All product candidates are investigational. For scientific exchange and education purposes only. Not for use in product promotion or commercialization.

Design of the New InsiGHTS Trial: A Multicenter, Phase 2, Randomized, Open-Label, Active Controlled Study to Investigate the Safety, Tolerability, and Efficacy of Lonapegsomatropin-tcgd in Prepubertal Individuals With Turner Syndrome

Karen O. Klein,[1] Philippe F. Backeljauw,[2] Sohair Abdelrahman,[3] Pamela M. Lai,[3] Meng Mao,[3] Aimee D. Shu[3]

[1]Rady Children's Hospital and University of California — San Diego, San Diego, CA; [2]Cincinnati Children's Hospital Medical Center, Cincinnati, OH; [3]Ascendis Pharma, Inc, Palo Alto, CA

THU-164

BACKGROUND

- Turner Syndrome (TS) is a rare condition that occurs 1 in 2500 live female births and is characterized by the complete or partial absence of one X chromosome[1]
 - Common clinical features include skeletal disproportion (growth failure, skeletal dysplasia, and low bone mass) and premature ovarian insufficiency in addition to a range of comorbidities including increased risk of thyroid disease, heart abnormalities, and diabetes mellitus[1,2]
- At this time, individuals with TS are treated with daily somatropin (human growth hormone [hGH]) which is effective at promoting height gain and may improve body composition[3]
 - However, daily somatropin therapy has been associated with poor adherence in growth disorders such as growth hormone deficiency (GHD), small gestational age (SGA), idiopathic short stature (ISS), Prader Willi Syndrome and TS which may result in reduced therapeutic efficacy[4,5]
- Lonapegsomatropin-tcgd (SKYTROFA™; TransCon™ hGH), a prodrug of somatropin, is approved for once-weekly treatment of pediatric GHD by the FDA (pediatric patients 1 year or older and weighing at least 11.5 kg), and by the EMA (pediatric patients age 3 years to 18 years)[6-8,11]
- The phase 2 New InsiGHTS trial will investigate the safety, tolerability, and efficacy of once-weekly lonapegsomatropin-tcgd in individuals with TS (NCT05690386)

Figure 1. Lonapegsomatropin-tcgd (TransCon® hGH) Design[6,10]

