Safety and Effectiveness of Lonapegsomatropin in the SkybriGHt Registry: Analysis of the First 100 Patients Enrolled With 2 Years of Follow-Up

SUN-201

Aristides K Maniatis,[1] Joshua H Yang,[2] M Jennifer Abuzzahab,[2] Andrew Dauber,[4] Bradley S Miller,[5] Terri H Lipman,[6] Steven Chernausek,[7] Gnanagurudasan Prakasam,[8] Daniel Flynn,[9] Allison S Komirenko,[10] Pamela M Lai,[10] Laurie Domrzalski,[10] Carol Zhao,[10] Jennifer M Iklé,[10] Aimee D Shu,[10] Philippe Backeljauw,[11] Alan D Rogol[12]

[1]Rocky Mountain Pediatric Endocrinology, PC, Centennial, CO, USA; [2]Orlando Health Arnold Palmer Hospital for Children Endocrinology and Diabetes Center, Orlando, FL, USA; [3]Children's Minnesota, Minneapolis, MN, USA; [4]Children's National Hospital, Washington, DC, USA; [5]University of Minnesota Medical School, Minneapolis, MN, USA; [6]University of Pennsylvania School of Nursing, Philadelphia, PA, USA; [7]University of Oklahoma Health Sciences Center, Oklahoma City, OK, USA; [8]Sutter Medical Center, Sacramento, CA, USA; [9]St Luke's Boise Medical Center, Boise, ID, USA; [10]Ascendis Pharma, LLC, Palo Alto, CA, USA; [11]Cincinnati Children's Hospital Medical Center, Cincinnati, OH, USA; [12]University of Virginia, Charlottesville, VA, USA

Conclusions

This prospective, real-world study evaluated lonapegsomatropin in the first 100 children and adolescents with GH deficiency for 2 years following enrollment in the SkybriGHt registry.

Lonapegsomatropin dosing remained stable, with continued increases in height SDS:

- Height SDS increased from enrollment to month 24.
- AHV was consistent with clinical expectations for age and treatment duration.
- As expected, AHV was higher among participants without prior GH therapy than among those previously treated with GH therapy.

Mean values of IGF-I SDS were maintained within a typical treatment target range (0 to +2) over time, consistent with adequate GH replacement.

Over 24 months, mean BMI SDS remained stable near 0.

There were no serious or severe treatment-related AEs, no AEs that led to discontinuation, and no deaths.

Safety and effectiveness were consistent with clinical trial findings and similar to outcomes reported with daily somatropin, supporting once-weekly lonapegsomatropin use in routine clinical practice.

Background

- Lonapegsomatropin (TransCon® hGH), a prodrug that provides sustained release of unmodified somatropin, is approved in the US as a once-weekly treatment for children and adults with GH deficiency and in Europe and other regions for pediatric GH deficiency.[1-3]
- Phase 3 heiGHt and fliGHt trials established robust efficacy and a safety profile of lonapegsomatropin comparable to that of daily somatropin therapy; data from enliGHten demonstrated sustained efficacy and established longer term safety profile.[4-7]
- Real-world data complement clinical trial findings by providing insights into disease management, treatment utilization, and long-term outcomes in routine clinical practice.[8]
- SkybriGHt is a US-based, multicenter, prospective, long-term study evaluating the safety and effectiveness of lonapegsomatropin in children and adolescents in the post-marketing setting.
- We report outcomes of lonapegsomatropin treatment for the first 100 children, 2 years post-enrollment in the registry.

Methods

SkybriGHt Registry Study

- Enrollment in SkybriGHt is ongoing across the US and will occur over a 5-year period.
- Children prescribed lonapegsomatropin in routine clinical practice are eligible.
- The decision to treat with lonapegsomatropin is made prior to and independent of study participation.
- The study is planned to continue for a total duration of 10 years.
- Lonapegsomatropin is administered subcutaneously using the GH Auto-Injector, which automatically reconstitutes the preservative-free lyophilized drug from prefilled cartridges.

Methods (cont'd)

SkybriGHt Registry Study (cont'd)

- Clinical outcomes evaluated include height SDS, AHV, and IGF-I SDS.
- Safety is assessed through AE reporting

Statistical Analyses

- Data cut-off: November 14, 2025
- Analyses were based on the first 100 children at 24 months of follow-up after enrollment.
- Descriptive statistics were used:
 - Subgroup analyses for children initiating GH therapy with lonapegsomatropin and children switching from prior GH therapy

