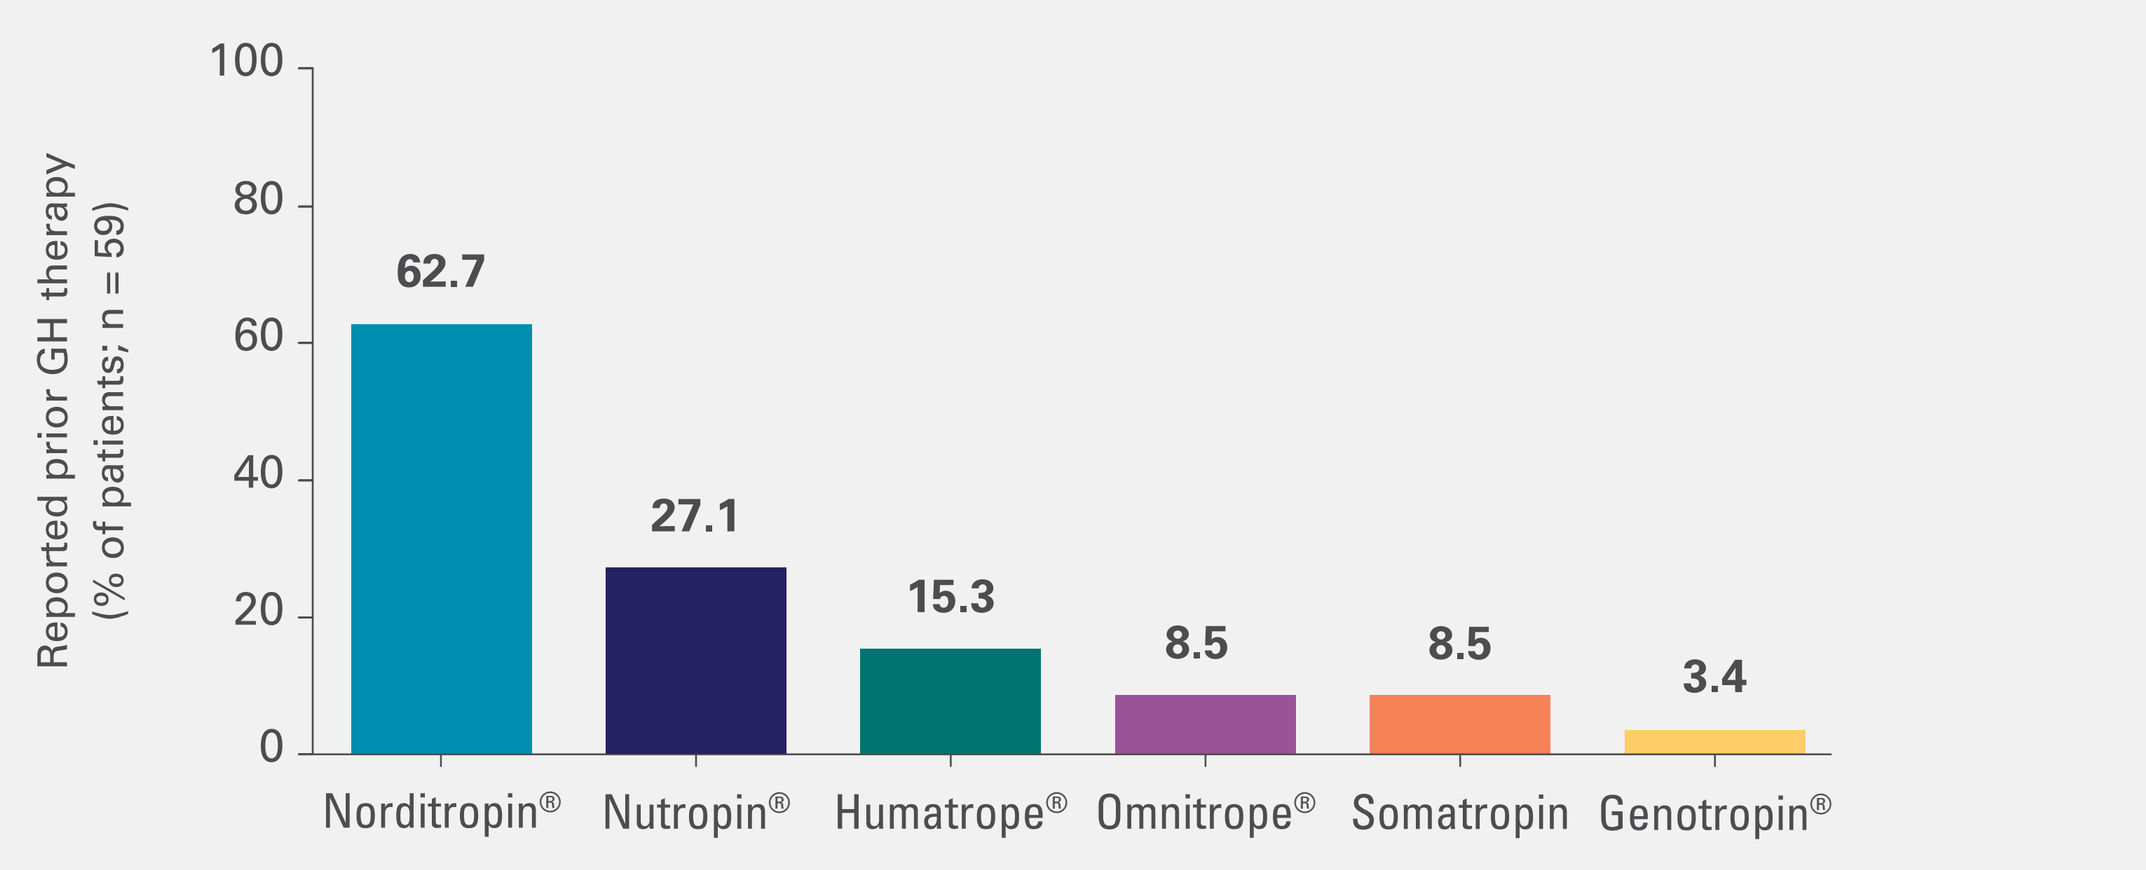
Reported prior GH therapy
(% of patients; n = 59)
100
80
60
40
20
0
62.7
27.1
15.3
8.5
8.5
3.4
Norditropin®
Nutropin®
Humatrope®
Omnitrope®
Somatropin
Genotropin®