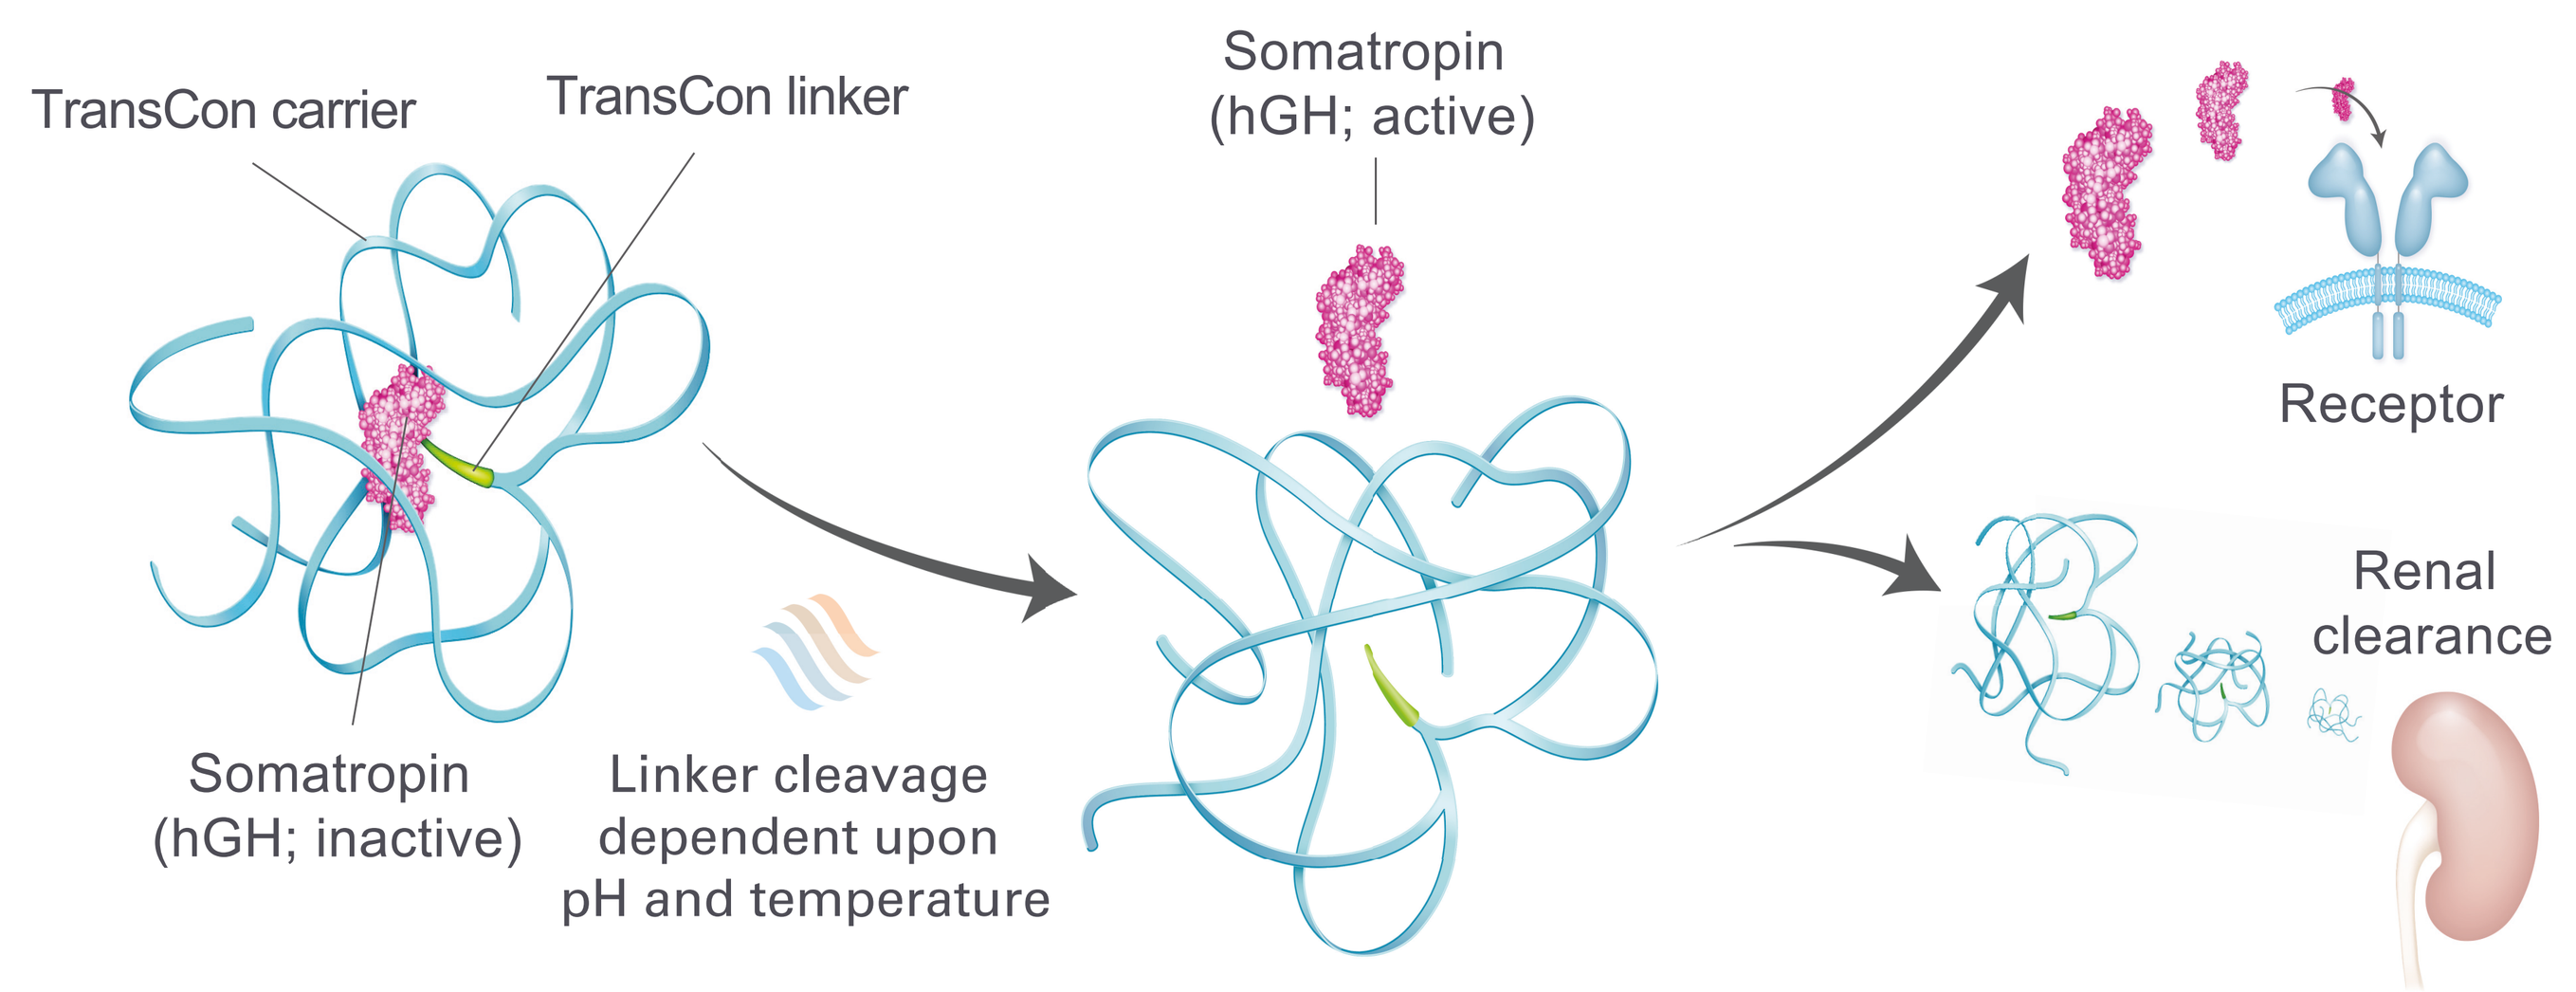
TransCon carrier
TransCon linker
Somatropin (hGH; inactive)
Linker cleavage dependent upon pH and temperature
Somatropin (hGH; active)
Receptor
Renal clearance

- Lonapegsomatropin-tcgd is a prodrug of somatropin, administered once-weekly and designed to provide sustained release of active, unmodified somatropin
- The unmodified, unbound somatropin released from lonapegsomatropin-tcgd has the identical 191 amino-acid sequence and size (22 kDa) as endogenous growth hormone (GH)