Figure 1. **SkybriGHt Registry Design (NCT05820672)**

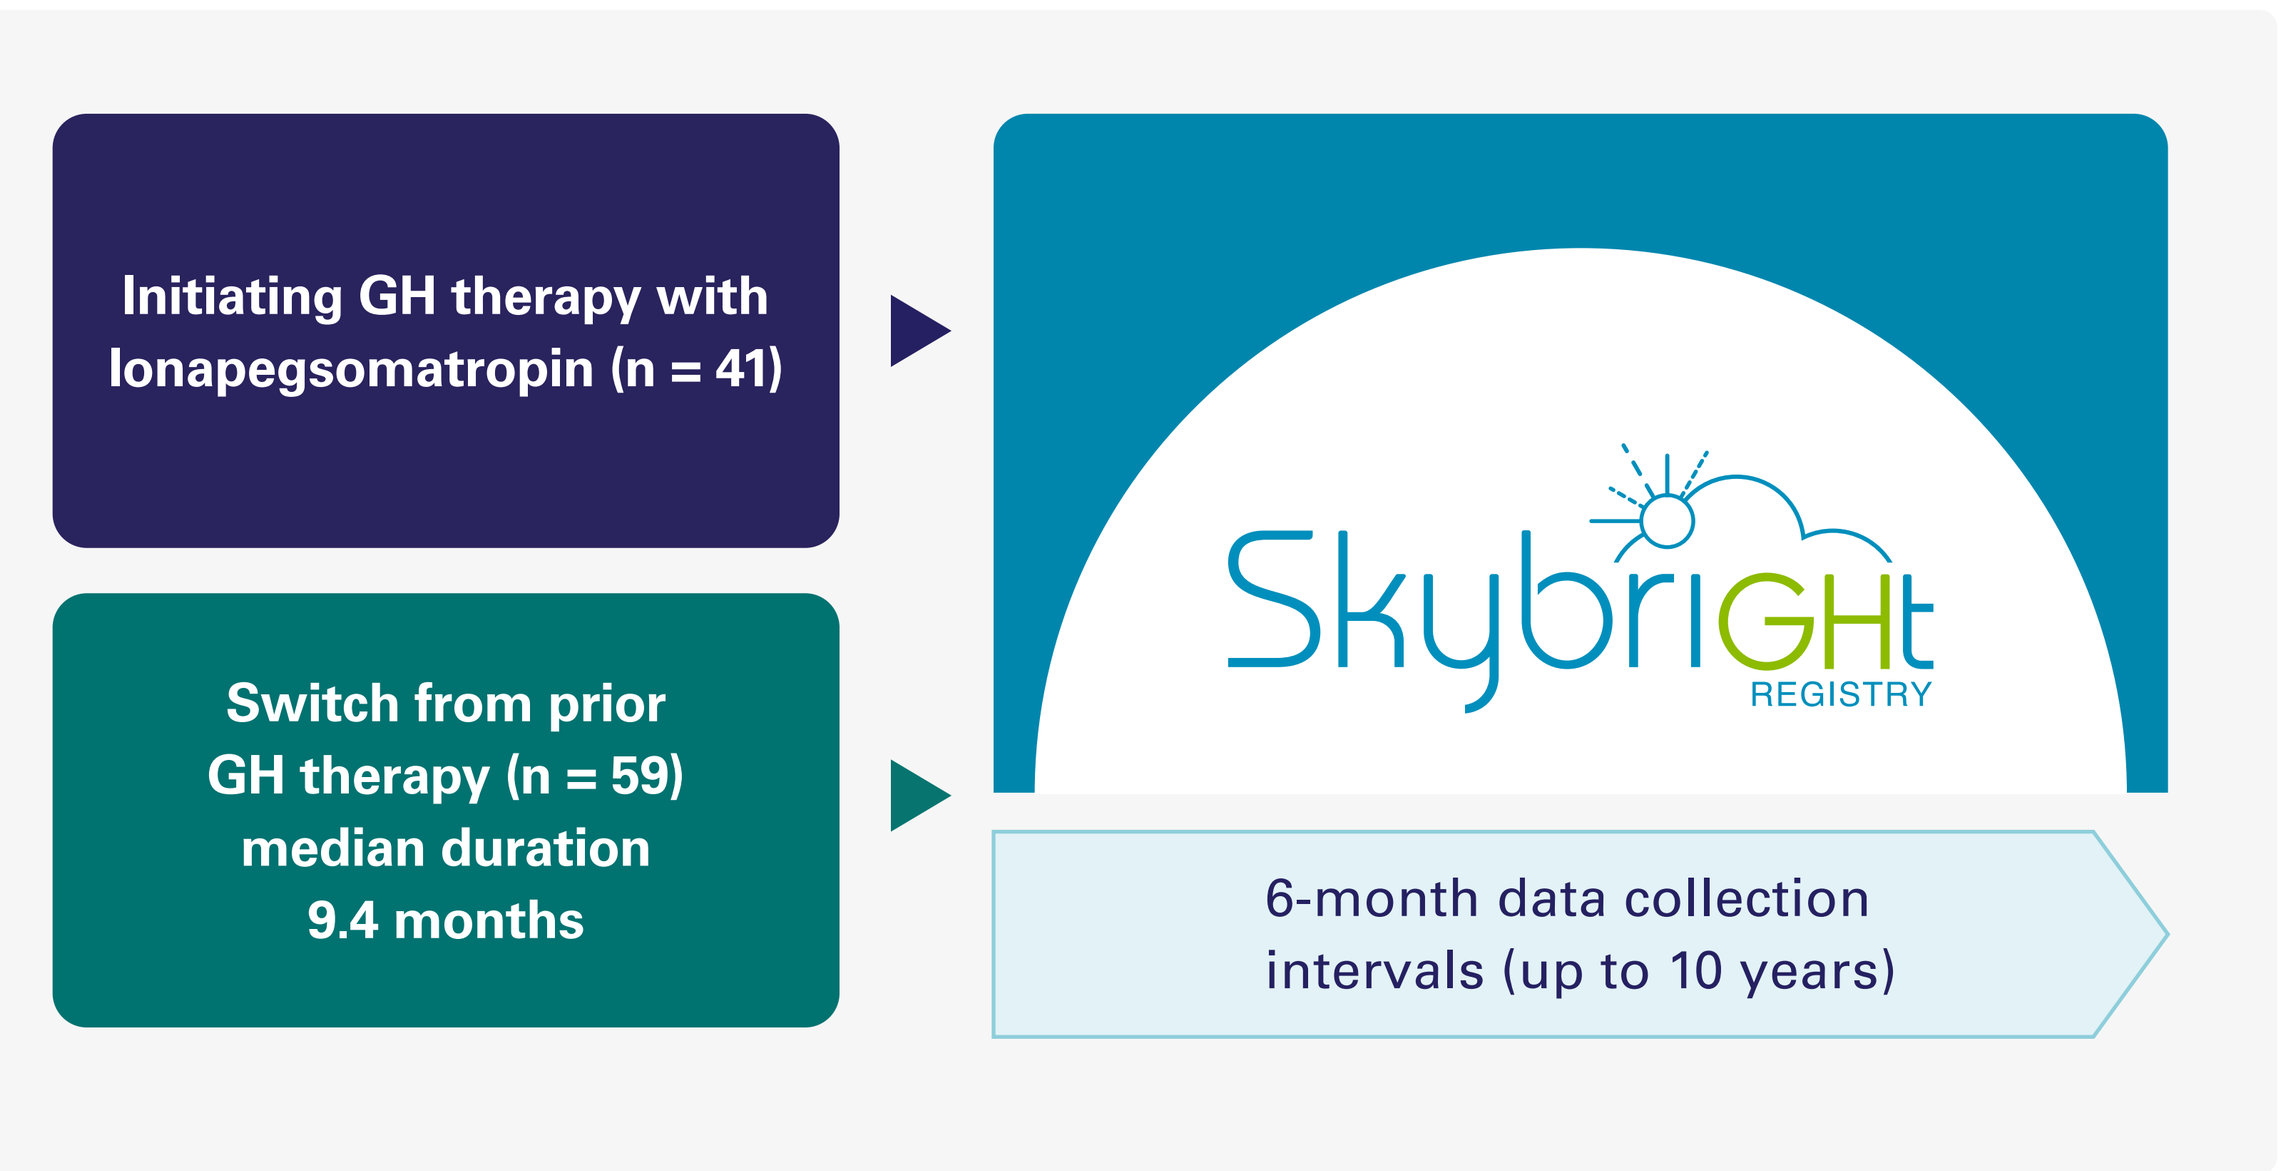
Initiating GH therapy with lonapegsomatropin (n = 41)
Switch from prior GH therapy (n = 59) median duration 9.4 months
SkybriGHt REGISTRY
6-month data collection intervals (up to 10 years)