- 59% of participants had been previously treated with daily GH
 - The median (Q1, Q3) prior daily GH dose at SkybriGHt enrollment was 0.31 (0.30, 0.31) mg hGH/kg/week and the median (Q1, Q3) duration of prior treatment was 9.1 (4.3, 13.5) months
- No participants had been previously treated with another long-acting GH

Somatropin indicates brand not specified. Patients could have received multiple brands of GH, and therefore, the summed percentages exceed 100%.

Table 3. **Lonapegsomatropin Exposure**

Variable	Category	Total (N = 100)
Duration of treatment[a] **(weeks)**	n	100
	Median	91.0
	Min, Max	56.0, 408.0
Initial prescribed[b] **dose (mg hGH/kg/week)**	n	77
	Median	0.23
	Min, Max	0.12, 0.36
Prescribed[c] **dose at Month 6 (mg hGH/kg/week)**	n	54
	Median	0.24
	Min, Max	0.15, 0.37
Prescribed[c] **dose at Month 12 (mg hGH/kg/week)**	n	28
	Median	0.26
	Min, Max	0.12, 0.37

[a]Total duration of treatment since first dose of lonapegsomatropin.
[b]Dose as recorded in patient's chart at time of first dose of lonapegsomatropin.
[c]Average prescribed dose within ±3-month window of each timepoint.