METHODS

TRIAL DESIGN AND STUDY POPULATION

Design:

- The New InsiGHTS trial is a phase 2, multicenter, open-label, active controlled, parallel arm, dose-finding study randomizing 48 individuals, 1:1:1:1, to lonapegsomatropin-tcgd (0.24, 0.30, 0.36 mg hGH/kg/week) or daily somatropin (0.05 mg/kg/day [0.35 mg/kg/week], a dose consistent with treatment guidelines) and stratifying them by chronological age (< 4 and ≥ 4 years) (NCT05690386)

Objectives:

- The primary objective is to evaluate and compare three different doses of once-weekly lonapegsomatropin-tcgd to daily somatropin in promoting linear growth at 26 weeks

Subject population:

- Prepubertal individuals with Turner Syndrome (TS) (confirmed by genetic testing), exhibiting impaired growth that are growth hormone (GH) and GH secretagogue-naïve, from the US

Table 1. Inclusion and Exclusion Criteria

Key Inclusion Criteria	Key Exclusion Criteria
• Diagnosed with TS confirmed by genetic testing • Ages 1 year to 10 years (inclusive) • Tanner Stage 1 • GH/GH secretagogue-naive • Biochemically euthyroid (including when on thyroid hormone supplementation) • Exhibit impaired growth defined by at least one of the following: – If ≥ 2 years: AHV < 6 cm/year or < 25th percentile over a time span of 6 to 18 months – If < 2 years: Height (or length) < 10th percentile for sex and age • Bone age within normal limits (no more than 20% above or below chronological age in months)	• Any known or history of clinically significant condition(s) likely to affect growth • Diagnosis of diabetes mellitus • Presence of Y-chromosomal material provided there is no history of gonadectomy • Receiving prior or concurrent treatment with any agent that might influence growth or interfere with GH secretion or action such as, but not limited to, nonsteroidal anabolic agents, sex steroids, etc