Results

Figure 2. **Treatment Exposure**

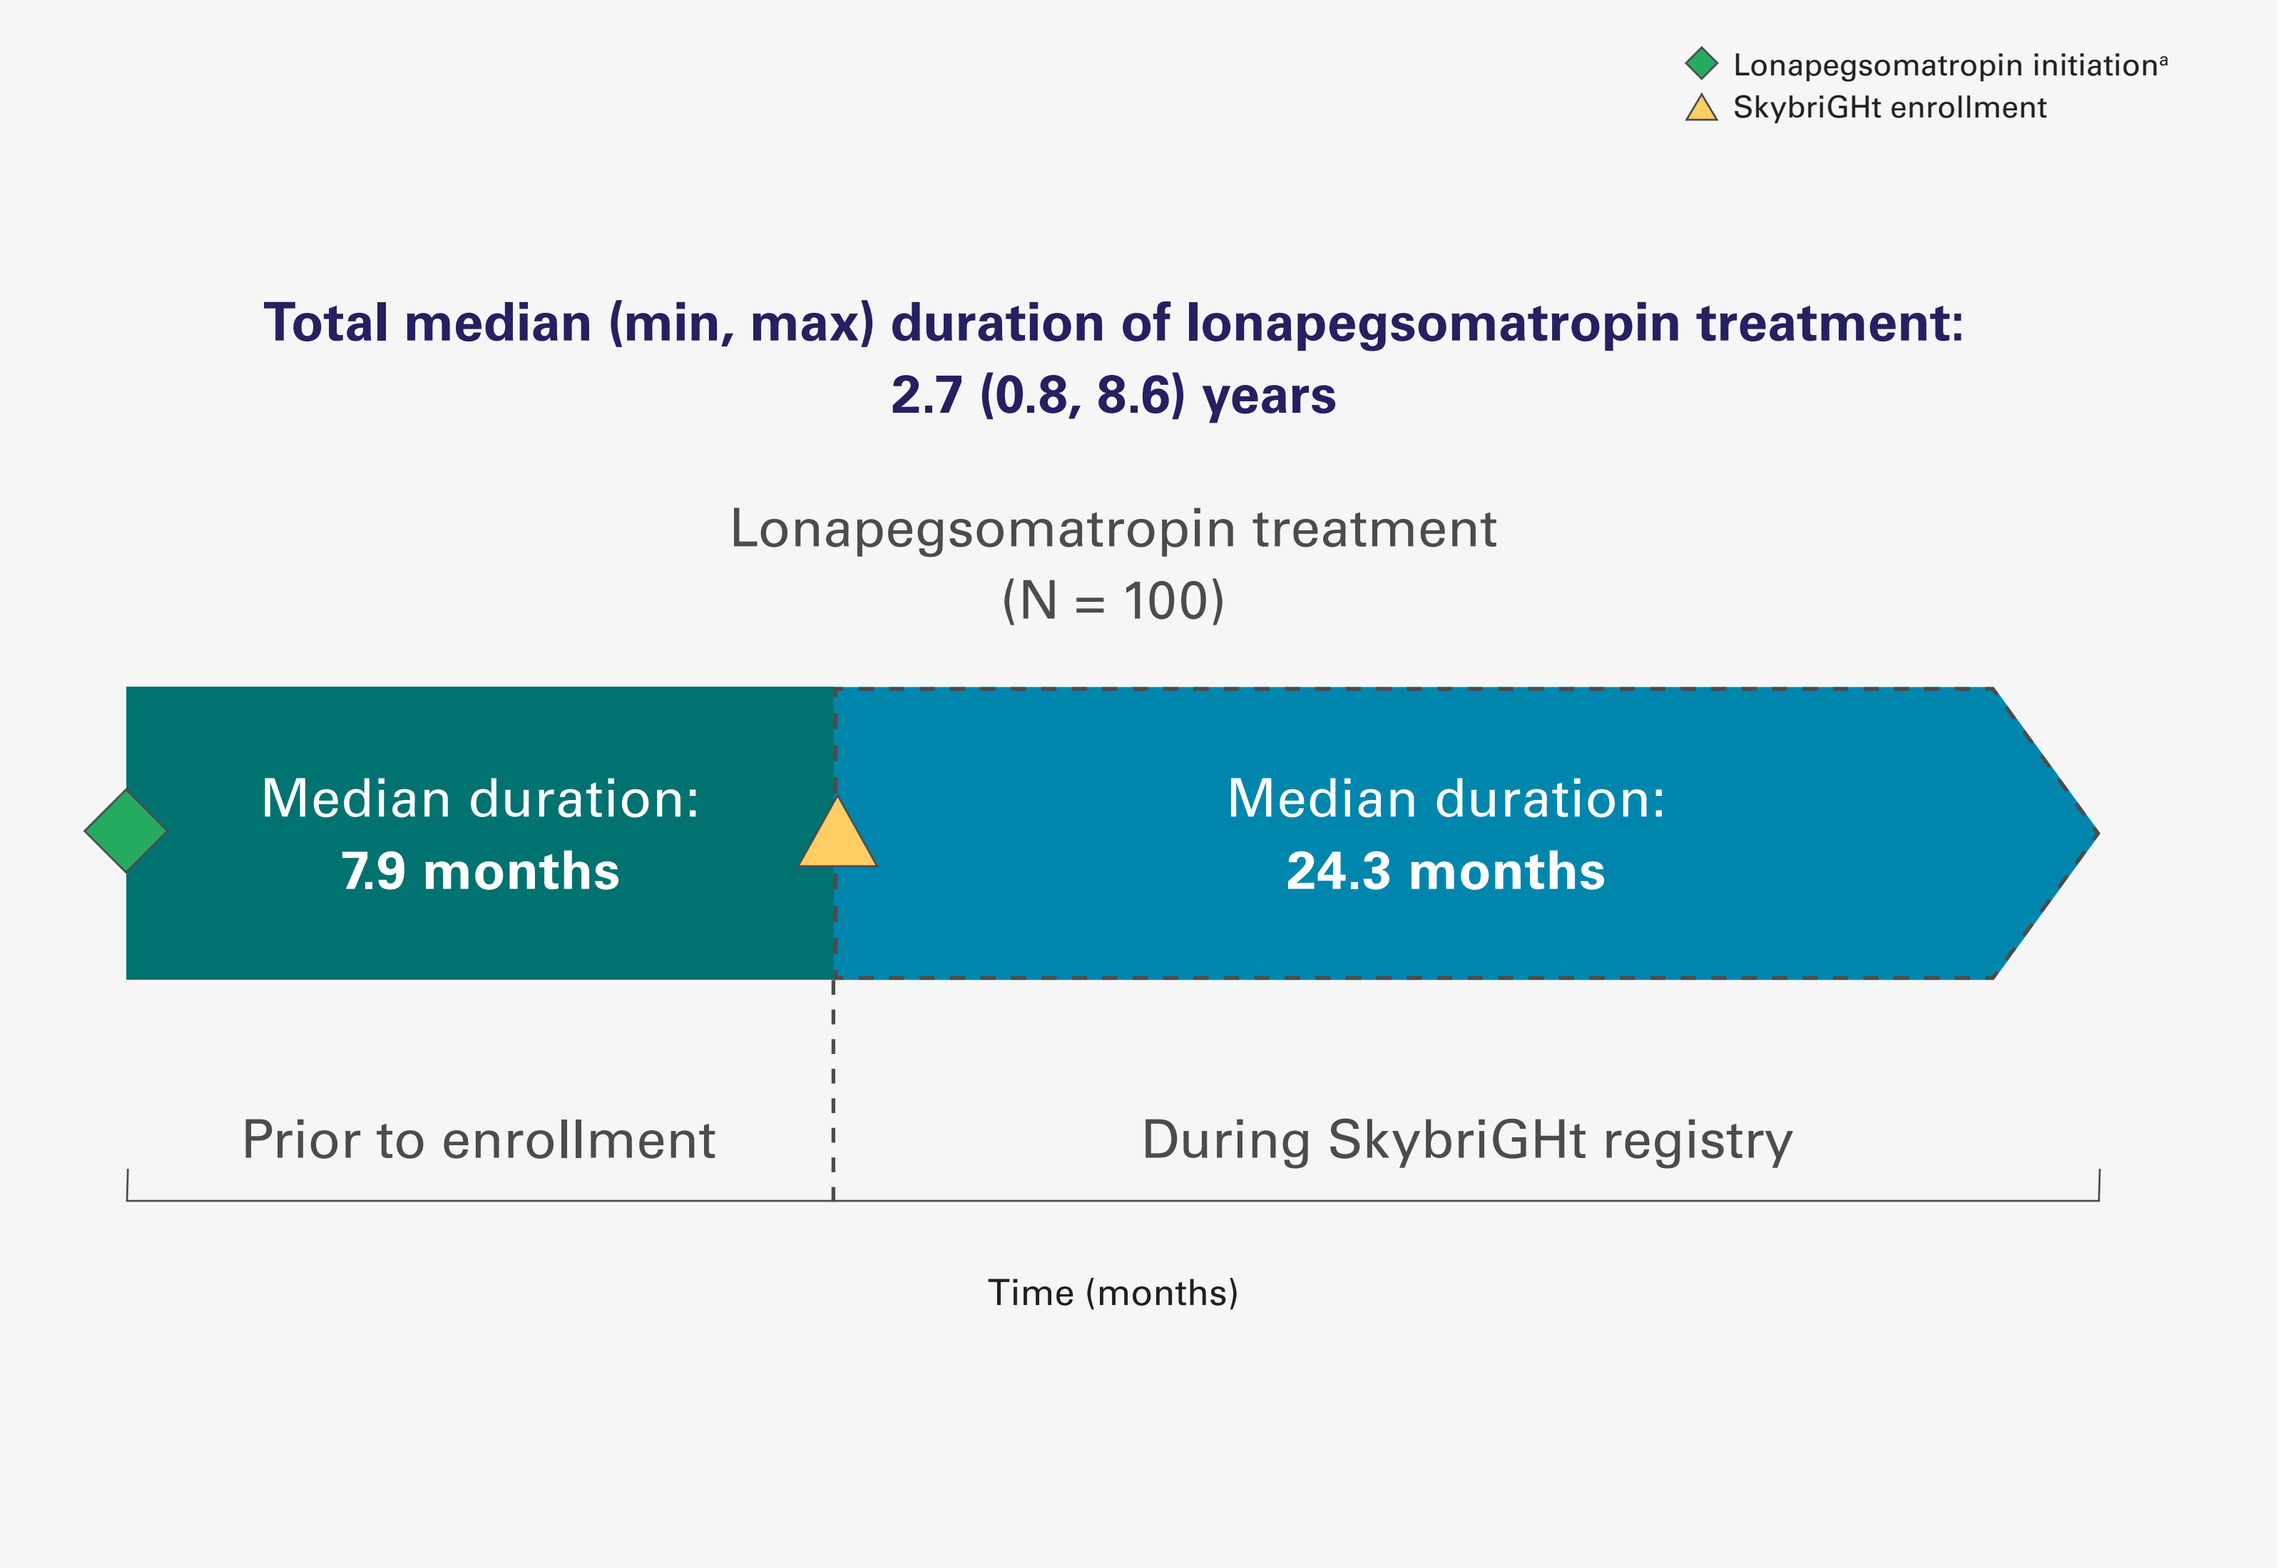
Lonapegsomatropin initiation[a]
SkybriGHt enrollment
Total median (min, max) duration of lonapegsomatropin treatment: 2.7 (0.8, 8.6) years
Lonapegsomatropin treatment (N = 100)
Median duration: 7.9 months
Median duration: 24.3 months
Prior to enrollment
During SkybriGHt registry
Time (months)