Figure 3. **Height SDS**

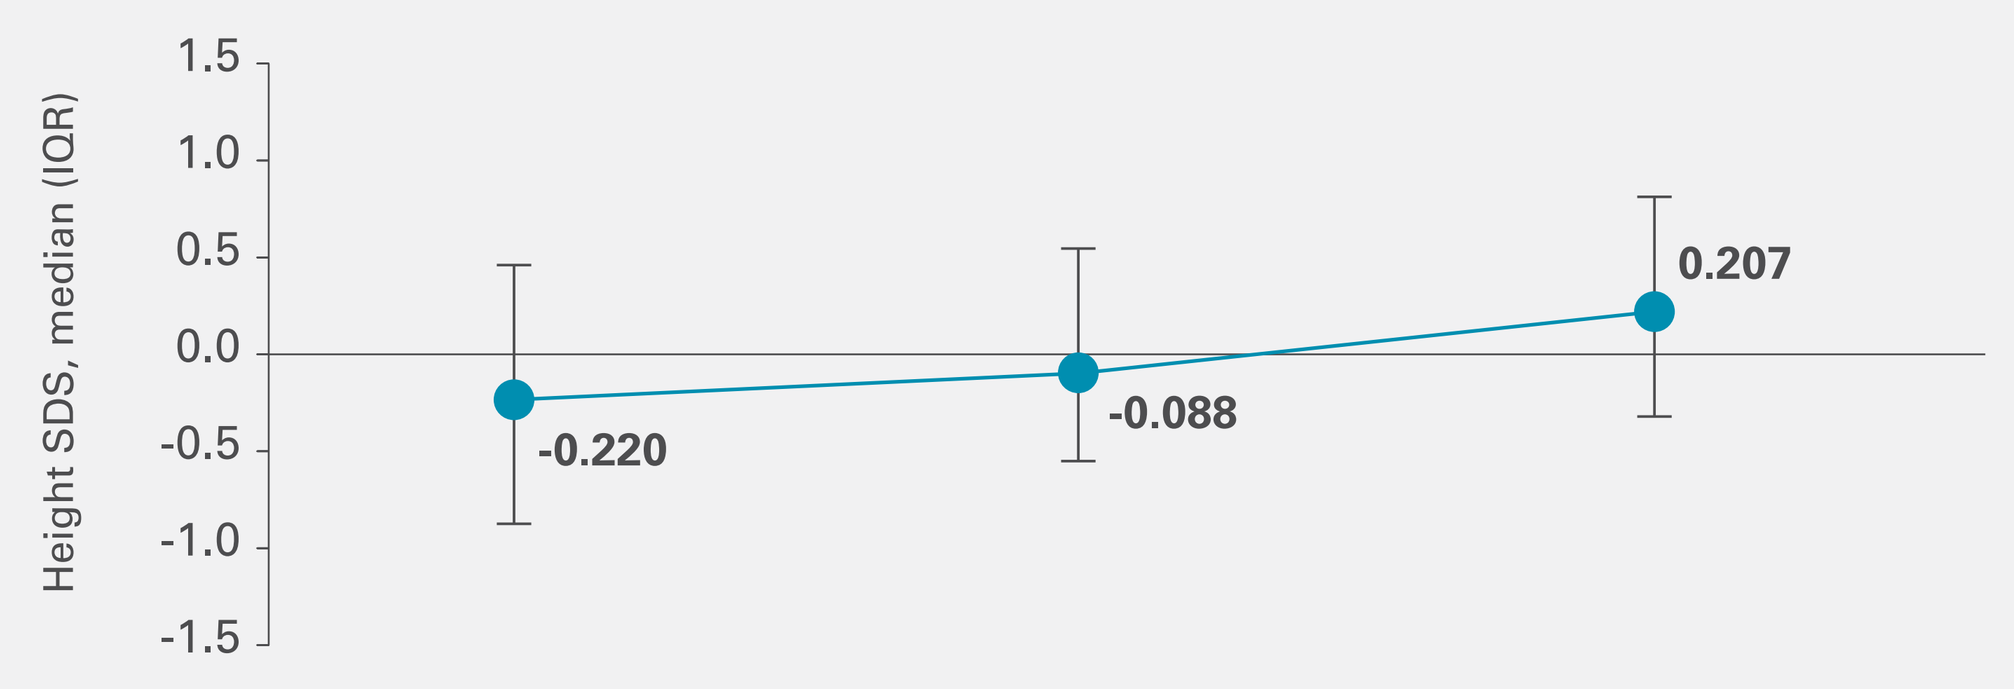
Height SDS, median (IQR)
1.5
1.0
0.5
0.0
-0.5
-1.0
-1.5
-0.220
-0.088
0.207