Figure 2. Inclusion and Exclusion Criteria

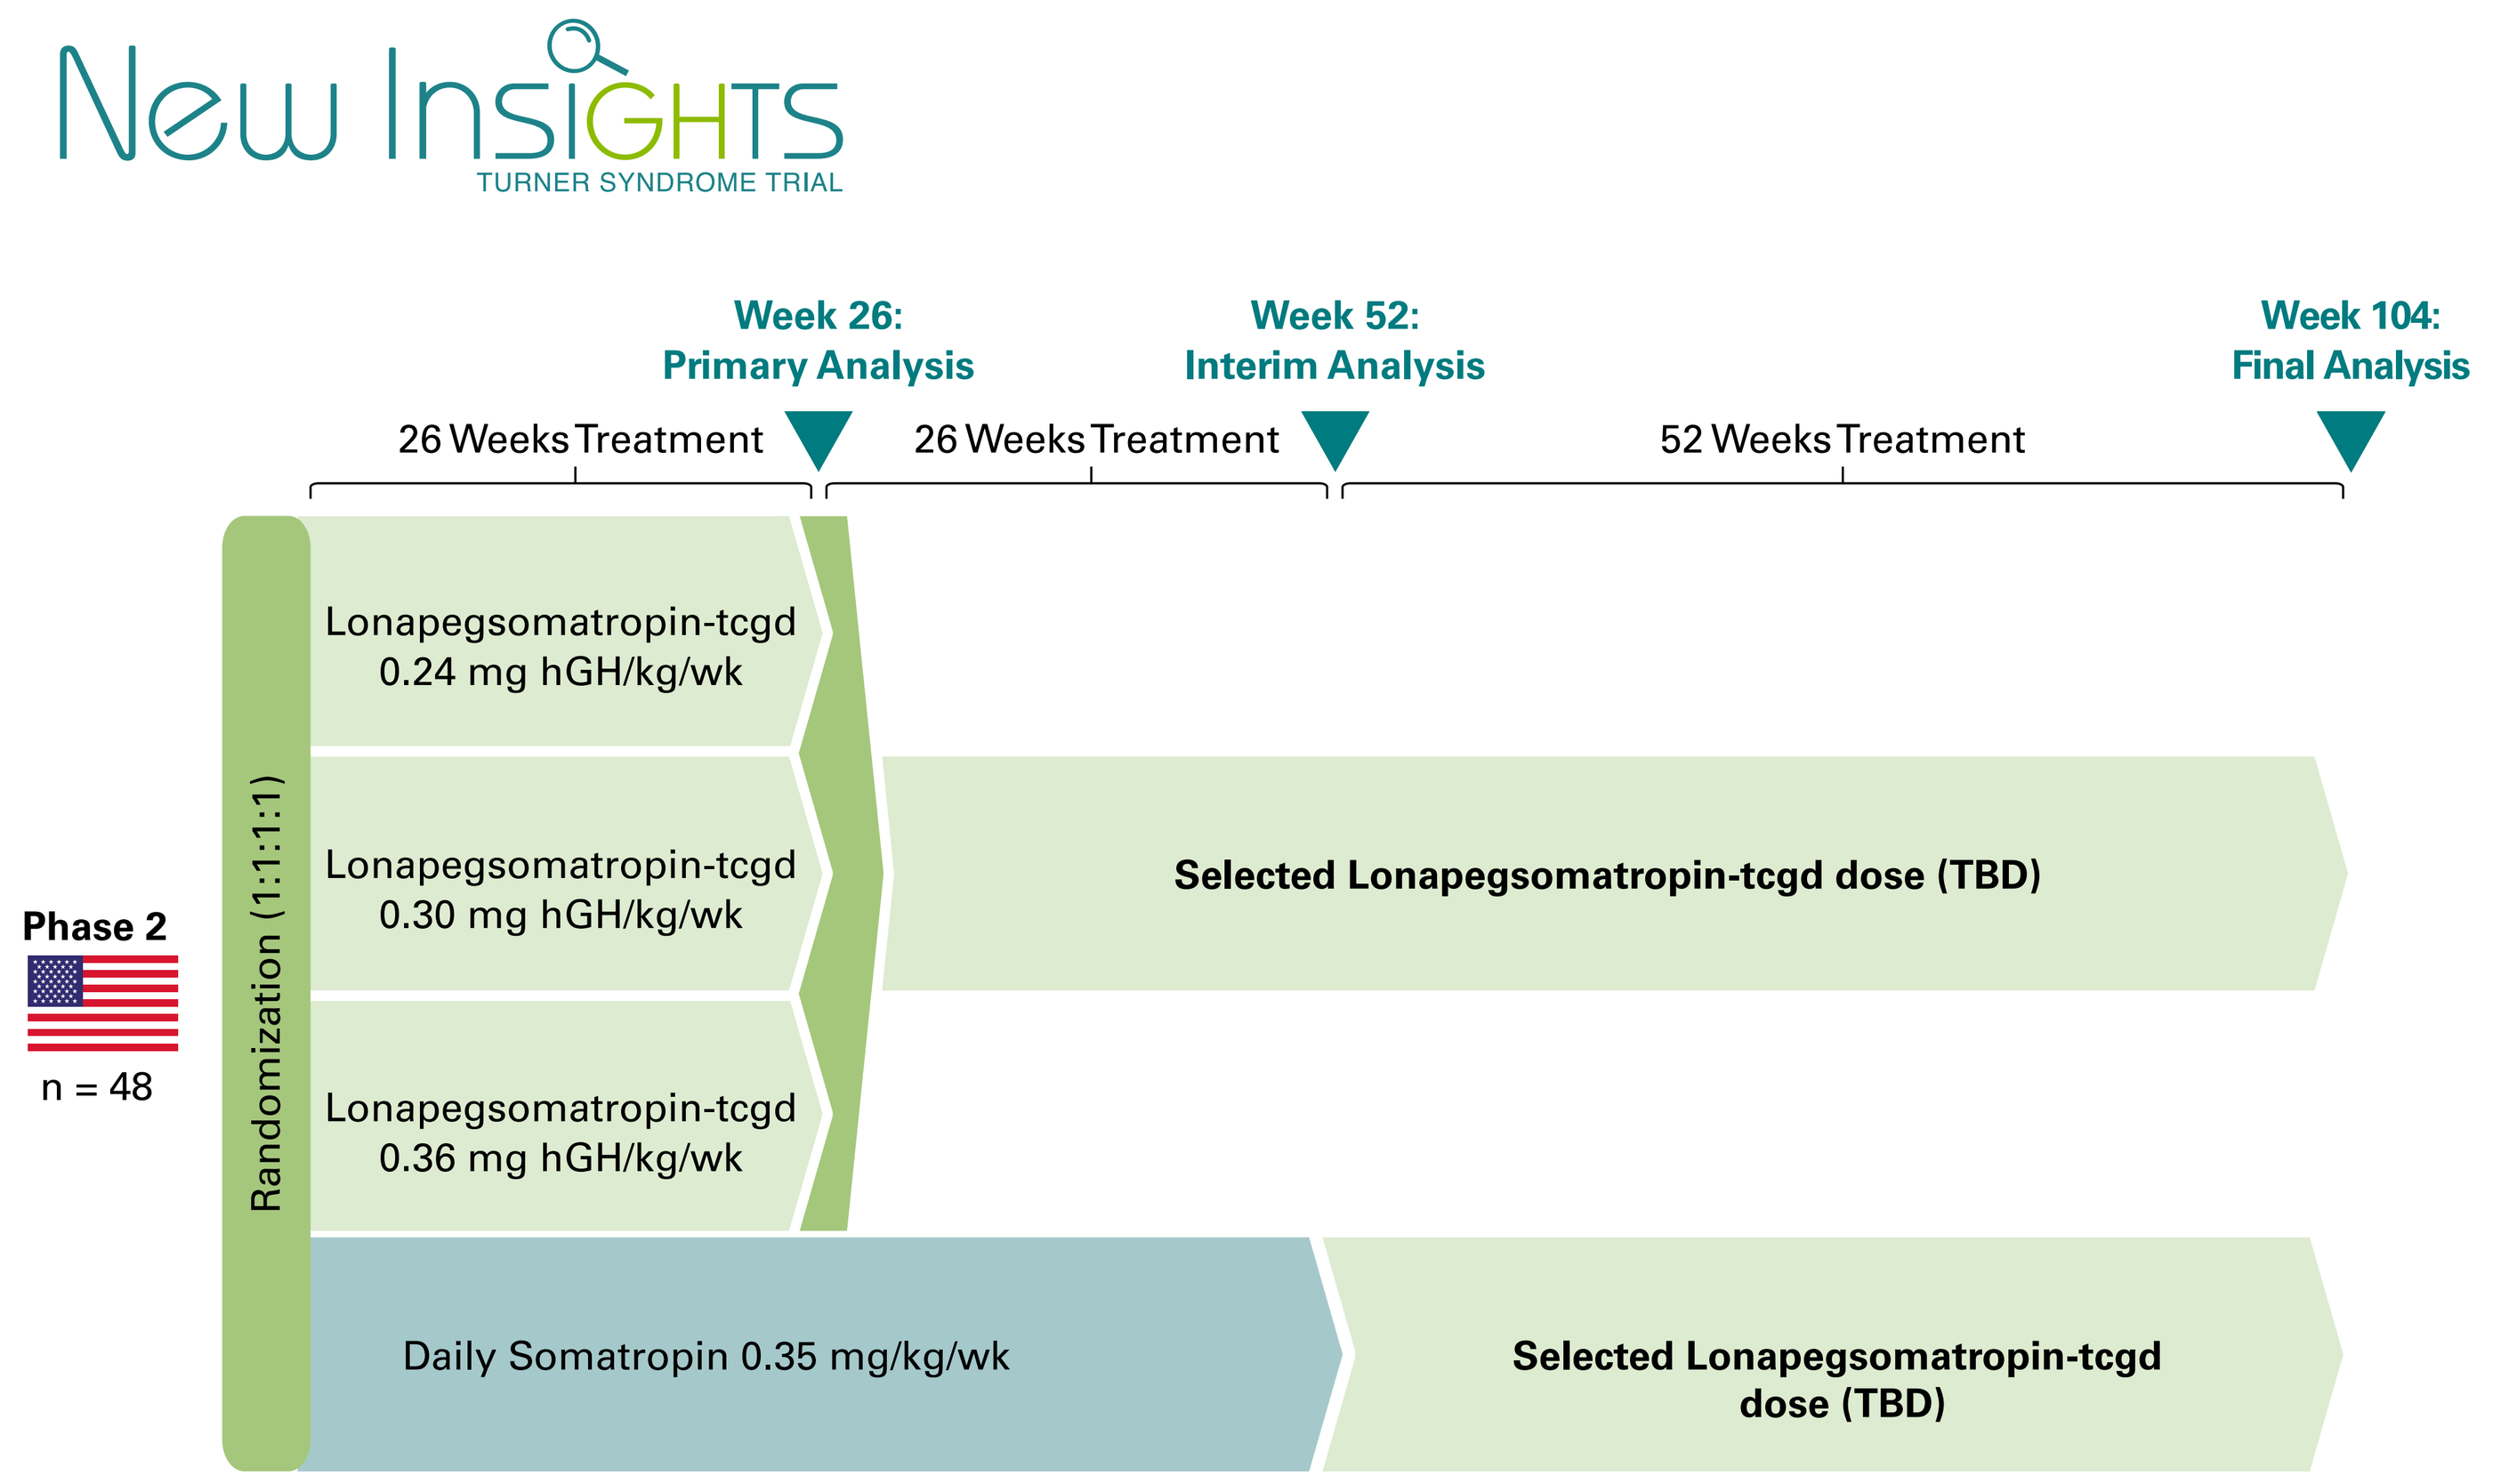
New InsiGHTS
TURNER SYNDROME TRIAL
Week 26: Primary Analysis
Week 52: Interim Analysis
Week 104: Final Analysis
26 Weeks Treatment
26 Weeks Treatment
52 Weeks Treatment
Phase 2
n = 48
Randomization (1:1:1:1)
Lonapegsomatropin-tcgd 0.24 mg hGH/kg/wk
Lonapegsomatropin-tcgd 0.30 mg hGH/kg/wk
Lonapegsomatropin-tcgd 0.36 mg hGH/kg/wk
Selected Lonapegsomatropin-tcgd dose (TBD)
Daily Somatropin 0.35 mg/kg/wk
Selected Lonapegsomatropin-tcgd dose (TBD)