[a]Lonapegsomatropin treatment may have been initiated at varying times and for varying duration prior to enrollment. Patients may have participated in lonapegsomatropin clinical trials prior to SkybriGHt. Initiation of lonapegsomatropin may have been preceded by other GH therapies such as daily somatropin or another long-acting GH.

Results

Table 1. **Demographics and Baseline Characteristics**

	Variable		Total (N = 100)
	Sex, n (%)	Male	82 (82)
		Female	18 (18)
	Race, n (%)	White	86 (86)
		Asian	6 (6)
		Black or African American	3 (3)
		Other	1 (1)
		Not reported	4 (4)
At diagnosis	Indication for GH treatment, n (%)	GH deficiency	100 (100)
	Age, mean (SD)[a]	Years	8.0 (3.4), n = 99
		Min, max	1.3, 15.4
At first dose of lonapegsomatropin	Age, mean (SD)[a]	Years	9.7 (3.0)
		Min, max	2.7, 15.6
	Height SDS, mean (SD)[b]		-0.79 (0.99)
	IGF-I SDS, mean (SD)[b]		0.2 (1.5), n = 64
	BMI SDS, mean (SD)[b]		-0.06 (0.95)
At SkybriGHt enrollment	Age, mean (SD)[a]	Years	11.1 (3.0)
		Min, max	3.1, 16.3
	Height SDS, mean (SD)		-0.59 (0.98)
	IGF-I SDS, mean (SD)		1.20 (1.05), n = 67
	BMI SDS, mean (SD)		0.01 (0.91), n = 99
	Bone age/chronological age ratio, mean (SD)		0.9 (0.1), n = 32
	Tanner stage, n (%)[c]	Stage I	50 (50)
		Stage II	7 (7)
		Stage III	11 (11)
		Stage IV	6 (6)
		Stage V	1 (1)

Note: Unless otherwise specified, values are based on the total study population (N = 100). For variables with missing data, the number of participants with available assessments is indicated.
[a]Age was determined by assigning June 15 as the birth date within the reported birth year.
[b]Defined as the last assessment prior to the first dose of lonapegsomatropin.
[c]Tanner stage was reported for n = 75 patients.

Figure 3. **Lonapegsomatropin Dosing**[2]