Months	0	6	12
n[a]	100	89	73
Δ height SDS[b]		0.105	0.204

[a]At each post-enrollment visit, n is the number of patients with available measurement.
[b]Δ height SDS is the change from enrollment, presented as median.

Table 4. **Annualized Height Velocity at 12 Months Following Enrollment**

Variable	Category	Total (N = 100)
AHV at month 12 (cm/year)	n	73
	Median	7.82
	Q1, Q3	6.54, 8.81

Table 5. **IGF-I SDS**

Variable	Category	Total (N = 100)
Enrollment IGF-I SDS[a]	n	67
	Median	1.30
	Q1, Q3	0.80, 1.70
Month 6 IGF-I SDS	n	57
	Median	1.30
	Q1, Q3	0.80, 1.90
Month 12 IGF-I SDS	n	43
	Median	1.20
	Q1, Q3	0.50, 1.70
Median (Q1, Q3) IGF-I SDS change from enrollment	Month 6	0 (-0.60, 0.60)
	Month 12	0 (-0.80, 0.30)

[a]Timing of IGF-I sample collection was variable and may not always represent the average weekly IGF-I. IGF-I SDS reflects value reported at the site in EDC.

Table 6. **Adverse Event Summary**

Variable	Total (N = 100)
Treatment-Emergent Adverse Events (TEAEs), n (%)	26 (26)
Related TEAEs,[a] **n (%)**	10 (10)
Serious TEAEs, n (%)	0
Serious related TEAEs, n (%)	0
Severe TEAEs, n (%)	0
Severe related TEAEs, n (%)	0
TEAEs leading to discontinuation of study drug, n (%)	0

[a]Related TEAEs included: insulin-like growth factor increased, injection site pain, and pain in extremity.

Table 7. **Adverse Events by System Organ Class**

Variable	Total (N = 100)
Patients with at least one TEAE, n (%)	26 (26)
Infections and infestations	10 (10)
Investigations	9 (9)
Injury, poisoning, and procedural complications	5 (5)
Nervous system disorders	4 (4)
General disorders and administration site conditions	2 (2)
Musculoskeletal and connective tissue disorders	1 (1)
Respiratory, thoracic and mediastinal disorders	1 (1)

TEAEs occurring in ≥ 5% of patients by system organ class included: Infections and infestations (bronchitis, otitis media, upper respiratory tract infection, cellulitis, conjunctivitis, gastroenteritis, nasopharyngitis, and pneumonia); Investigations (insulin-like growth factor increased); Injury, poisoning, and procedural complications (head injury, limb injury, skin laceration, torus fracture, wrist fracture).