Table 2. Outcomes

Primary	• Annualized Height Velocity (AHV) (cm/year) at 26 weeks
Secondary	• AHV (cm/year) at 52 and 104 weeks • Change from baseline in height standard deviation score (SDS) at 26, 52 and 104 weeks • Change from baseline in bone age at 52 and 104 weeks • Change from baseline in ratio of bone age/chronological age at 104 weeks
Other	• Insulin-like growth factor 1 (IGF-1) SDS at 26, 52 and 104 weeks

Figure 3. New InsiGHTS Trial Site Locations

- States with registered study sites for the New InsiGHTS trial across the US

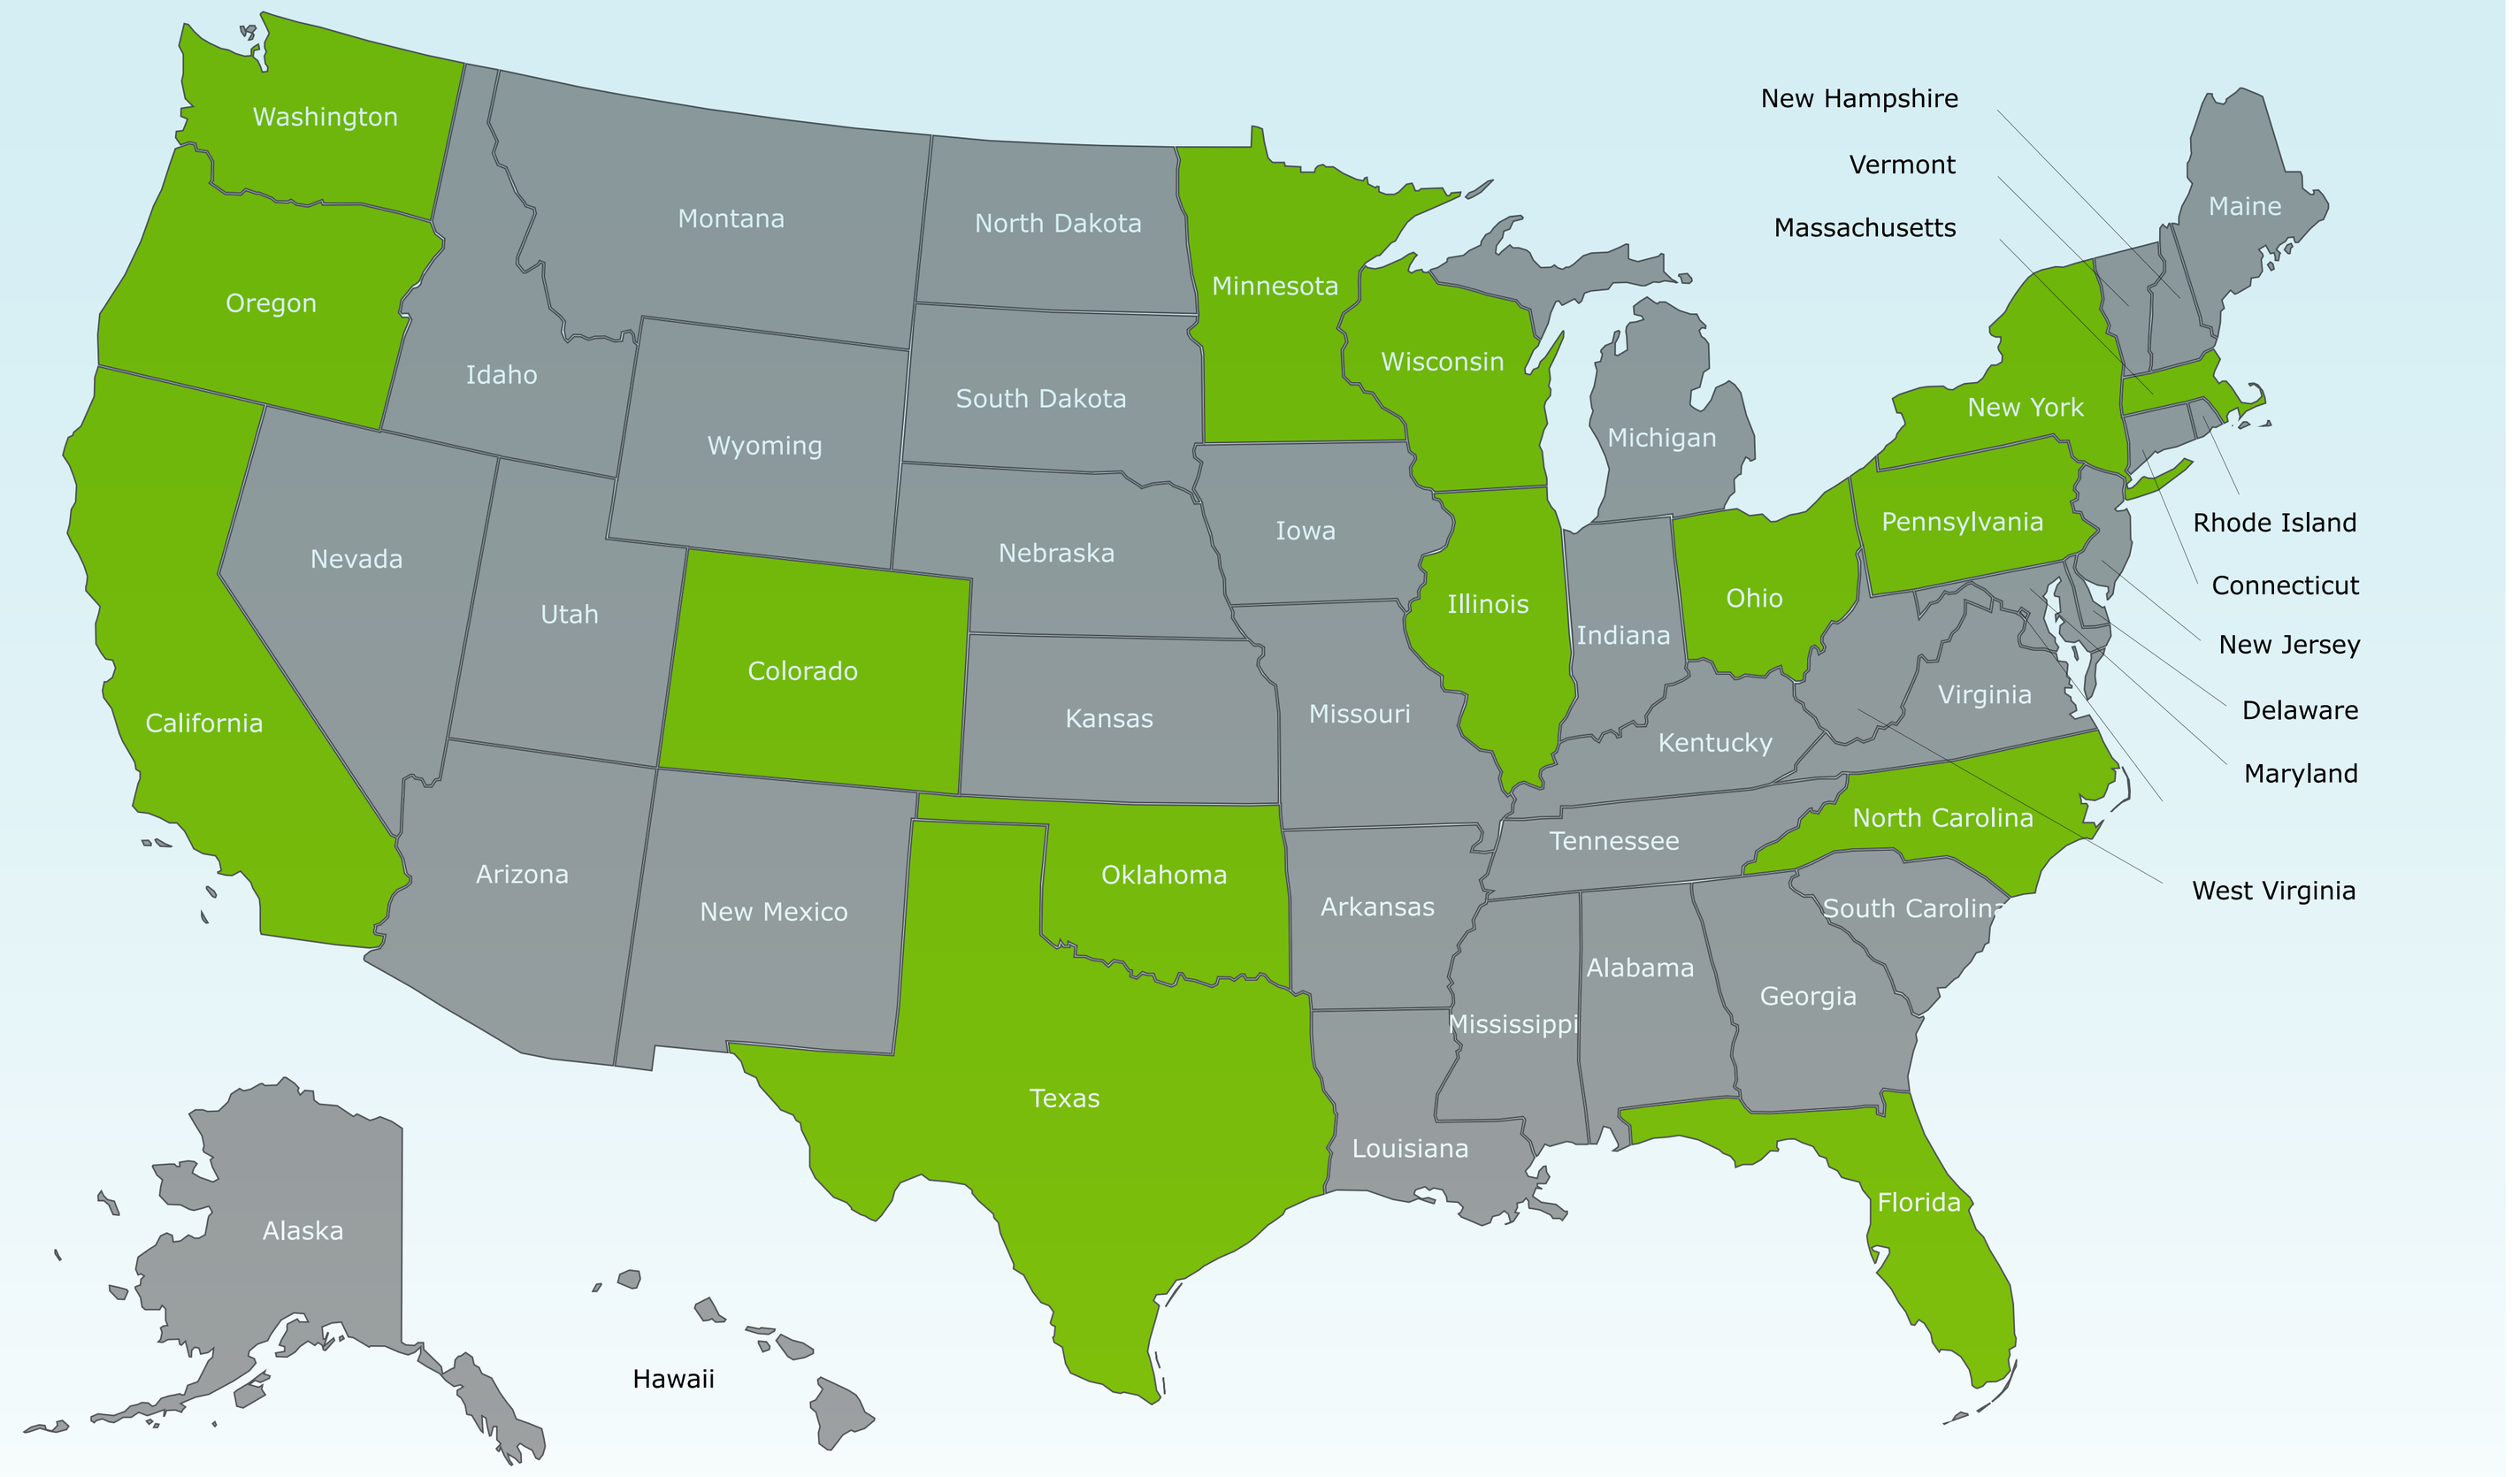
Washington
Oregon
California
Idaho
Nevada
Montana
Wyoming
Utah
Arizona
Colorado
New Mexico
North Dakota
South Dakota
Nebraska
Kansas
Oklahoma
Texas
Minnesota
Iowa
Missouri
Arkansas
Louisiana
Wisconsin
Illinois
Michigan
Indiana
Ohio
Kentucky
Tennessee
Mississippi
Alabama
Georgia
Florida
South Carolina
North Carolina
Virginia
Pennsylvania
New York
Maine
New Hampshire
Vermont
Massachusetts
Rhode Island
Connecticut
New Jersey
Delaware
Maryland
West Virginia
Alaska
Hawaii