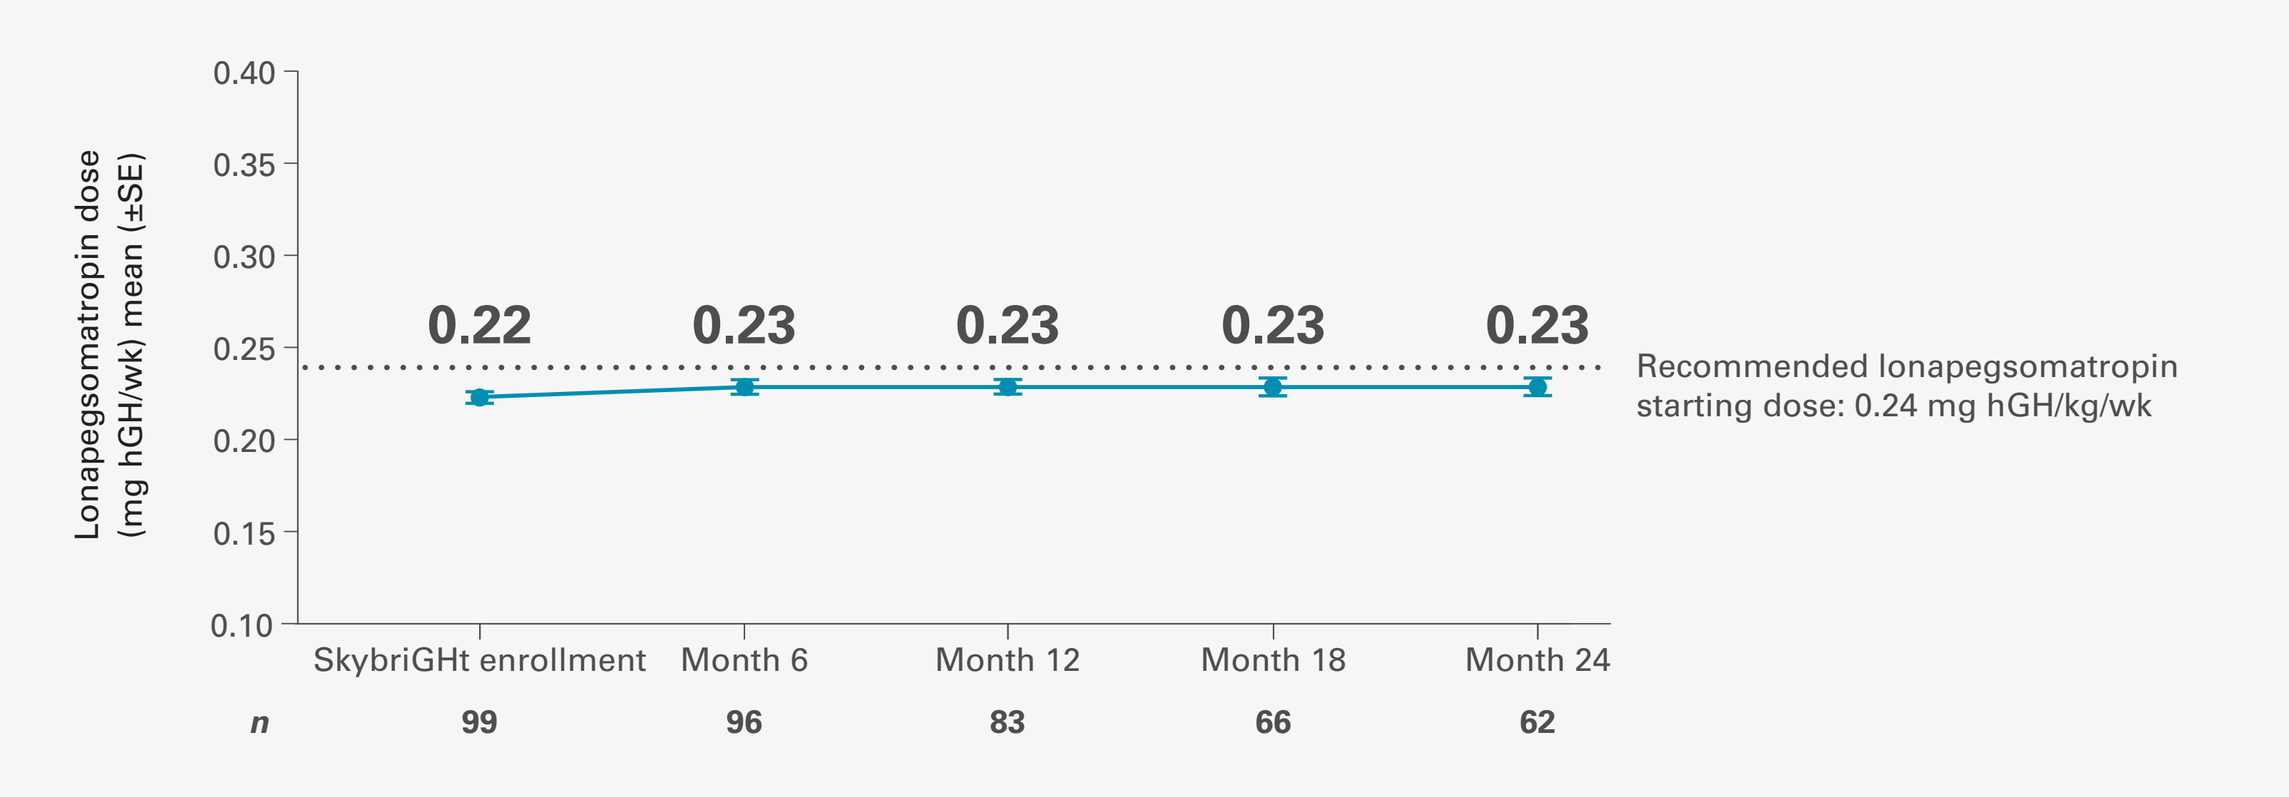
Lonapegsomatropin dose (mg hGH/wk) mean (±SE)
0.22
0.23
0.23
0.23
0.23
Recommended lonapegsomatropin starting dose: 0.24 mg hGH/kg/wk
SkybriGHt enrollment
Month 6
Month 12
Month 18
Month 24
n
99
96
83
66
62

Note: Enrollment is the dose when enrolled into the SkybriGHt registry.

Figure 4. **Height SDS From SkybriGHt Enrollment to Month 24**

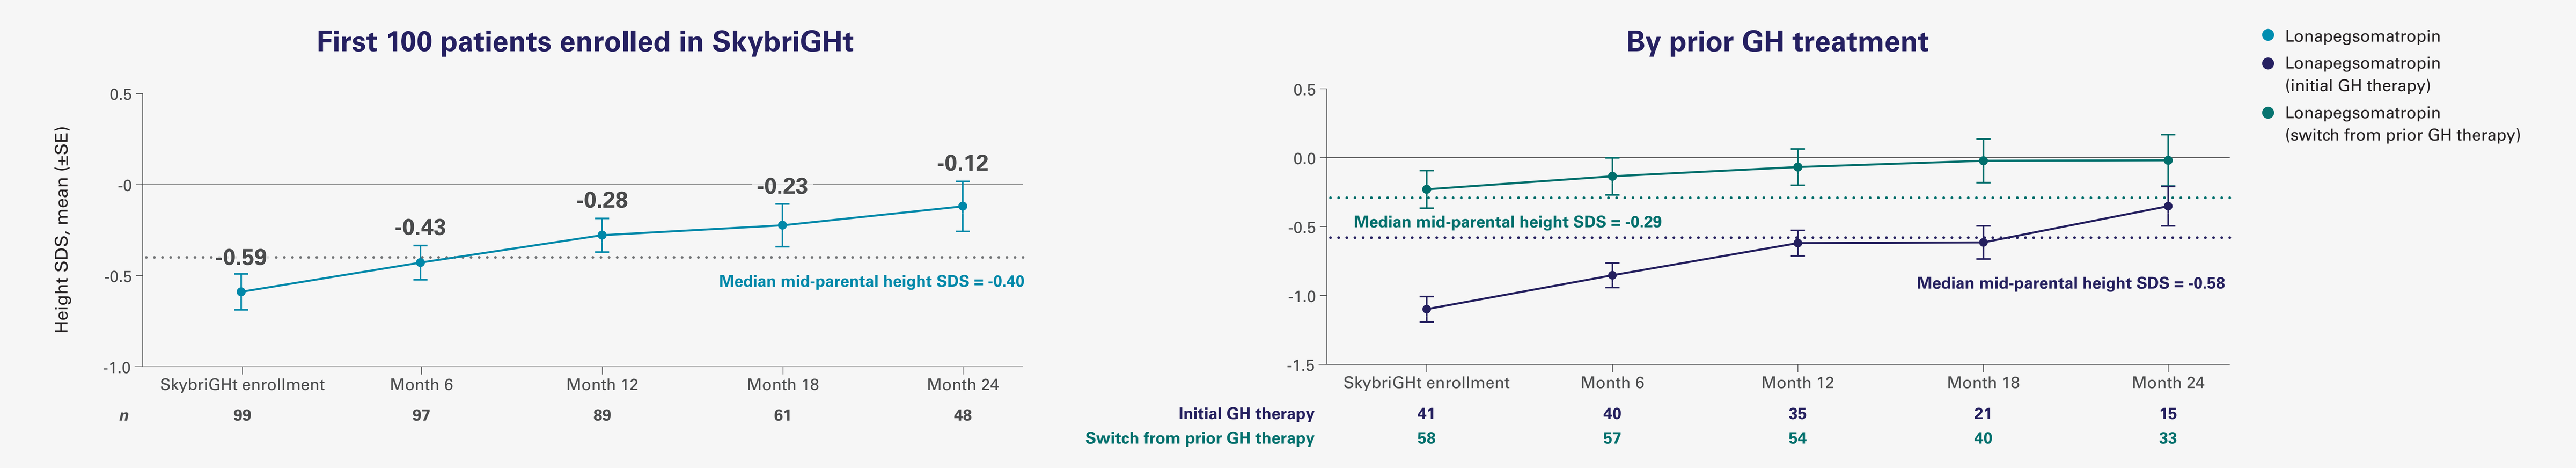
First 100 patients enrolled in SkybriGHt
Height SDS, mean (±SE)
-0.59
-0.43
-0.28
-0.23
-0.12
Median mid-parental height SDS = -0.40
SkybriGHt enrollment
Month 6
Month 12
Month 18
Month 24
n
99
97
89
61
48
By prior GH treatment
Median mid-parental height SDS = -0.29
Median mid-parental height SDS = -0.58
Initial GH therapy
41
40
35
21
15
Switch from prior GH therapy
58
57
54
40
33
Lonapegsomatropin
Lonapegsomatropin (initial GH therapy)
Lonapegsomatropin (switch from prior GH therapy)