Abbreviations

AE = adverse event; **AHV** = annualized height velocity; **CDC** = Centers for Disease Control and Prevention; **EDC** = electronic data capture; **GH** = growth hormone; **GHD** = growth hormone deficiency; **hGH** = human growth hormone; **IGF-I** = insulin-like growth factor I; **IQR** = interquartile range; **SDS** = standard deviation score; **TEAE** = treatment emergent adverse event

References

1. SKYTROFA. US Prescribing Information (USPI). Ascendis Pharma; 2024. Accessed May 30, 2024. https://ascendispharma.us/products/pi/skytrofa/skytrofa_pi.pdf; **2.** SKYTROFA. SmPC. EPAR product information. EMA. https://www.ema.europa.eu/en/documents/product-information/skytrofa-previously-lonapegsomatropin ascendis-pharma-epar-product-information_en.pdf. Accessed March 29, 2023; **3.** Thornton PS, Maniatis AK, Aghajanova E, et al. Weekly Lonapegsomatropin in Treatment-Naive Children With Growth Hormone Deficiency: The Phase 3 heiGHt Trial. *J Clin Endocrinol Metab.* 2021;106(11):3184-3195; **4.** Maniatis AK, Nadgir U, Saenger P, et al. Switching to Weekly Lonapegsomatropin from Daily Somatropin in Children with Growth Hormone Deficiency: The fliGHt Trial. *Horm Res Paediatr.* 2022;95(3):233-243; **5.** Maniatis AK, Casella SJ, Nadgir UM, et al. Safety and Efficacy of Lonapegsomatropin in Children With Growth Hormone Deficiency: enliGHten Trial 2-Year Results. *J Clin Endocrinol Metab.* 2022;107(7):e2680-e2689; **6.** Maniatis AK, Thornton PS, Nadgir UM, et al. Children with Growth Hormone Deficiency Treated With Lonapegsomatropin Demonstrated Sustained Height Improvements for up to 6 Years- enliGHten Trial Final Results. *Horm Res Paediatr.* 2025. Online ahead of print; **7.** Grimberg A, Huerta-Saenz L, Grundmeier R, et al. Gender Bias in U.S. Pediatric Growth Hormone Treatment. *Sci Rep.* 2015;9:5:11099. **8.** Kuczmarski RJ, Ogden CL, Guo SS, et al. 2000 CDC Growth Charts for the United States: Methods and Development. *Vital Health Stat 11.* May 2002;(246):1-190.

Disclosures and Funding

This study was sponsored by Ascendis Pharma Endocrinology Division A/S.

MS: employment with Rocky Mountain Pediatric Endocrinology; **AM:** honoraria from, advisory board role for, and speakers' bureau participation for Alexion, Ascendis, BioMarin, Novo Nordisk, and Pfizer; and institutional research funding from Alexion, Ascendis, Novo Nordisk, OPKO, and Pfizer; **MJA:** advisory board role for Ascendis and Pfizer; consulting role for Endo and Rhythm; institutional research funding from Ascendis, Lumos, Mannkind, Medtronic, Novo Nordisk, Rhythm, and Soleno; and other relationship as DSMB for LG; **SC:** advisory board role for Ascendis; **TL** and **DF:** nothing to disclose; **GP:** honoraria, research funding, travel support, advisory board role, consulting role, and speakers' bureau participation for Ascendis, AstraZeneca, Dexcom, Eli Lilly, Johnson and Johnson, Mannkind, Medtronic, Novo Nordisk, Pfizer, and Ultragenyx; and grants from Mannkind Corporation; **AR:** honoraria from, advisory board role for, consulting role for, and travel support from Ascendis, Tolmar, ALK Pharma, and Halozyme Pharma; patents, royalties, or other intellectual property from UpToDate; and other relationships with United Stated Anti-doping Agency and World Anti-doping agency; **EH:** former employee of Ascendis Pharma, Inc.; **CZ:** employee of Ascendis Pharma, Inc.; **PFB:** advisory board role for Ascendis, BioMarin, Novo Nordisk; and speakers' bureau participation for Ascendis and Biomarin.

Medical writing support was provided by Sarah Franco, PhD, an employee of Ascendis Pharma, Inc.

Ascendis, Ascendis Pharma, Ascendis Pharma logo, the company logo and TransCon are trademarks owned by the Ascendis Pharma group.
© April 2025 Ascendis Pharma A/S

MED-US-TC-PGHD-2500030
November 2025