CONCLUSIONS

- The phase 2 New InsiGHTS trial is the first time that once-weekly lonapegsomatropin-tcgd will be studied in individuals with TS
- The New InsiGHTS will provide an opportunity to examine the impact of lonapegsomatropin-tcgd in a population whose short stature is not due to GH deficiency

ABBREVIATIONS

AHV = Annualized Height Velocity; **EMA** = European Medicines Agency; **GH** = growth hormone; **GHD** = growth hormone deficiency; **hGH** = human growth hormone; **IGF-1** = insulin-like growth factor 1; **ISS** = idiopathic short stature; **SDS** = standard deviation score; **SGA** = small gestational age; **TS** = Turner Syndrome

REFERENCES:

1. Bondy CA. Turner syndrome 2008. *Horm Res.* 2009;71 Suppl 1:52-56. **2.** Gravholt CH, Andersen NH, Conway GS, et al. Clinical practice guidelines for the care of girls and women with Turner syndrome: proceedings from the 2016 Cincinnati International Turner Syndrome Meeting. *Eur J Endocrinol.* 2017;177(3):G1-G7. **3.** Irzyniec, T, Jeż, W., Lepska, K. et al. Childhood growth hormone treatment in women with Turner syndrome - benefits and adverse effects. *Sci Rep.* 9, 15951 (2019). **4.** Hartmann K, Ittner J, Müller-Rossberg E, et al. Growth hormone treatment adherence in prepubertal and pubertal children with different growth disorders. *Horm Res Paediatr.* 2013;80(1):1-5. **5.** Cutfield W., Derraik J., Gunn A., et al. Non-Compliance with Growth Hormone Treatment in Children Is Common ands Impairs Linear Growth. *PLoS ONE.* 2011;6(1):e16223. **6.** Thornton PS, Maniatis AK, Aghajanova E, et al. Weekly Lonapegsomatropin in Treatment-Naïve Children With Growth Hormone Deficiency: The Phase 3 heiGHt Trial. *J Clin Endocrinol Metab.* 2021;106(11):3184-3195. **7.** SKYTROFA. US Prescribing Information (USPI). Ascendis Pharma; 2022. https://ascendispharma.us/products/pi/skytrofa/skytrofa_pi.pdf. Accessed March 29, 2023. **8.** Maniatis AK, Nadgir U, Saenger P, et al. Switching to Weekly Lonapegsomatropin from Daily Somatropin in Children with Growth Hormone Deficiency: The fliGHt Trial. *Horm Res Paediatr.* 2022;95(3):233-243. **9.** Maniatis AK, Casella SJ, Nadgir UM, et al. Safety and Efficacy of Lonapegsomatropin in Children With Growth Hormone Deficiency: enliGHten Trial 2-Year Results. *J Clin Endocrinol Metab.* 2022;107(7):e2680-e2689. **10.** Sprogøe K, Mortensen E, Karpf DB, Leff JA. The rationale and design of TransCon Growth Hormone for the treatment of growth hormone deficiency. *Endocr Connect.* 2017;6(8):R171-R181. **11.** SKYTROFA. SmPC. EPAR product information. EMA. https://www.ema.europa.eu/en/medicines/human/EPAR/skytrofa-previously-lonapegsomatropin-ascendis-pharma. Accessed March 29, 2023.

DISCLOSURES AND FUNDING

This study was sponsored by Ascendis Pharma Endocrinology Division A/S.

Individual author disclosures: KOK: Advisory Board Member; Abbvie, Tolmar. Consulting Fee; Self; Abbvie, Tolmar. Grant Recipient; Self; Azurity. Research Investigator; Self; Abbvie, Azuriity. Speaker; Self; Abbvie. **P.B:** Advisory Board Member; Self; Ascendis Pharma. Consulting Fee; Self; Novo Nordisk, Novartis Pharmaceuticals, Ascendis Pharma, BioMarin Pharmaceutical, Tolmar, Cavalry Biosciences, Ipsen. Research Investigator; Self; Novo Nordisk, Novartis Pharmaceuticals, Ipsen . Speaker; Self; Ascendis Pharma. **SA:** Employee; Self; Ascendis Pharma. P.L: Employee; Self; Ascendis Pharma. **MM:** Employee; Self; Ascendis Pharma. **ADS:** Employee; Self; Ascendis Pharma.

Study design and development support by **Ari Bitton**, an employee of Ascendis Pharma, Inc. Medical writing support by **Manasa Akkineni**, an employee/consultant to Ascendis Pharma, Inc.

All product candidates are investigational. Intended for education and scientific exchange only. Not for use in promotion or product commercialization.

Ascendis, Ascendis Pharma, Ascendis Pharma logo, the company logo and TransCon are trademarks owned by the Ascendis Pharma group.
© June 2023 Ascendis Pharma A/S

MED-US-TC-PGHD-2500031
November 2025