Figure 5. **Annualized Height Velocity**

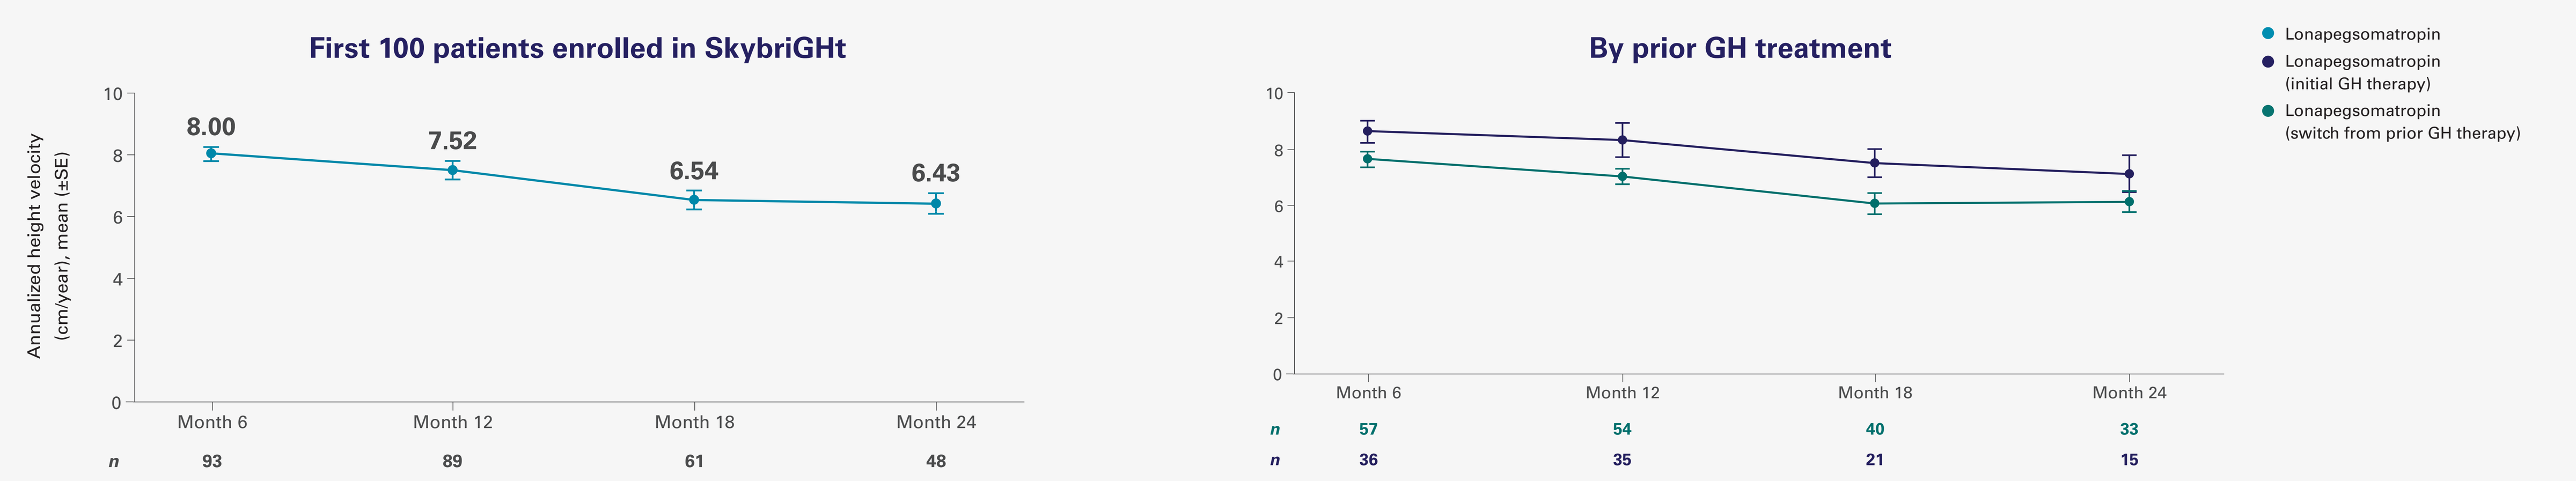
First 100 patients enrolled in SkybriGHt
Annualized height velocity (cm/year), mean (±SE)
8.00
7.52
6.54
6.43
Month 6
Month 12
Month 18
Month 24
n
93
89
61
48
By prior GH treatment
n
57
54
40
33
n
36
35
21
15
Lonapegsomatropin
Lonapegsomatropin (initial GH therapy)
Lonapegsomatropin (switch from prior GH therapy)

Figure 6. **IGF-I SDS Over 24 Months**

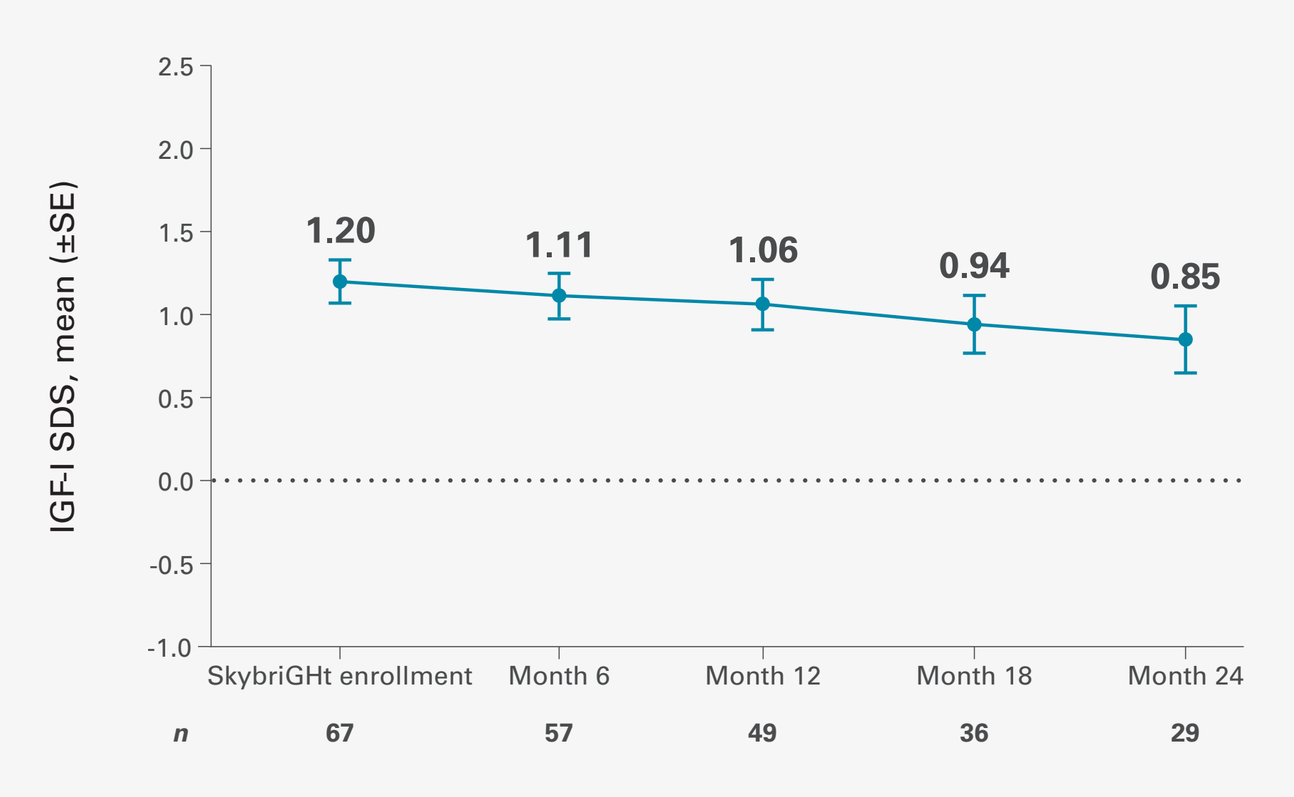
IGF-I SDS, mean (±SE)
1.20
1.11
1.06
0.94
0.85
SkybriGHt enrollment
Month 6
Month 12
Month 18
Month 24
n
67
57
49
36
29

Increases in IGF-I (defined as a level that necessitated a dose reduction) occurred in 14 (14%) participants and were not associated with symptoms.

Note: At each visit, n is the number of children with non-missing IGF-I data. Timing of postdose IGF-I sampling was not reported for all children; therefore, values presented as measured and may not reflect weekly average levels.

Figure 7. **BMI SDS Over 24 Months**

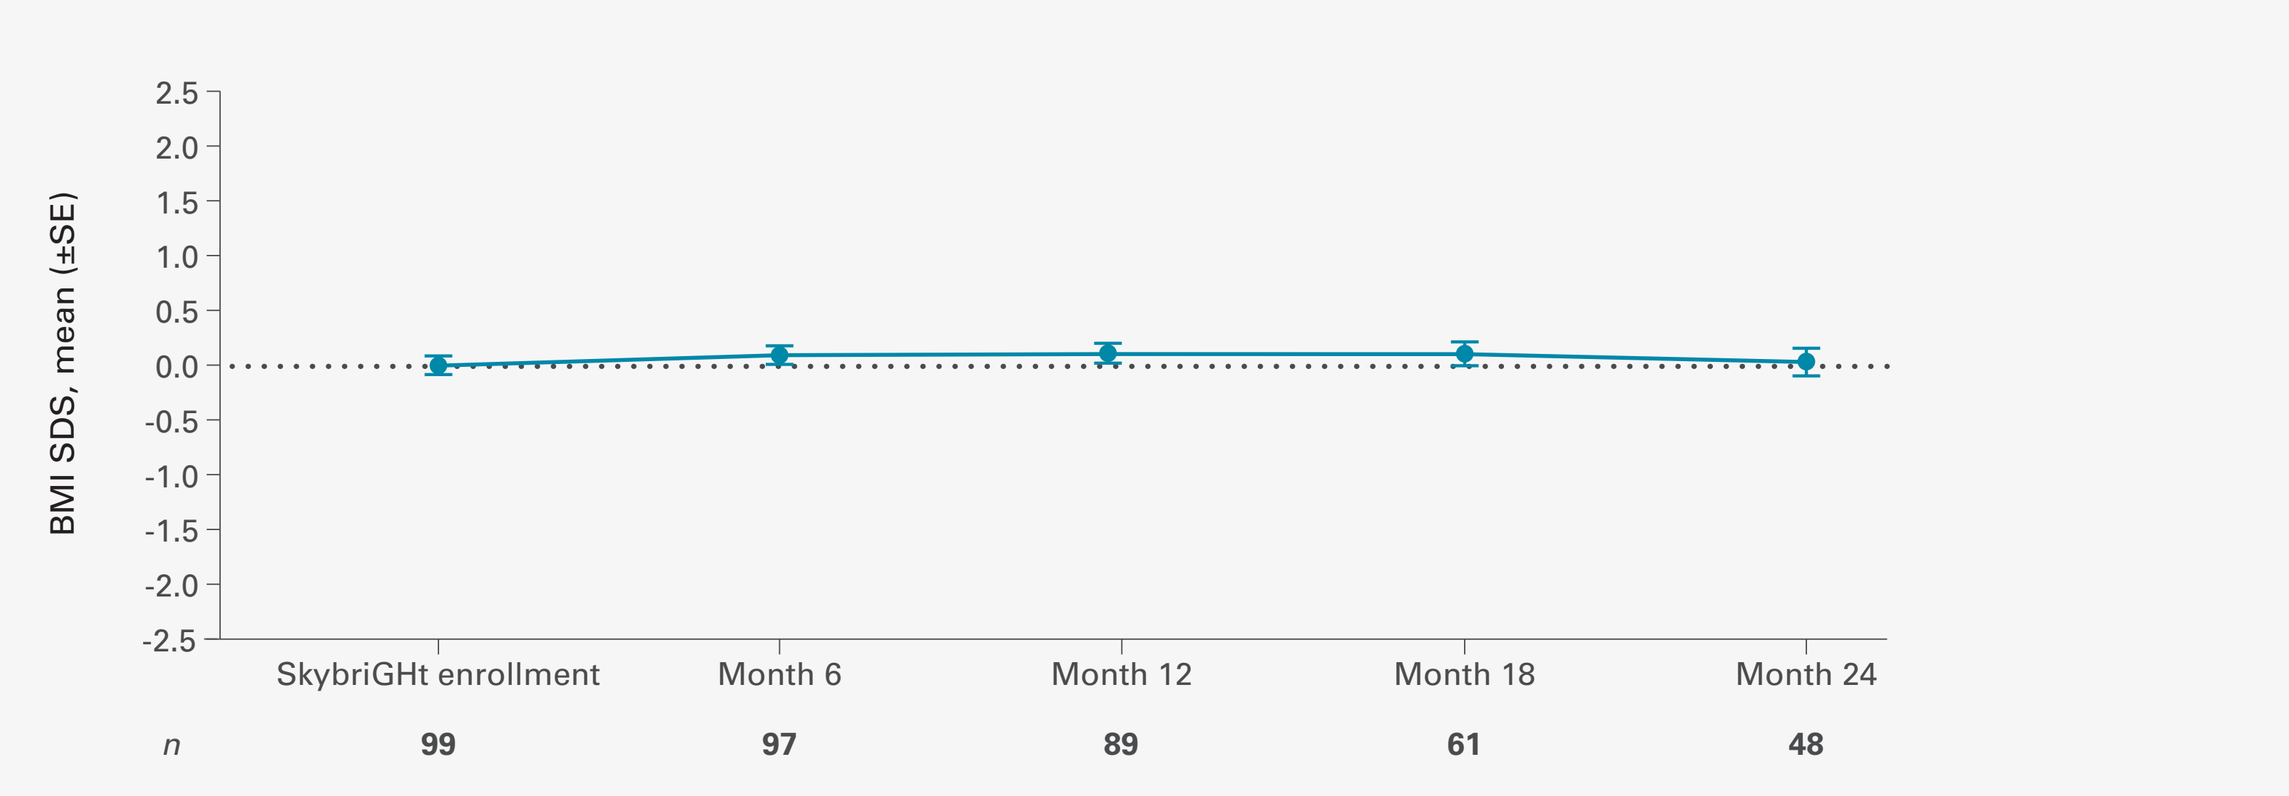
BMI SDS, mean (±SE)
SkybriGHt enrollment
Month 6
Month 12
Month 18
Month 24
n
99
97
89
61
48

Adverse Event Summary

- AEs were reported in 48/100 (48%) children; most were mild or moderate.
- The most commonly reported clinical AEs were upper respiratory tract infection (4%); scoliosis (4%); and otitis media, limb injury, arthralgia, and headache (each 3%).
- No serious or severe treatment-related AEs were reported.
- No AEs led to treatment discontinuation.
- One case of injection site reaction was reported.
- No cases of benign intracranial hypertension or slipped capital femoral epiphysis were observed.

Abbreviations

AE = adverse event; **AHV** = annualized height velocity; **BMI** = body mass index; **GH** = growth hormone; **IGF-I** = insulin-like growth factor I; **SD** = standard deviation; **SDS** = standard deviation score; **SE** = standard error; **US** = United States

References

1. Sprogøe K, Mortensen E, Karpf DB, Leff JA. The rationale and design of TransCon Growth Hormone for the treatment of growth hormone deficiency. *Endocr Connect.* 2017;6(8):R171–r181. doi:10.1530/ec-17-0203. **2.** SKYTROFA® [package insert]. Palo Alto, CA: Ascendis Pharma. October 2022. **3.** EMA Skytrofa: Summary of product characteristics. 2022. **4.** Thornton PS, Maniatis AK, Aghajanova E, et al. Weekly Lonapegsomatropin in Treatment-Naive Children With Growth Hormone Deficiency: The Phase 3 heiGHt Trial. *J Clin Endocrinol Metab.* 2021;106(11):3184–3195. doi:10.1210/clinem/dgab529 **5.** Maniatis AK, Casella SJ, Nadgir UM, et al. Safety and Efficacy of Lonapegsomatropin in Children With Growth Hormone Deficiency: enliGHten Trial 2-Year Results. *J Clin Endocrinol Metab.* 2022;107(7):e2680–e2689. doi:10.1210/clinem/dgac217 **6.** Maniatis AK, Nadgir U, Saenger P, et al. Switching to Weekly Lonapegsomatropin from Daily Somatropin in Children with Growth Hormone Deficiency: The fliGHt Trial. *Horm Res Paediatr.* 2022;95(3):233–243. doi:10.1159/000524003 **7.** Maniatis AK, Thornton PS, Nadgir UM, et al. Children with Growth Hormone Deficiency Treated with Lonapegsomatropin Demonstrated Sustained Height Improvements for up to 6 Years: enliGHten Trial Final Results. *Horm Res Paediatr.* 2025;1–13. doi:10.1159/000545064 **8.** Blonde L, Khunti K, Harris SB, et al. Interpretation and impact of real-world clinical data for the practicing clinician. *Adv Ther.* 2018; 35:1763-1774.

Disclosures and Funding

This study was sponsored by Ascendis Pharma Endocrinology Division A/S

AKM: received honoraria from Ascendis, Novo Nordisk, Pfizer; on the advisory board of Ascendis Pharma, Novo Nordisk, Pfizer; consulted for Ascendis Pharma, Pfizer, and Novo Nordisk; has been speaker bureau for Ascendis Pharma, Novo Nordisk, and Pfizer; and received research funding from Ascendis Pharma, Novo Nordisk, OPKO, and Pfizer; **JHY:** consultant and speaker bureau for Ascendis, Novo Nordisk, Abbvie, Azurity, Tolmar, Sanofi, Kyowa Kirin and Alexion. Consultant for Pfizer and Eton. Principal investigator for Ascendis, Novo Nordisk and Lumos; **MJA:** consultant and research support from Ascendis Pharma and Soleno; research support from Medtronic, Lumos, Novo Nordisk; research support, and speakers' bureau for Rhythm; consultant for Eton, DayOne, Pfizer, and Rezolute; speakers' bureau for Endo and Neurocrine; **AD:** has served as a consultant for Novo Nordisk, Pfizer, BioMarin, Ascendis, QED, and Neurocrine; has received grant funding from BioMarin, Novo Nordisk and Pfizer and served as a site investigator for Lumos, BioMarin, Neurocrine, and Novo Nordisk; **BSM:** consultant for Amgen, Ascendis Pharma, Azurity, Bio Marin, Endo Pharmaceuticals, Eton Pharmaceuticals, Novo Nordisk, QED/BridgeBio, Pfizer, Soleno, Tolmarand Tyra; research support from Aardvark Therapeutics, Alexion, Abbvie, Aeterna Zentaris, Antares Pharma, Ascendis Pharma, BioMarin, Foresee, Foundation for Prader Willi Research, Lumos Pharma, Novo Nordisk, OPKO Health and Pfizer; **THL:** scientific advisory board for Ascendis Pharma; **SC:** serves as a scientific advisor for Ascendis Pharma; **GP:** honoraria, research funding, travel support, advisory board role, consulting role, and speakers' bureau participation for Ascendis, AstraZeneca, Dexcom, Eli Lilly, Johnson and Johnson, Mannkind, Medtronic, Novo Nordisk, Pfizer, and Ultragenyx; and grants from Mannkind Corporation; **DF:** nothing to disclose; **ASK, PML, LD, CZ, JMI, ADS** are employees of Ascendis Pharma, LLC; **PB:** consultant for/has received honoraria from Ascendis Pharma, BioMarin Pharmaceutical, Eton Pharmaceuticals, ICON, Lumos, Novo Nordisk, QED/BridgeBio, Tyra Biosciences, Wolters-Kluwer and Upsher-Smith Laboratories, and receives research support from the NIH/NHLBI (U01HL133883); **ADR:** honoraria from, advisory board role for, consulting role for, and travel support from Ascendis and Tolmar; patents, royalties, or other intellectual property from UpToDate; and other relationships with US Anti-doping Agency and World Anti-doping Agency.

Medical writing support was provided by Tisheeka Graham, PhD, and Sarah Franco, PhD, employees of Ascendis Pharma, LLC.

Ascendis®

MED-US-TC-PGHD-2600048
July 2026